MER1173 ESMO Posters (print) 1
09/09/2024 22:20

POSTER
943TiP

Petosemtamab compared with investigator's choice monotherapy in previously treated patients (pts) with recurrent/metastatic (r/m) head and neck squamous cell carcinoma (HNSCC): A randomized, open-label, phase 3 trial

Robert Haddad,[1] Maura Gillison,[2] Kevin J. Harrington,[3] Sung-Bae Kim,[4] Christophe Le Tourneau,[5] Ari Rosenberg,[6] Jim Ford,[7] Yu-Ming Shen,[7] David Yao,[7] Fabian Zohren,[7] Jean-Pascal Machiels[8]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]MD Anderson Cancer Center, Houston, TX, USA; [3]The Institute of Cancer Research/Royal Marsden, London, UK; [4]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Korea; [5]Institut Curie, Paris, France; [6]University of Chicago, Chicago, IL, USA; [7]Merus N.V., Utrecht, Netherlands; [8]Cliniques Universitaires Saint-Luc, Université Catholique de Louvain, Brussels, Belgium

LiGeR HN2
Bispecific antibody that targets EGFR & LGR5

INTRODUCTION

Petosemtamab is a human, common light chain, IgG1 bispecific antibody with ADCC-enhanced activity, targeting EGFR and LGR5

- EGFR and WNT are oncogenic and mitogenic drivers in several cancer types, including HNSCC[1–3]
- LGR5 is a receptor of WNT signaling and is upregulated in many cancer types[4,5]
- Petosemtamab monotherapy demonstrated meaningful preclinical and clinical antitumor activity in HNSCC[1,6]

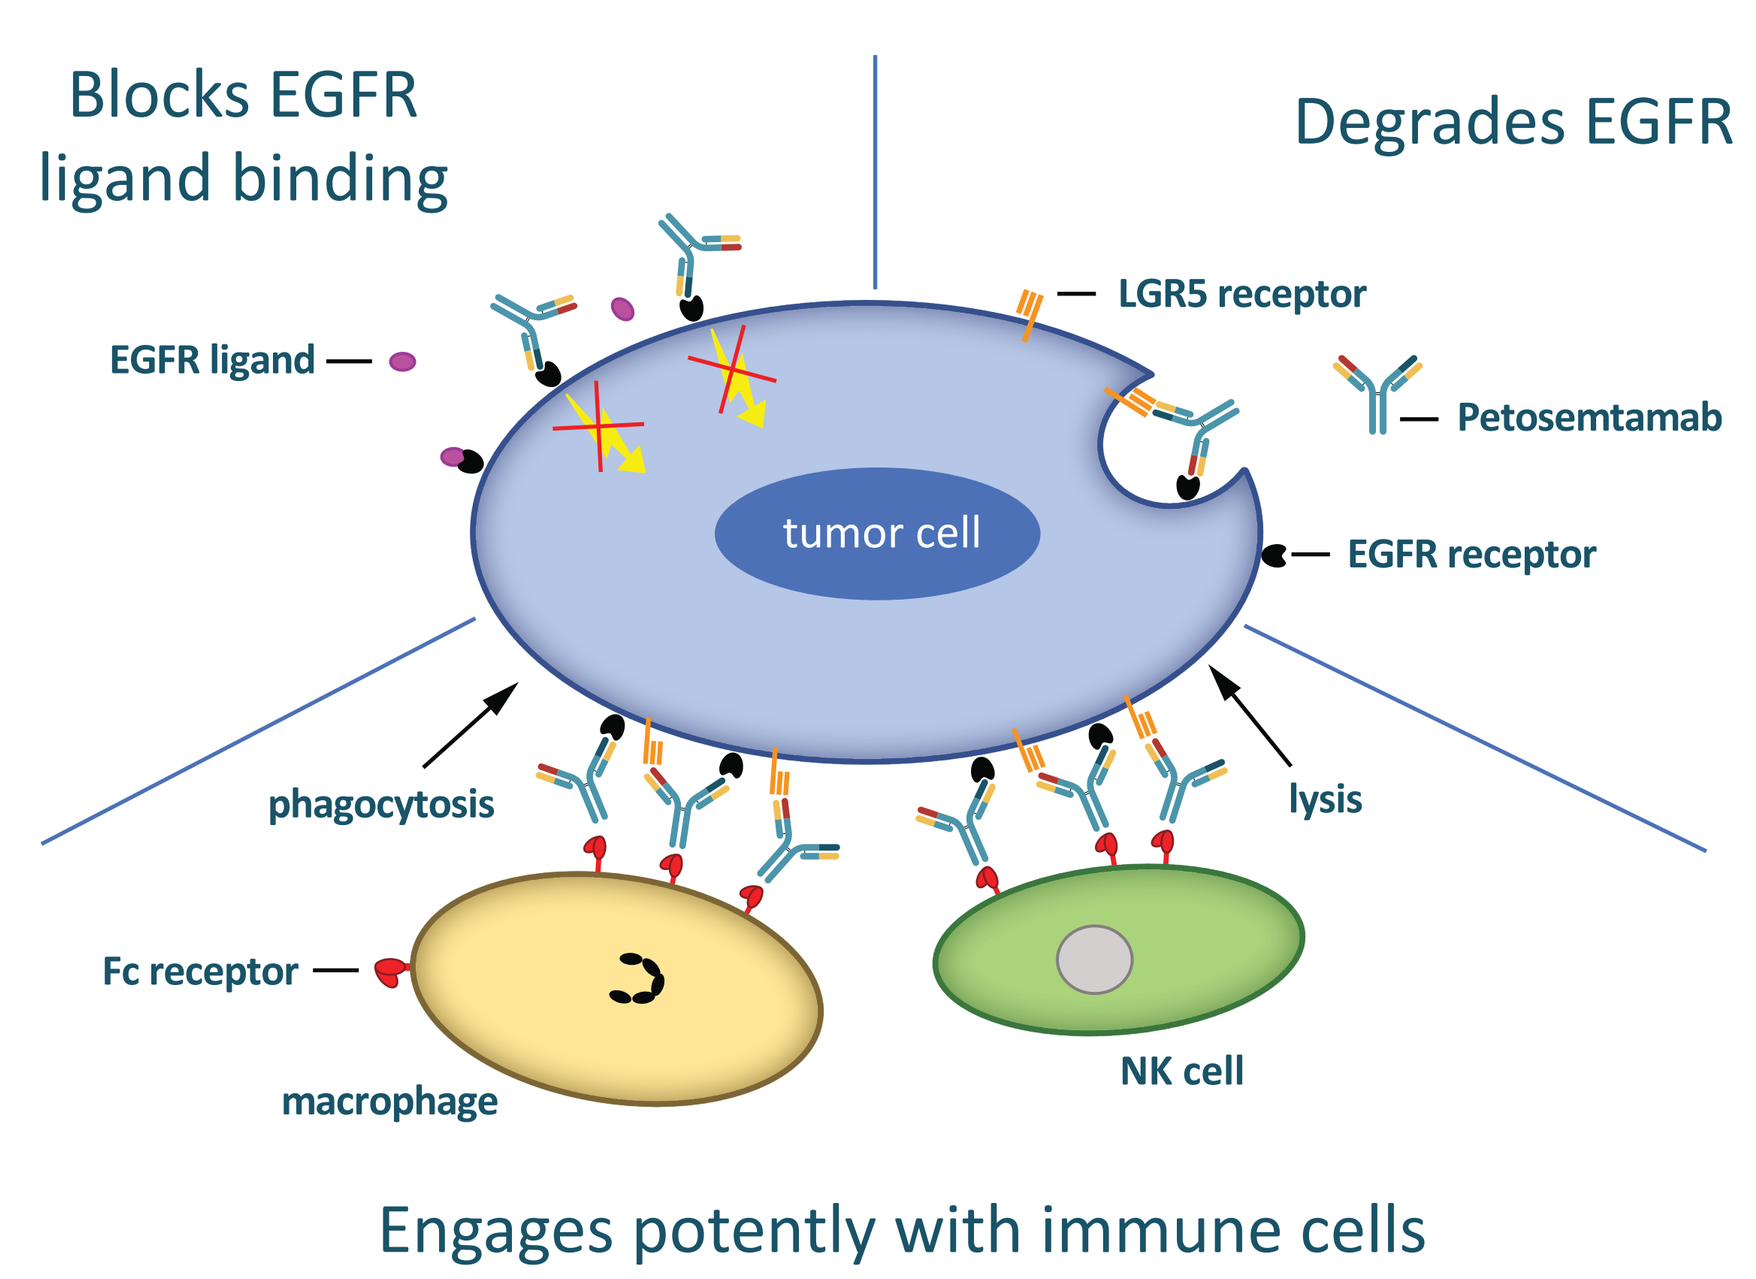
Blocks EGFR ligand binding
Degrades EGFR
EGFR ligand
LGR5 receptor
Petosemtamab
tumor cell
EGFR receptor
phagocytosis
lysis
Fc receptor
macrophage
NK cell
Engages potently with immune cells

Figure 1 | Petosemtamab mechanism of action

Mechanism of action[1]

Petosemtamab was designed to:

- Block EGFR ligand binding and inhibit signaling
- Degrade EGFR (via LGR5/E3 ligase)
- Facilitate interaction with immune cells via ADCP and enhanced ADCC

Head and neck squamous cell carcinoma

- High prevalence and mortality with dismal prognosis[7]
 - Sixth most common cancer worldwide in 2020[7] and ~930,000 new cases and 467,000 deaths[7]
- Unmet medical need in the platinum and anti-PD-1 refractory setting
 - Limited treatment options, and no standard of care, after platinum-based chemotherapy and pembrolizumab[8–11]
 - 6–19% ORR and median OS of 5–9 months in 2L+ setting with cetuximab, docetaxel, or methotrexate[8–11]
- LGR5 expression reported in 52–89% of HNSCC[12,13]
- EGFR overexpression reported in ~90% of HNSCC[14,15]

2L+: previously treated; ADCC: antibody-dependent cellular cytotoxicity; ADCP: antibody-dependent cellular phagocytosis; EGFR: epidermal growth factor receptor; Fc: fragment crystallizable; HNSCC: head and neck squamous cell carcinoma; IgG1: immunoglobulin G1; LGR5: leucine-rich repeat-containing G-protein coupled receptor 5; NK: natural killer; ORR: overall response rate; OS: overall survival; PD-1: programmed cell death protein 1; WNT: wingless/integrated.

LiGeR-HN2 PHASE 3 TRIAL DESIGN

LiGeR-HN2 is a phase 3, open-label, randomized, controlled, multicenter trial to compare petosemtamab vs. investigator's choice monotherapy in patients with HNSCC for the 2L and 3L treatment of incurable metastatic/recurrent disease

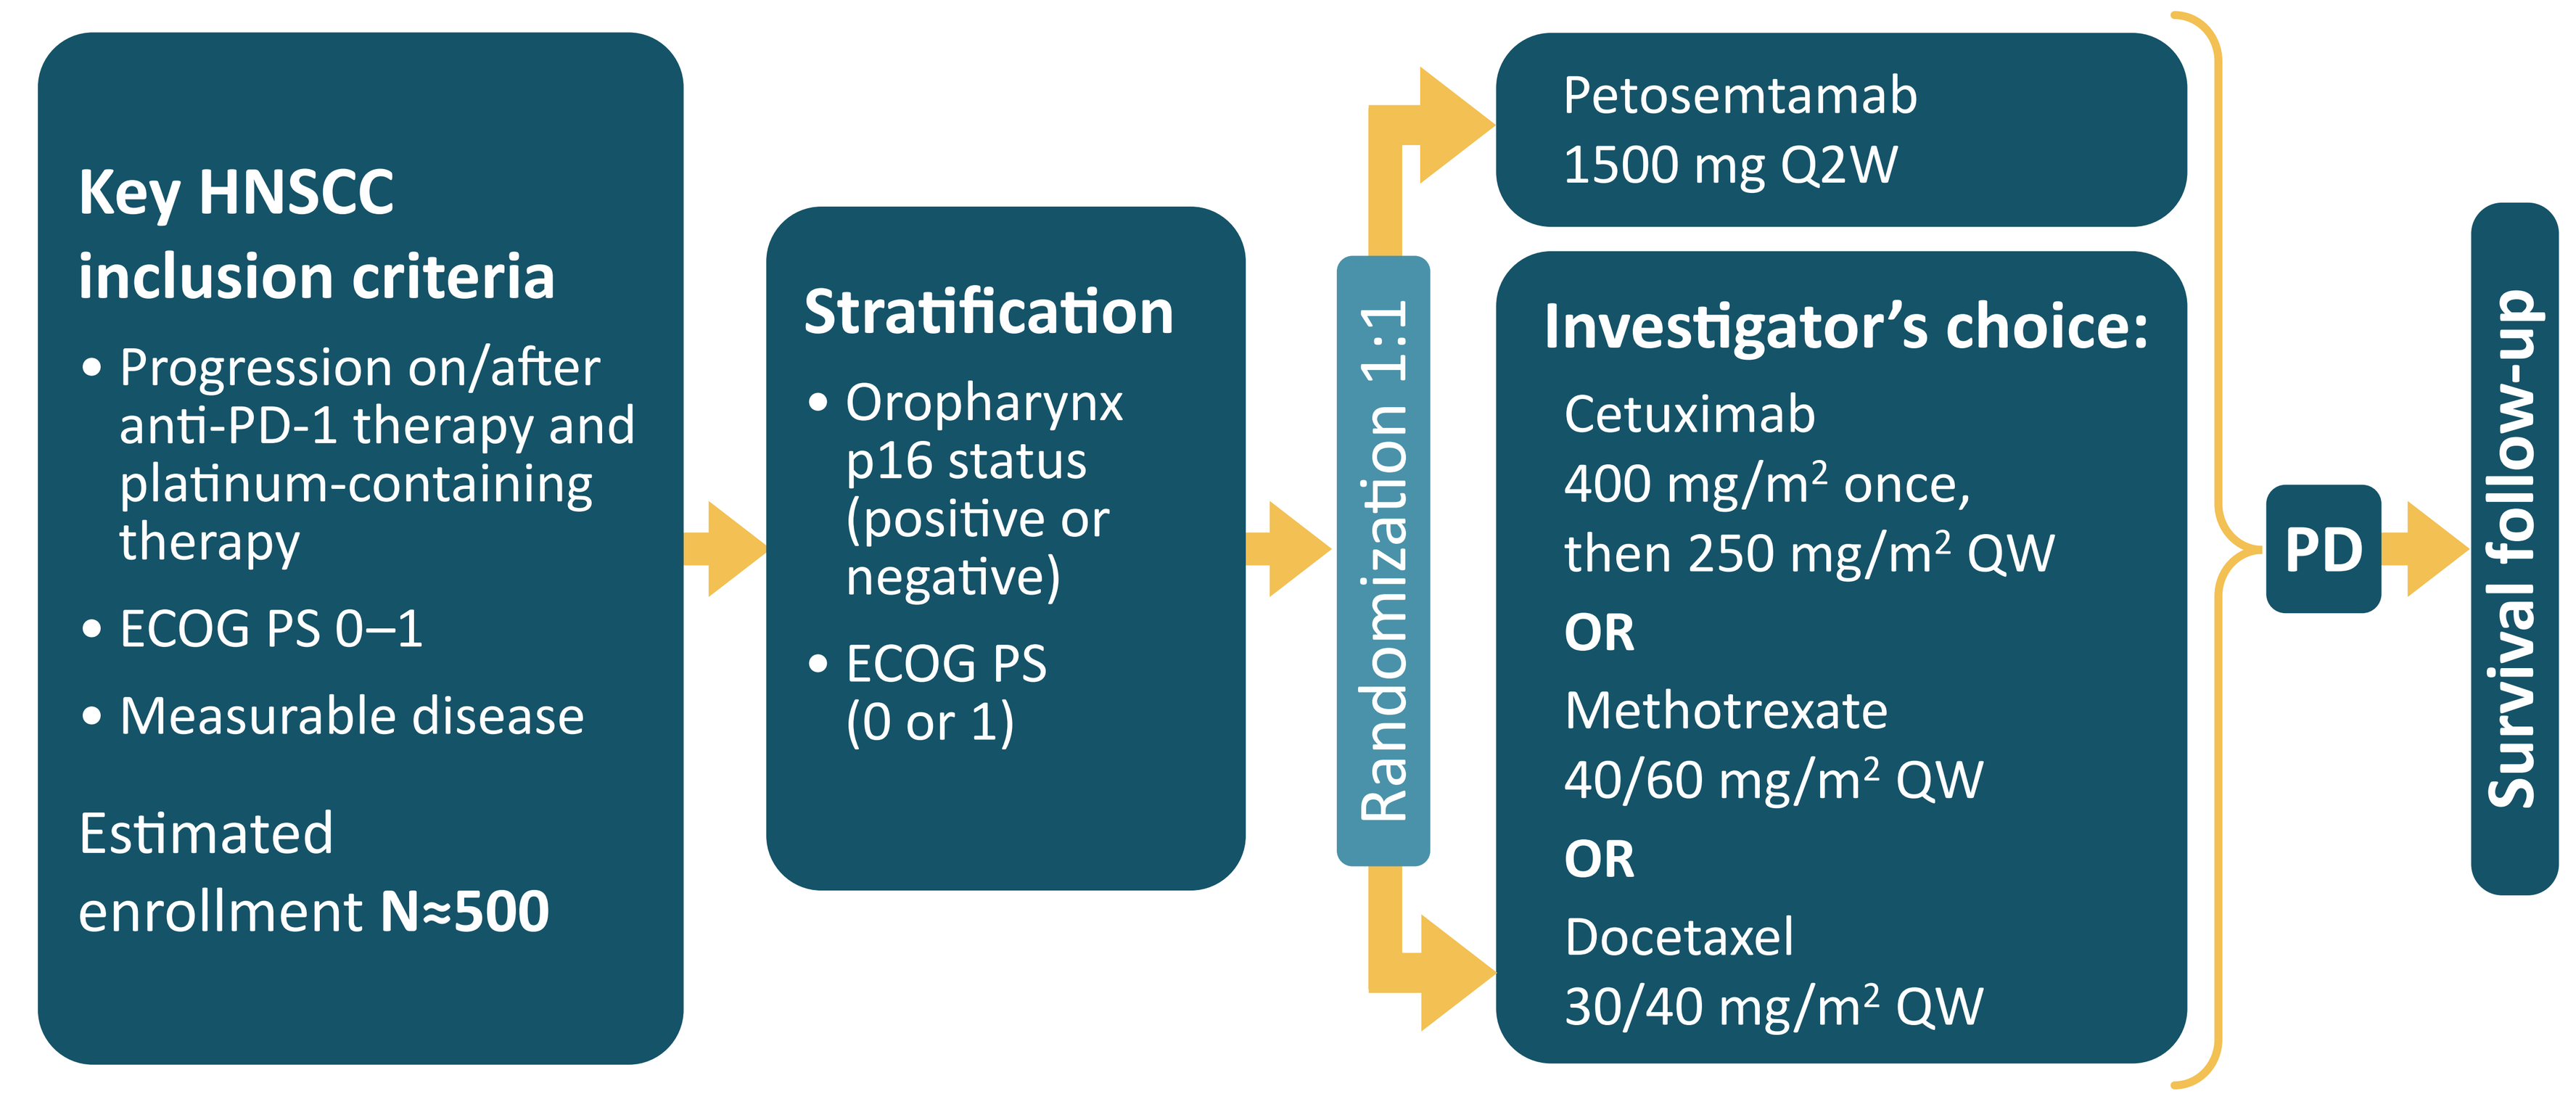
Key HNSCC inclusion criteria
• Progression on/after anti-PD-1 therapy and platinum-containing therapy
• ECOG PS 0–1
• Measurable disease
Estimated enrollment N≈500
Stratification
• Oropharynx p16 status (positive or negative)
• ECOG PS (0 or 1)
Randomization 1:1
Petosemtamab 1500 mg Q2W
Investigator's choice:
Cetuximab 400 mg/m² once, then 250 mg/m² QW
OR
Methotrexate 40/60 mg/m² QW
OR
Docetaxel 30/40 mg/m² QW
PD
Survival follow-up

Figure 2 | LiGeR-HN2 trial design

No crossover is permitted. 2L: second line; 3L: third line; ECOG PS: Eastern Cooperative Oncology Group performance status; PD: progressive disease; Q2W: once every 2 weeks; QW: once weekly.

PHASE 2 TRIAL INTERIM DATA: AACR® 2023

Overall response rate (RECIST v1.1, per investigator)

Best percent change in sum of target lesions from baseline (N=43)[6]

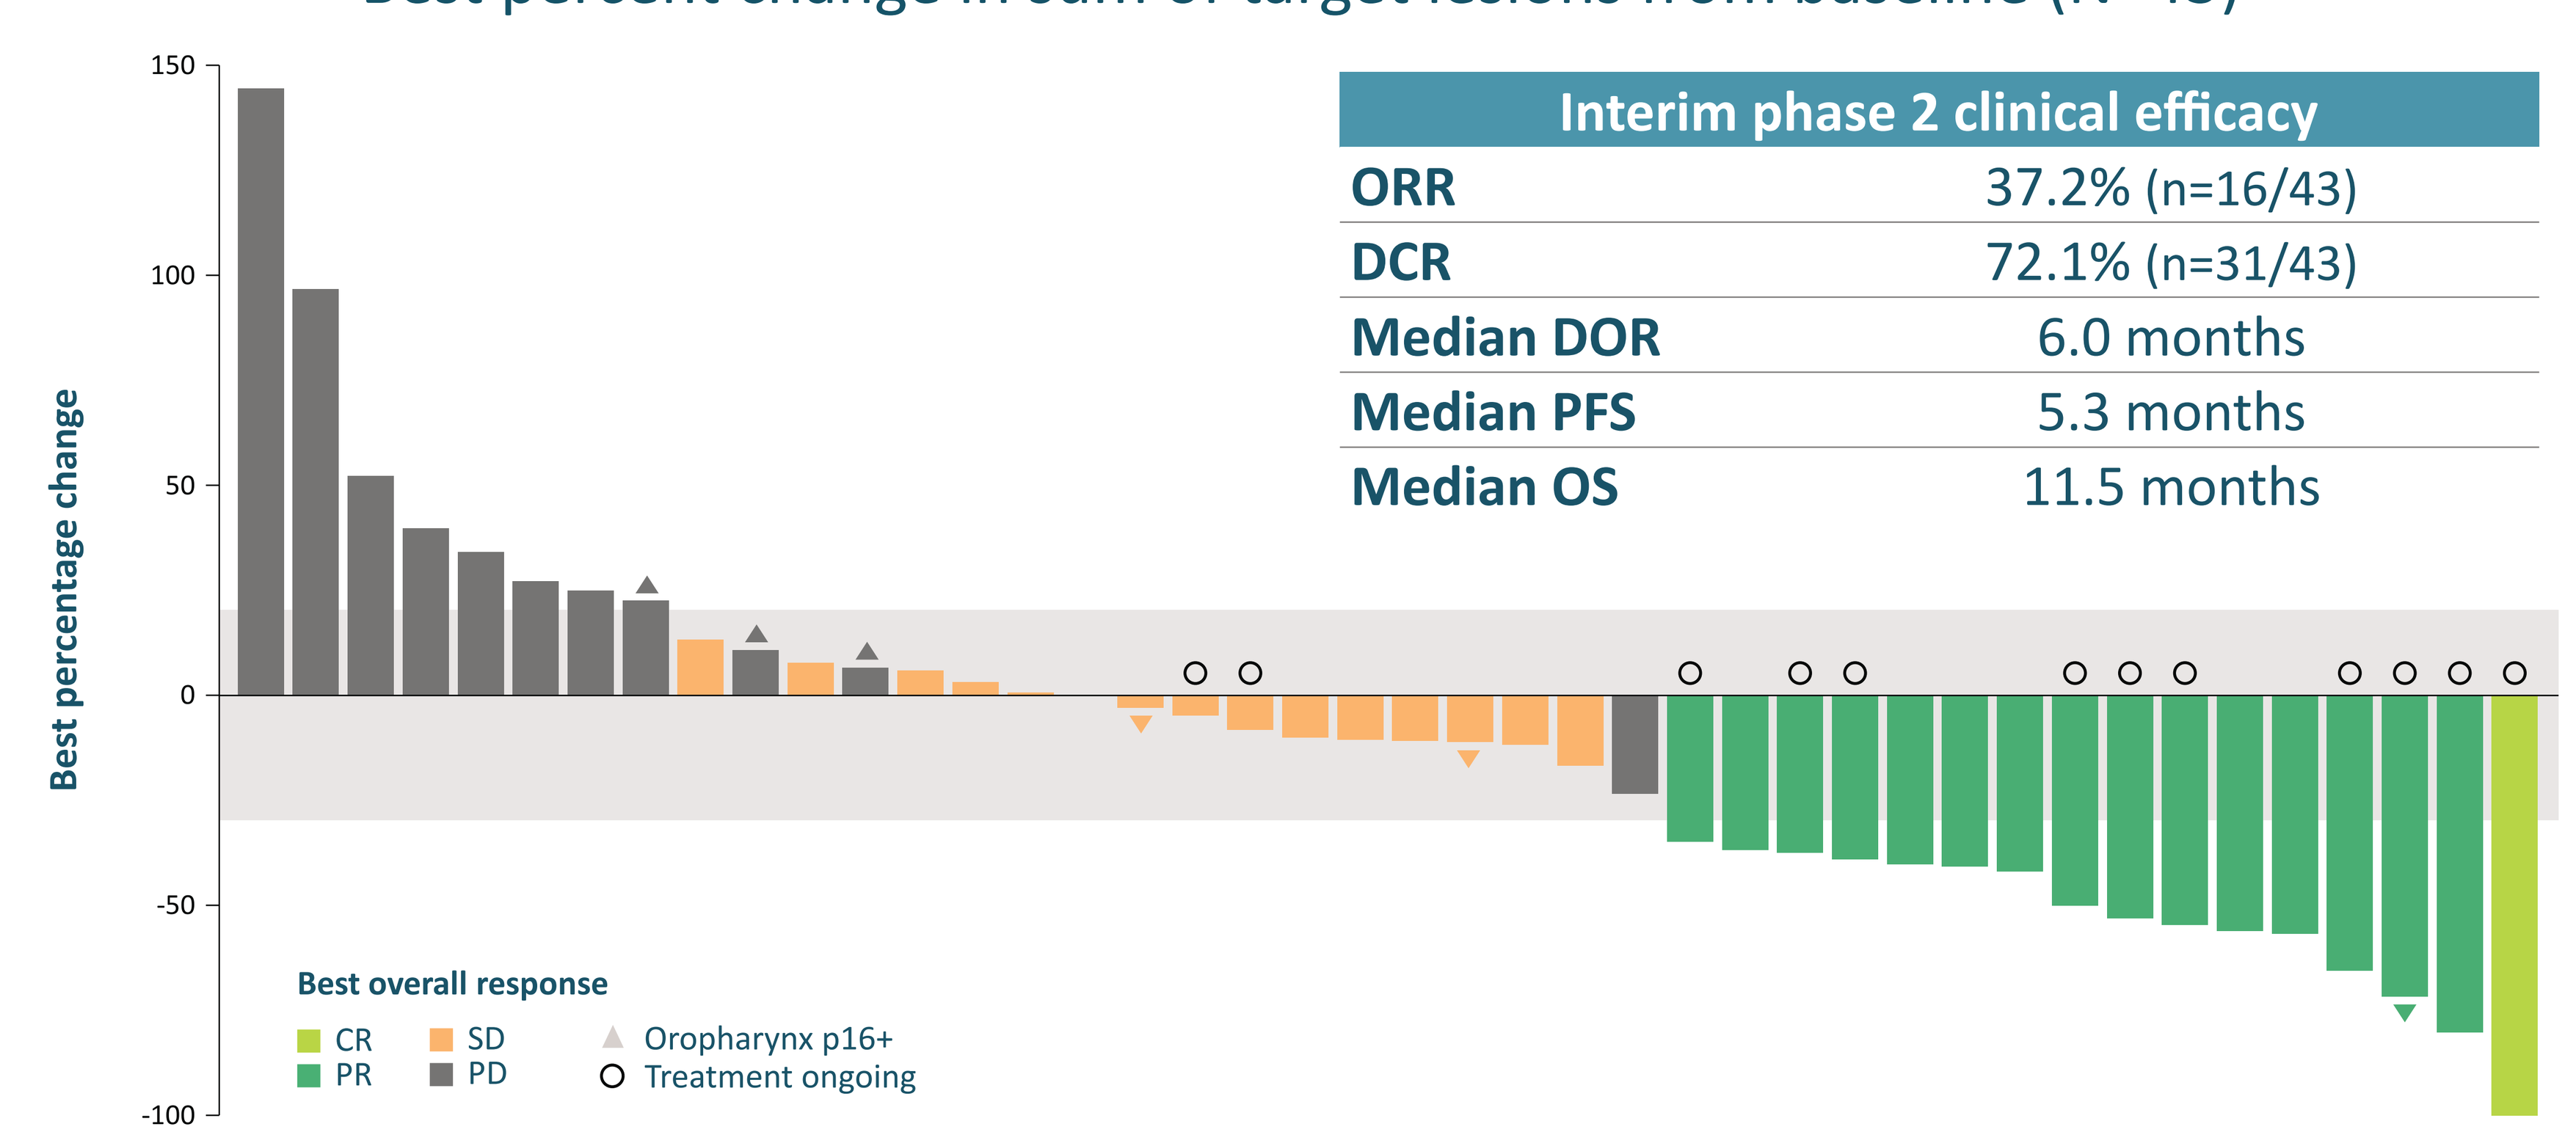
Best percentage change
150
100
50
0
-50
-100
Best overall response
CR
PR
SD
PD
Oropharynx p16+
Treatment ongoing

Interim phase 2 clinical efficacy	
ORR	37.2% (n=16/43)
DCR	72.1% (n=31/43)
Median DOR	6.0 months
Median PFS	5.3 months
Median OS	11.5 months

Figure 3 | Petosemtamab monotherapy in 2L+ r/m HNSCC: Phase 2 data (NCT03526835)

Petosemtamab monotherapy was observed to be well tolerated with a manageable safety profile[6]

Data cutoff date: 01 Feb 2023.
Data from one patient with best overall response of 'not evaluable' were not included due to no post-baseline tumor assessment; p16 status was available in 9 of the 15 patients with oropharyngeal cancer (6 positive and 3 negative) in the efficacy evaluable population.
AACR: American Association for Cancer Research; CR: complete response; DCR: disease control rate; DOR: duration of response; PFS: progression-free survival; PR: partial response; RECIST: Response Evaluation Criteria in Solid Tumors; r/m: recurrent/metastatic; SD: stable disease.

LiGeR-HN2 METHODS

Inclusion criteria

- Age ≥18 years at signing of ICF
- Histologically previously confirmed HNSCC with evidence of metastatic or locally advanced disease not amenable to standard therapy with curative intent
- Progressed on or after anti-PD-1 therapy and platinum-containing therapy
- HNSCC primary tumor locations are oropharynx, oral cavity, hypopharynx, and larynx
- Documentation of p16 status (positive or negative) by local laboratory IHC for patients with primary oropharyngeal cancer
- Measurable disease as defined by RECIST v1.1 by radiologic methods
- ECOG PS of 0 or 1

Exclusion criteria

- CNS metastases that are untreated or symptomatic, or require radiation, surgery, or continued steroid therapy to control symptoms within 14 days of study entry
- Known leptomeningeal involvement
- Any systemic anticancer therapy within 4 weeks of the first dose of study treatment
- Primary tumor site of nasopharynx (any histology)

Primary outcome measures

- Objective response rate as assessed by BICR
- Overall survival

Secondary outcome measures

- Assessed by BICR and by investigator:
 - Progression-free survival
 - Duration of response
 - Time to response
 - Clinical benefit rate
- Objective response rate as assessed by investigator
- Number of participants who experienced:
 - At least one TEAE
 - At least one serious TEAE
- Number of participants who:
 - Discontinued study treatment due to TEAEs
 - Had dose modification due to TEAEs
- Mean change from baseline in EORTC QLQ-C30 and QLQ-H&N43
- Concentrations predose and at end of infusion
- Pharmacokinetic parameters
- Incidence of antidrug antibody

BICR: blinded independent central review; CNS: central nervous system; EORTC: European Organisation for Research and Treatment of Cancer; ICF: informed consent form; IHC: immunohistochemistry; QLQ-C30: 30-item Quality of Life Questionnaire; QLQ-H&N43: 43-item Quality of Life Head and Neck Cancer Module; TEAE: treatment-emergent adverse event.

References

1. Herpers B, et al. (2022) Nat Cancer *3*, 418–436.
2. Schinke H, et al. (2022) Mol Cancer *21*, 178.
3. Parsons MJ, et al. (2021) Cancer Discov *11*, 2413–2429.
4. Xu L, et al. (2019) Stem Cell Res Ther *10*, 219.
5. Katoh M. (2017) Int J Oncol *51*, 1357–1369.
6. Cohen E, et al. (2023) Cancer Res *83(Suppl 8)*, CT012 [oral, AACR 2023].
7. Sung H, et al. (2021) CA Cancer J Clin *71*, 209–249.
8. Cohen E, et al. (2019) Lancet *393*, 156–167.
9. Ferris RL, et al. (2016) N Engl J Med *375*, 1856–1867.
10. Vermorken JB, et al. (2007) J Clin Oncol *25*, 2171–2177.
11. Fayette J, et al. (2023) Ann Oncol *34(Suppl 2)*, S554–S593 [oral, ESMO 2023].
12. Wu Z, et al. (2017) Int J Clin Exp Pathol *10*, 11267–11275.
13. Dalley AJ, et al. (2015) Oral Surg Oral Med Oral Pathol Oral Radiol *119*, 436–440.
14. Byeon HK, et al. (2019) Exp Mol Med *51*, 1–14.
15. Xu MJ, et al. (2017) Cancer Metastasis Rev *36*, 463–473.

Acknowledgments

We thank the investigators and their teams; Merus colleagues; CROs and vendors; and Sophie Houten at LiNK Health Group. Study sponsored by Merus NV.

Presenting Author Disclosures

RH is a panel chair guidelines member for the National Comprehensive Cancer Network (NCCN); on the Data Safety Monitoring Board for Hookipa, Nanobiotix, and PSI; an advisory board consultant for AstraZeneca, Bayer, Bristol Myers Squibb, Boehringer Ingelheim, Coherus, Eisai, EMD Serono, Exelixis, Genentech, Genmab, Merck, Merus, and Pfizer; and has received institutional research funding from AstraZeneca, Bristol Myers Squibb, EMD Serono, Incyte, Kura, and Merck.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the European Society for Medical Oncology (ESMO®) Annual Meeting; September 13–17, 2024; Barcelona, Spain

ClinicalTrials.gov identifier: NCT06496178

Contact information: David Yao (d.yao@merus.nl)