POSTER
944TiP

Petosemtamab plus pembrolizumab vs. pembrolizumab for first-line (1L) treatment of recurrent/metastatic (r/m) PD-L1–positive head and neck squamous cell carcinoma (HNSCC): A randomized, open-label, phase 3 trial

Jérôme Fayette,[1] Douglas Adkins,[2] Amanda Psyrri,[3] Lillian L. Siu,[4] Makoto Tahara,[5] William N. William Jr.,[6] Rebecca Pyle,[7] Yu-Ming Shen,[7] Fabian Zohren,[7] Eduardo Pennella,[7] Everett Vokes[8]

[1]Léon Bérard Center, University of Lyon, Lyon, France; [2]Washington University School of Medicine, St. Louis, MO, USA; [3]National Kapodistrian University of Athens Medical School, Athens, Greece; [4]Princess Margaret Cancer Centre, Toronto, ON, Canada; [5]National Cancer Center Hospital East, Kashiwa, Japan; [6]Grupo Oncoclínicas, São Paulo, Brazil; [7]Merus N.V., Utrecht, Netherlands; [8]University of Chicago Medical Center, Chicago, IL, USA

LiGeR HN1
Bispecific antibody that targets EGFR & LGR5

INTRODUCTION

Petosemtamab is a human, common light chain, IgG1 bispecific antibody with ADCC-enhanced activity, targeting EGFR and LGR5

- EGFR and WNT are oncogenic and mitogenic drivers in several cancer types, including HNSCC[1–3]; LGR5 is a receptor of WNT signaling and is upregulated in many cancer types[4,5]
- Petosemtamab monotherapy demonstrated meaningful preclinical and clinical antitumor activity in HNSCC[1,6]
- The combination of petosemtamab plus pembrolizumab demonstrated clinically meaningful activity in 1L PD-L1+ HNSCC[7]

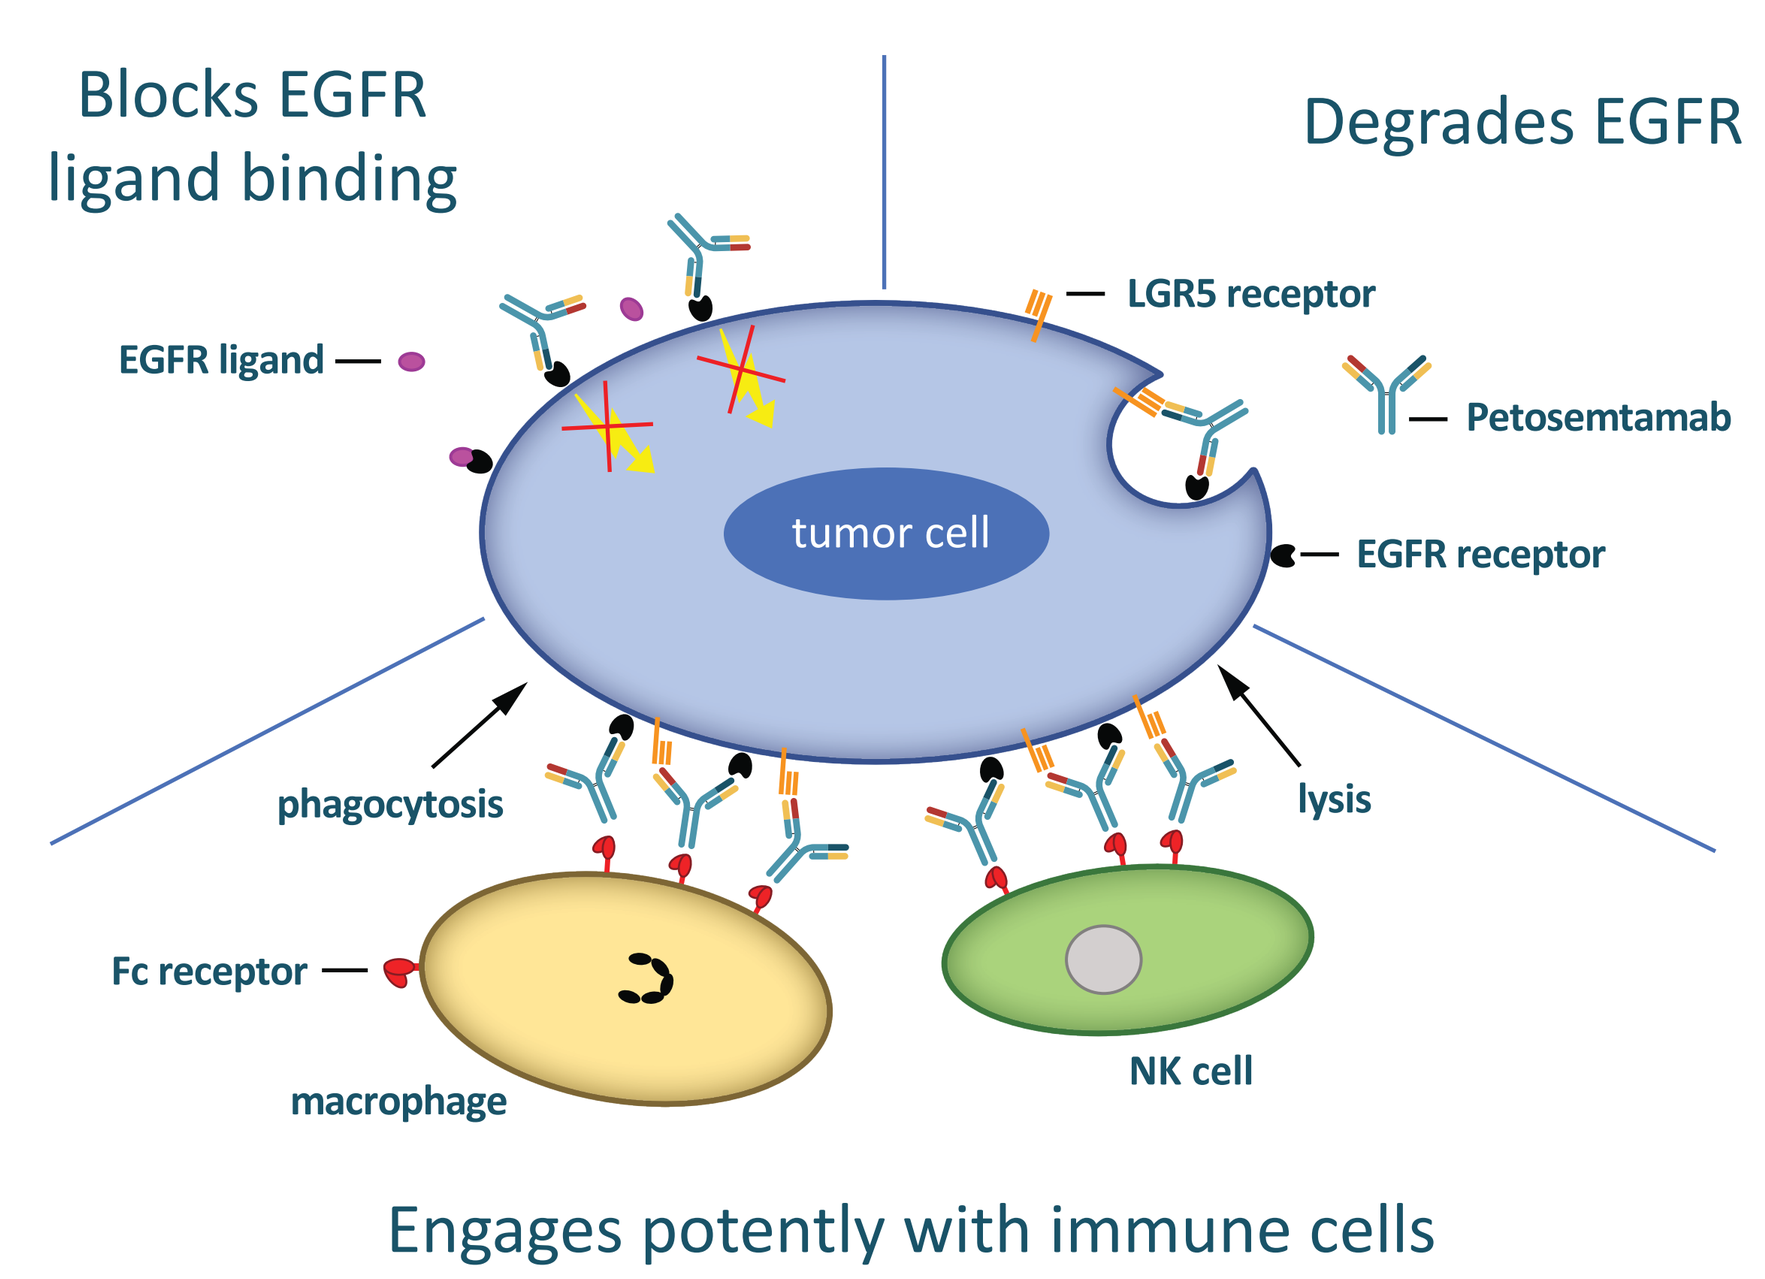

Blocks EGFR ligand binding
Degrades EGFR
EGFR ligand
LGR5 receptor
Petosemtamab
tumor cell
EGFR receptor
phagocytosis
lysis
Fc receptor
macrophage
NK cell
Engages potently with immune cells

Figure 1 | Petosemtamab mechanism of action

Mechanism of action[1]

Petosemtamab was designed to:

- Block EGFR ligand binding and inhibit signaling
- Degrade EGFR (via LGR5/E3 ligase)
- Facilitate interaction with immune cells via ADCP and enhanced ADCC

Head and neck squamous cell carcinoma

- High prevalence and mortality with dismal prognosis[8]
 - Sixth most common cancer worldwide in 2020 with ~930,000 new cases and 467,000 deaths[8]
- Unmet need for a more effective and tolerable 1L treatment of PD-L1+ r/m HNSCC
 - Pembrolizumab with or without chemotherapy is the current standard of care for 1L treatment of PD-L1+ r/m HNSCC[9]
 - In a study evaluating pembrolizumab in 1L PD-L1+ HNSCC, ORR was 19% and median OS was 12.3 months[10]
 - This highlights poor survival prognosis and an unmet need for improved therapies for this patient population
- LGR5 expression reported in 52–89% of HNSCC[11,12]
- EGFR overexpression reported in ~90% of HNSCC[13,14]

1L: first-line; ADCC: antibody-dependent cellular cytotoxicity; ADCP: antibody-dependent cellular phagocytosis; EGFR: epidermal growth factor receptor; Fc: fragment crystallizable; HNSCC: head and neck squamous cell carcinoma; IgG1: immunoglobulin G1; LGR5: leucine-rich repeat-containing G-protein coupled receptor 5; NK: natural killer; ORR: overall response rate; OS: overall survival; PD-L1(+): programmed cell death ligand 1 (positive); r/m: recurrent/metastatic; WNT: wingless/integrated.

LiGeR-HN1 PHASE 3 TRIAL DESIGN

LiGeR-HN1 is a randomized, open-label, phase 3 trial to evaluate the efficacy and safety of petosemtamab plus pembrolizumab vs. pembrolizumab monotherapy in patients with 1L r/m PD-L1+ HNSCC

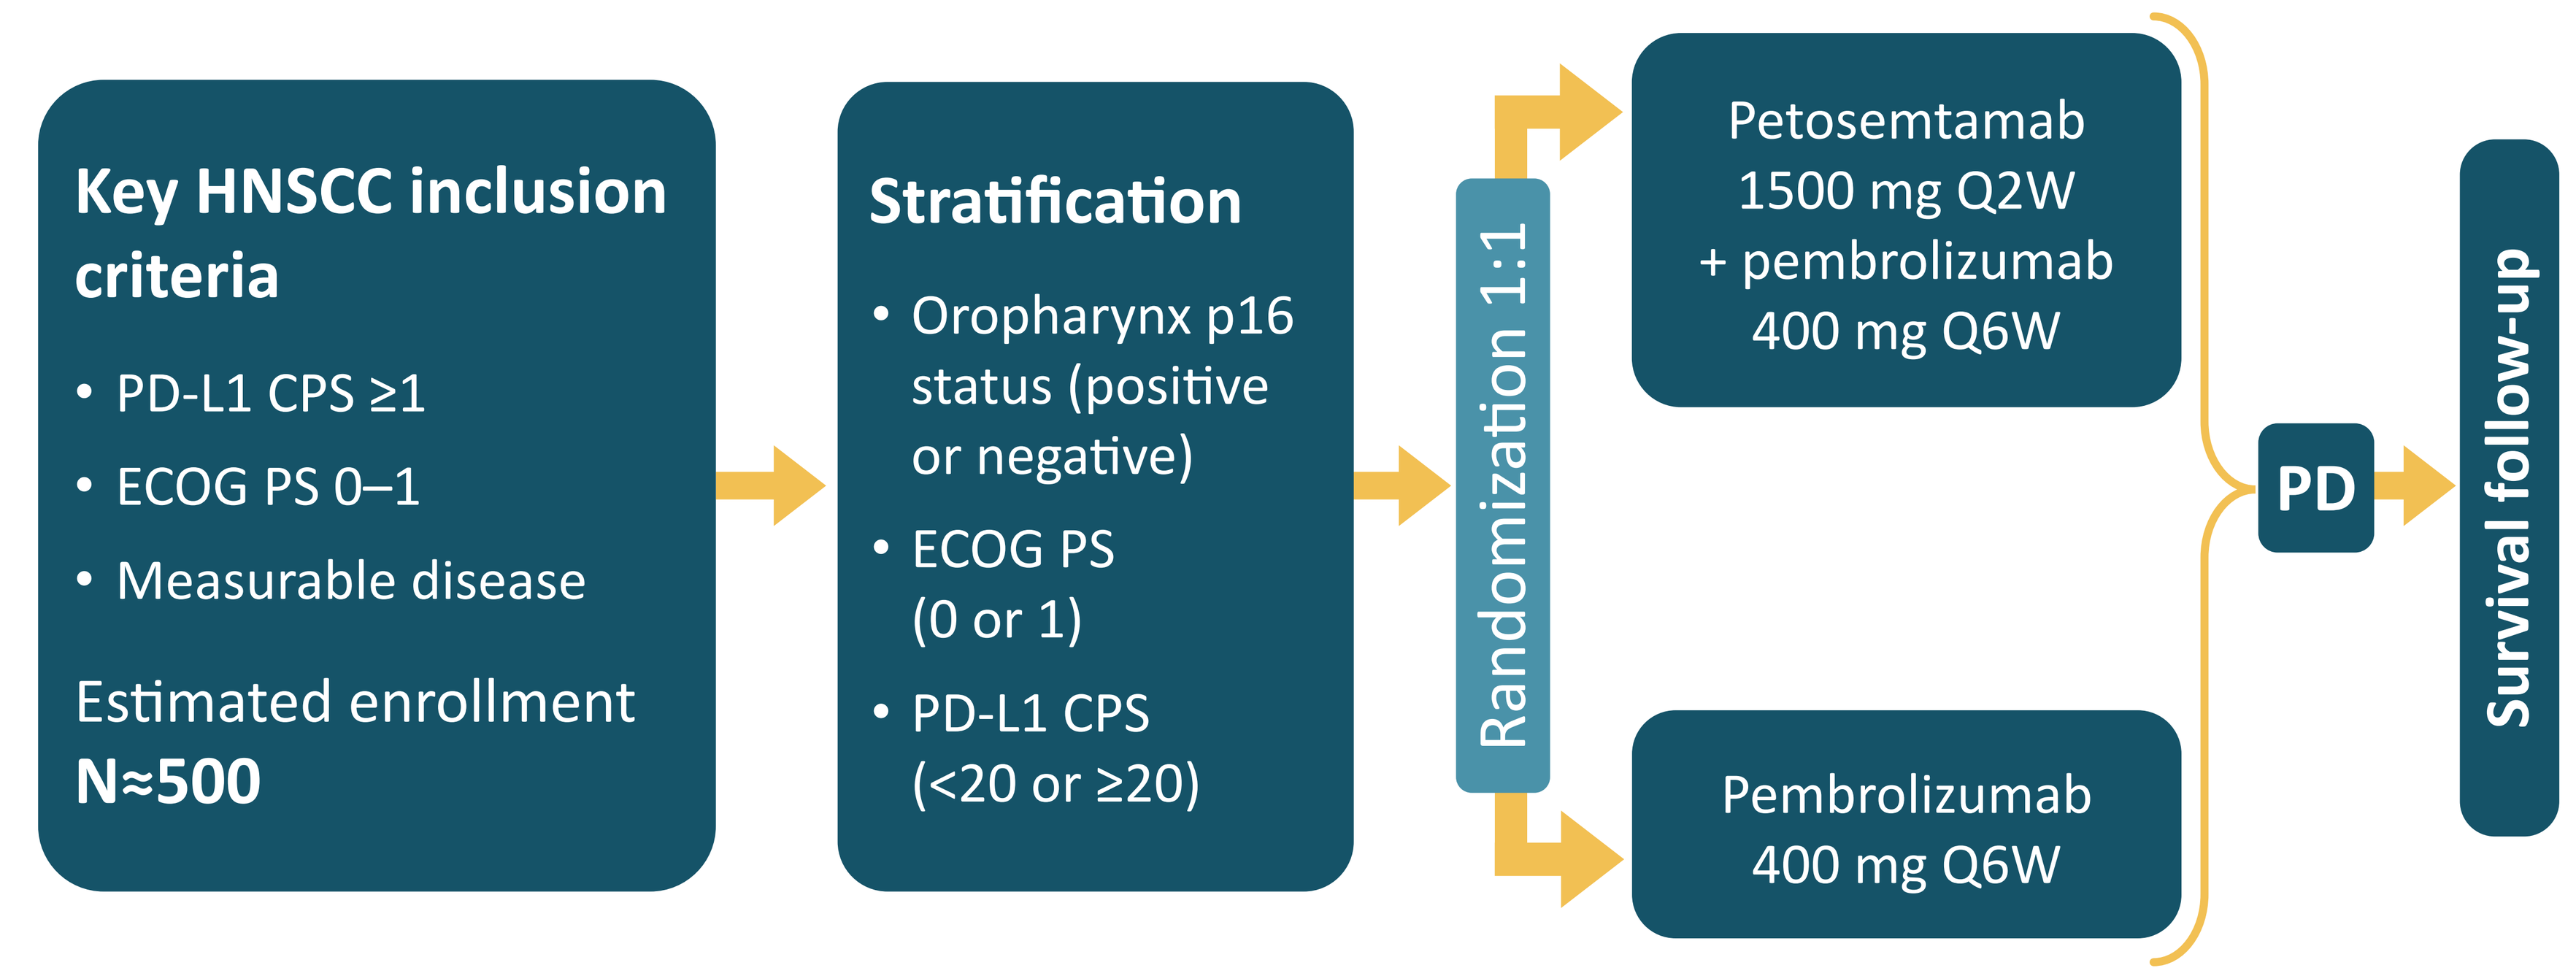

Key HNSCC inclusion criteria
• PD-L1 CPS ≥1
• ECOG PS 0–1
• Measurable disease
Estimated enrollment
N≈500
Stratification
• Oropharynx p16 status (positive or negative)
• ECOG PS (0 or 1)
• PD-L1 CPS (<20 or ≥20)
Randomization 1:1
Petosemtamab 1500 mg Q2W + pembrolizumab 400 mg Q6W
Pembrolizumab 400 mg Q6W
PD
Survival follow-up

Figure 2 | LiGeR-HN1 trial design

No crossover is permitted. CPS: combined positive score; ECOG PS: Eastern Cooperative Oncology Group performance status; PD: progressive disease; Q2W: once every 2 weeks; Q6W: once every 6 weeks.

PHASE 2 TRIAL INTERIM DATA: 2024 ASCO®

Overall response rate (RECIST v1.1, per investigator)

Best percent change in sum of target lesions from baseline (N=24)[7]

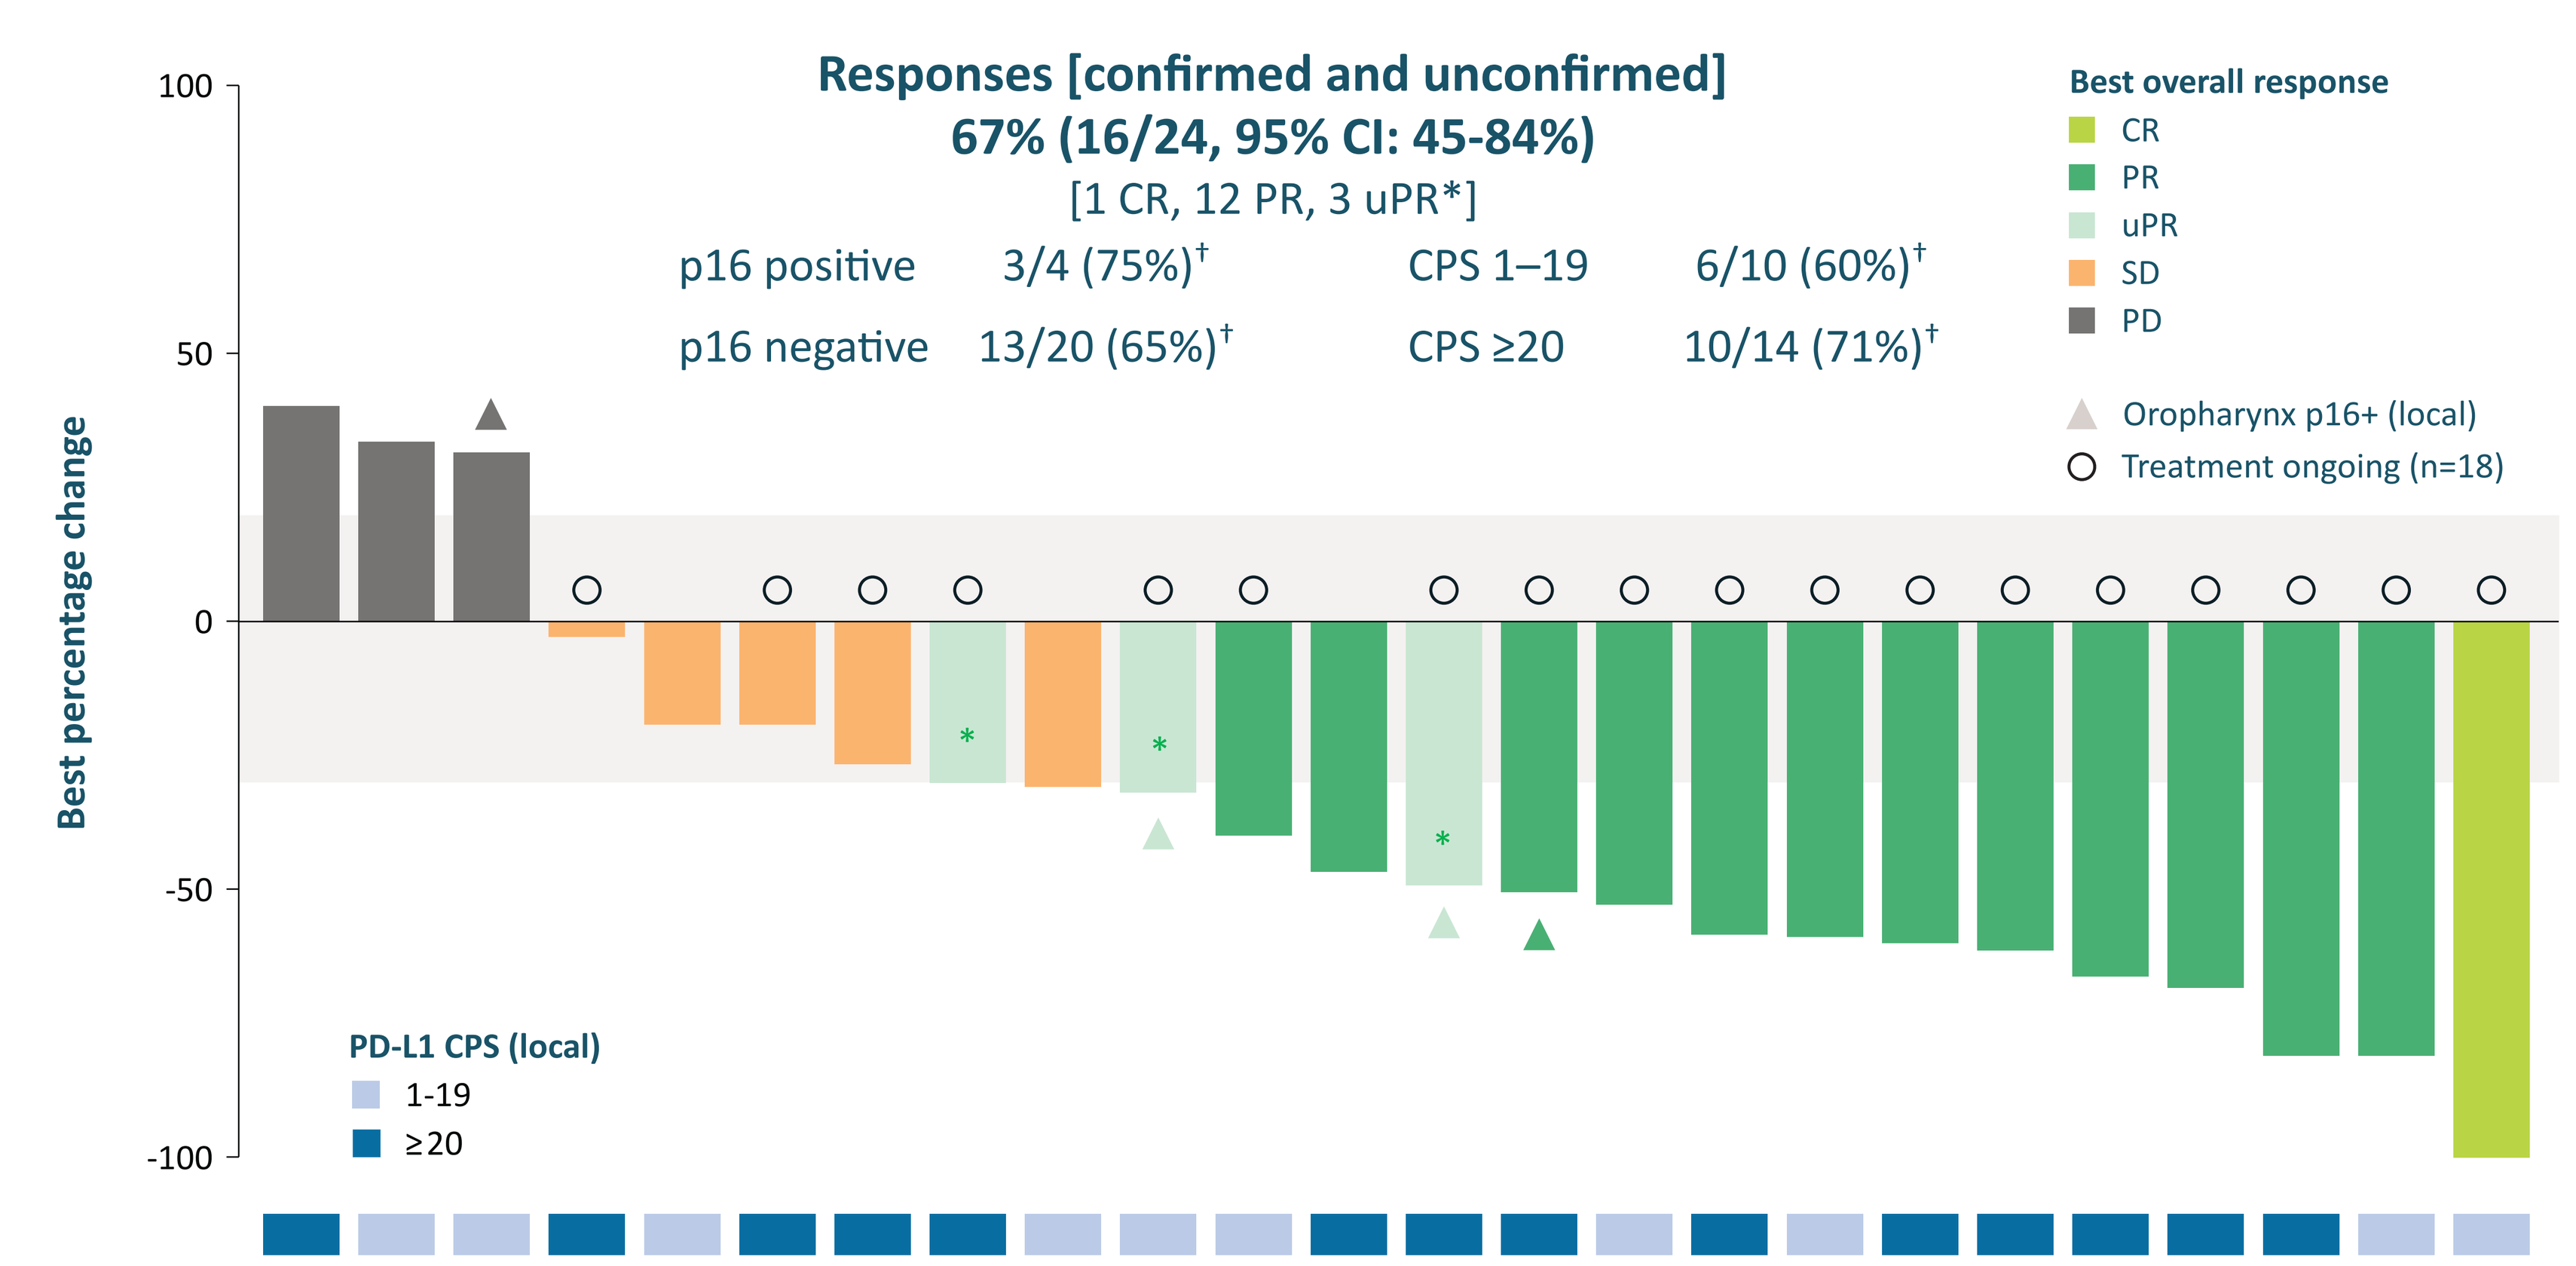

Responses [confirmed and unconfirmed]
67% (16/24, 95% CI: 45-84%)
[1 CR, 12 PR, 3 uPR*]
p16 positive 3/4 (75%)†
p16 negative 13/20 (65%)†
CPS 1–19 6/10 (60%)†
CPS ≥20 10/14 (71%)†
Best overall response
CR
PR
uPR
SD
PD
Oropharynx p16+ (local)
Treatment ongoing (n=18)
Best percentage change
100
50
0
-50
-100
PD-L1 CPS (local)
1-19
≥20

Figure 3 | Petosemtamab plus pembrolizumab in 1L r/m HNSCC: Phase 2 data (NCT03526835)

Petosemtamab plus pembrolizumab was observed to have a favorable safety profile including manageable infusion-related reactions[7]

Data cutoff date: 06 Mar 2024.
*All three uPRs were confirmed as PR after data cutoff date. †Response values for p16 and PD-L1 CPS subgroups include CR, PR, and uPR.
ASCO: American Society of Clinical Oncology; CI: confidence interval; CR: complete response; PR: partial response;
RECIST: Response Evaluation Criteria in Solid Tumors; SD: stable disease; uPR: unconfirmed partial response.

LiGeR-HN1 METHODS

Inclusion criteria

- Age ≥18 years at signing of ICF
- Histologically confirmed HNSCC with evidence of metastatic or locally recurrent disease not amenable to local therapy with curative intent
- HNSCC primary tumor locations are oropharynx, oral cavity, hypopharynx, and larynx; both HPV+ and HPV– are allowed
- Eligible to receive pembrolizumab as 1L monotherapy with tumors expressing PD-L1, CPS ≥1
- No previous systemic therapy administered in the incurable r/m setting
- Measurable disease as defined by RECIST v1.1 by radiologic methods
- ECOG PS of 0–1

Exclusion criteria

- CNS metastases that are untreated or already treated but symptomatic, or require radiation, surgery, or continued steroid therapy to control symptoms within 21 days of study entry
- Known leptomeningeal involvement
- Any systemic anticancer therapy within 4 weeks of the first dose of study treatment
- History of prior malignancies*
- Diagnosis of immunodeficiency or is receiving systemic steroid therapy or any form of immunosuppressive therapy within 7 days prior to the first dose
- Primary tumor site of nasopharynx, or sinonasal carcinoma (any histology)

Primary outcome measures

- Objective response rate per RECIST v1.1 as assessed by BICR
- Overall survival

Secondary outcome measures

- Progression-free survival per RECIST v1.1 as assessed by BICR
- Duration of response per RECIST v1.1 as assessed by BICR
- Safety of petosemtamab plus pembrolizumab (incidence and severity of AEs and SAEs)

*With the exception of excised local cancer, or treated cancer deemed at low risk for recurrence with no evidence of disease for ≥3 years.
AE: adverse event; BICR: blinded independent central review; CNS: central nervous system; HPV: human papillomavirus; ICF: informed consent form; SAE: serious AE.

References

1. Herpers B, et al. (2022) Nat Cancer *3*, 418–436.
2. Schinke H, et al. (2022) Mol Cancer *21*, 178.
3. Parsons MJ, et al. (2021) Cancer Discov *11*, 2413–2429.
4. Xu L, et al. (2019) Stem Cell Res Ther *10*, 219.
5. Katoh M. (2017) Int J Oncol *51*, 1357–1369.
6. Cohen E, et al. (2023) Cancer Res *83(Suppl 8)*, CT012 [oral, AACR 2023].
7. Fayette J, et al. (2024) J Clin Oncol *42(Suppl 16)*, Abstract 6014 [oral, ASCO 2024].
8. Sung H, et al. (2021) CA Cancer J Clin *71*, 209–249.
9. Harrington KJ, et al. (2023) J Clin Oncol *41*, 790–802.
10. Merck. (2024) Keytruda Prescribing Information. Available at: https://www.merck.com/product/usa/pi_circulars/k/keytruda/keytruda_pi.pdf (accessed 30 Aug 2024).
11. Wu Z, et al. (2017) Int J Clin Exp Pathol *10*, 11267–11275.
12. Dalley AJ, et al. (2015) Oral Surg Oral Med Oral Pathol Oral Radiol *119*, 436–440.
13. Byeon HK, et al. (2019) Exp Mol Med *51*, 1–14.
14. Xu MJ, et al. (2017) Cancer Metastasis Rev *36*, 463–473.

Acknowledgments

We thank the investigators and their teams; Merus colleagues; CROs and vendors; and Sophie Houten at LiNK Health Group.
Study sponsored by Merus NV.

Presenting Author Disclosures

JF is principal investigator for clinical trials sponsored by AstraZeneca, Calliditas, ISA, Merus, MSD, and Pfizer; and an advisory board consultant for AstraZeneca, Hookipa, Innate Pharma, MSD, Merck Serono, Pfizer, and Roche.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the European Society for Medical Oncology (ESMO®) Annual Meeting; September 13–17, 2024; Barcelona, Spain

ClinicalTrials.gov identifier: NCT06525220

Contact information: Shekeab Jauhari (s.jauhari@merus.nl)