Merus

Abstract 336

Mechanism of action of MCLA-129, a bispecific antibody that targets EGFR and c-MET and impairs growth of EGFR exon 20 insertion mutant non-small cell lung cancer

David JJ de Gorter, Marie N O'Connor, Alexandre Deshiere, Martijn van Rosmalen, Szabolcs Fatrai, Jeroen Lammerts van Bueren and Cecile AW Geuijen

Merus N.V., Utrecht, The Netherlands

INTRODUCTION

- EGFR and c-MET activate the same intracellular signal transduction pathways to drive proliferation, survival and invasion
- MET/HGF amplification plays a potential role in resistance to EGFR targeted treatment in solid tumors[1]
- In non-small cell lung cancer (NSCLC), resistance to TKI inhibition of mutant EGFR is associated with increased c-MET signalling[2]

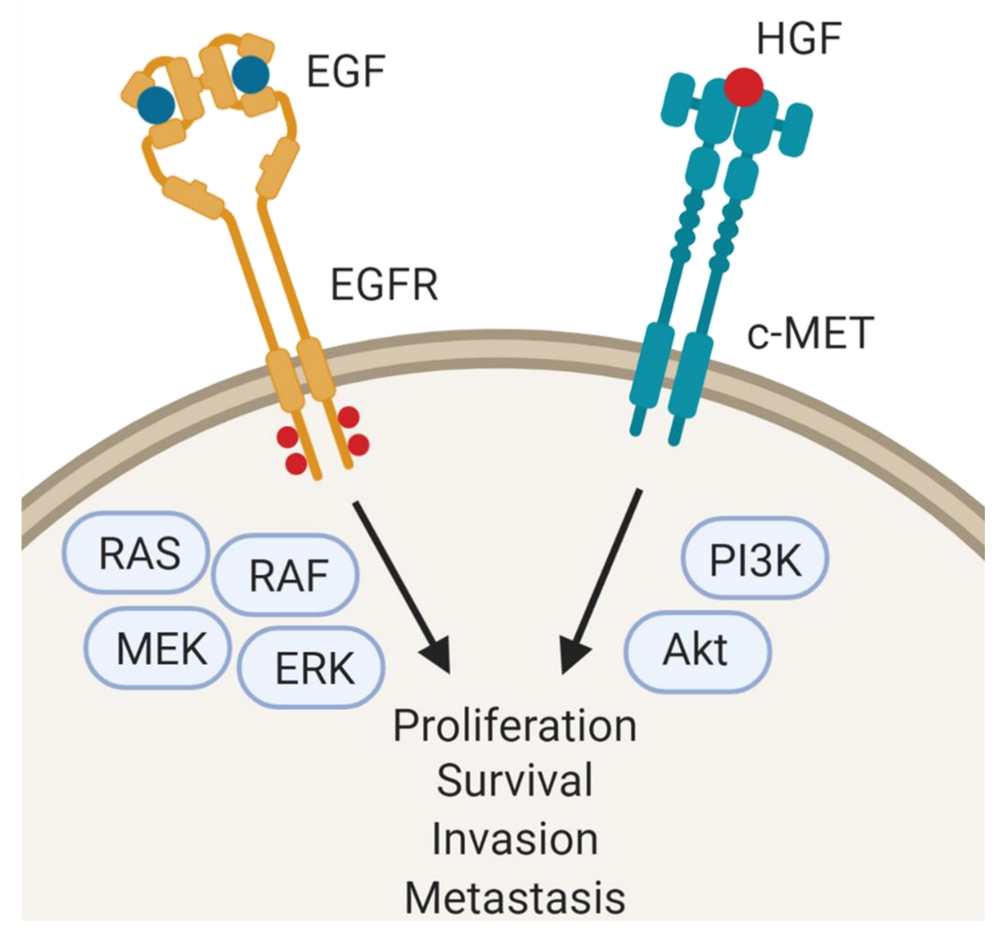
EGF
HGF
EGFR
c-MET
RAS
RAF
MEK
ERK
PI3K
Akt
Proliferation
Survival
Invasion
Metastasis

THE MCLA-129 BICLONICS®

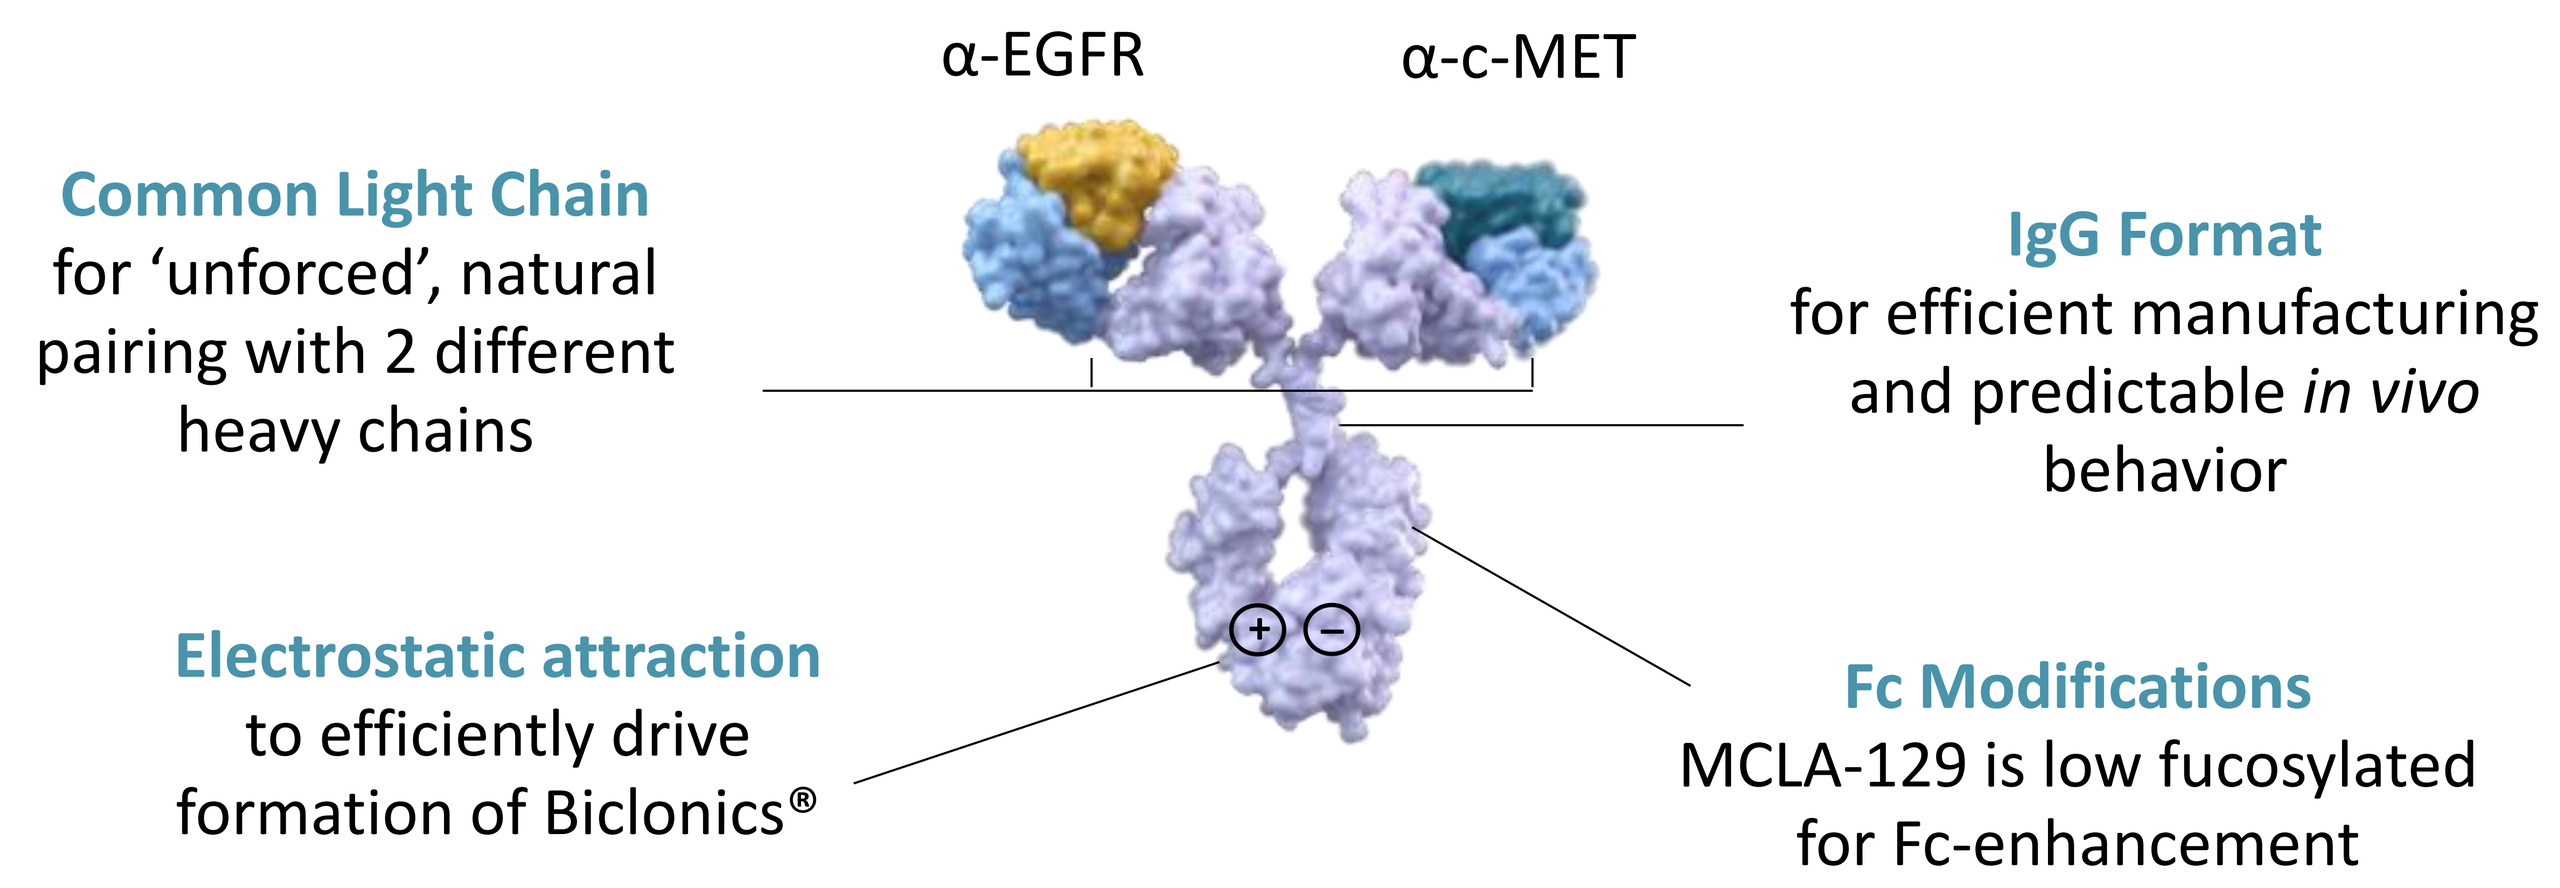
α-EGFR
α-c-MET
Common Light Chain
for 'unforced', natural pairing with 2 different heavy chains
IgG Format
for efficient manufacturing and predictable *in vivo* behavior
Electrostatic attraction
to efficiently drive formation of Biclonics®
Fc Modifications
MCLA-129 is low fucosylated for Fc-enhancement

EPITOPE FOR MCLA-129 OVERLAPS WITH EGF BINDING SITE

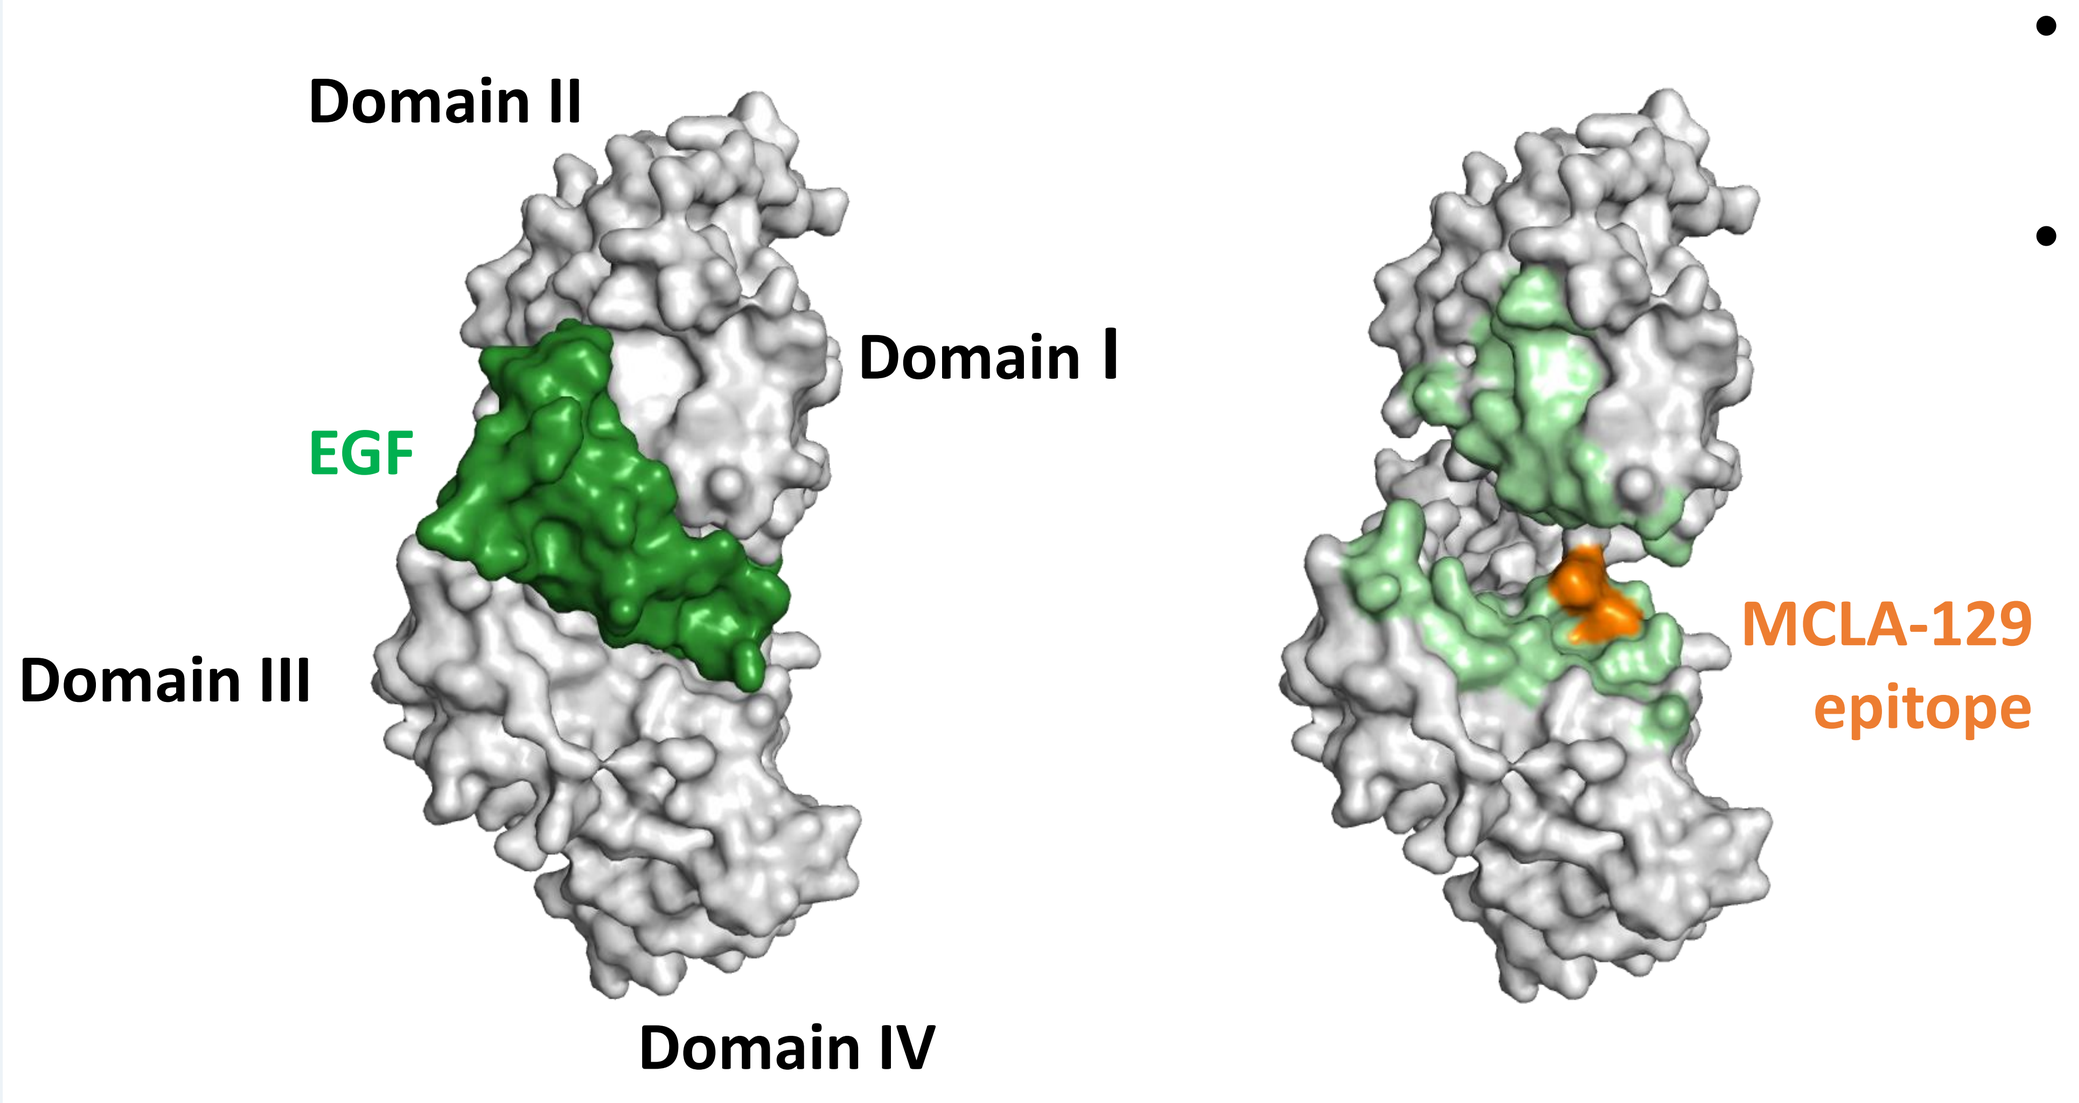
Domain II
Domain I
EGF
Domain III
Domain IV
MCLA-129 epitope

- The MCLA-129 epitope is located on EGFR domain III
- The EGFR residues critical for MCLA-129 binding overlap with the EGF-binding site

Figure 1 | The MCLA-129 epitope identified by shotgun mutagenesis epitope mapping. EGF (green) binds EGFR domain I and III (left). The EGF footprint (light green) on the surface of EGFR (grey) highlights all atoms within 4 Å of bound EGF. The residues critical for MCLA-129 binding are indicated in orange (right). (Crystal structure reference PDB ID: 1IVO).

MCLA-129 INHIBITS EGFR AND c-MET DIMERIZATION

- MCLA-129 inhibits EGF and HGF ligand-induced dimerization of the receptors EGFR and c-MET
- Reference antibody was the anti-RSV IgG negative control

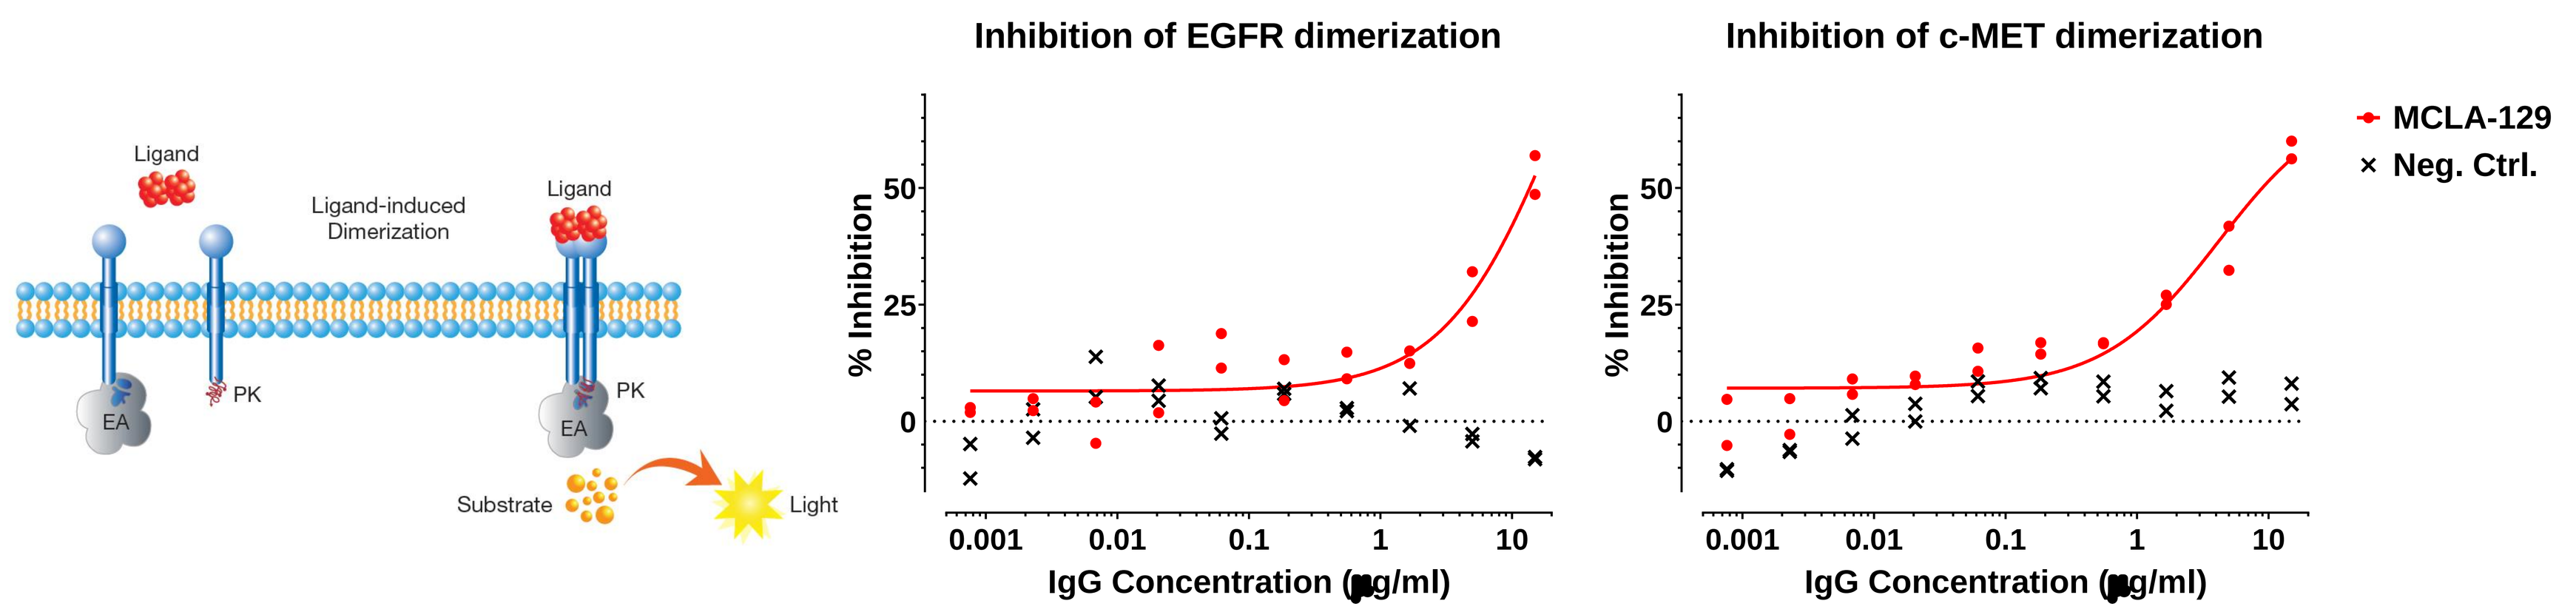
Ligand
Ligand-induced Dimerization
PK
EA
Substrate
Light
Inhibition of EGFR dimerization
Inhibition of c-MET dimerization
% Inhibition
IgG Concentration (µg/ml)
MCLA-129
Neg. Ctrl.

Figure 2 | β-galactosidase enzyme complementation assay to determine the inhibitory effect of MCLA-129 on ligand-induced EGFR and c-MET receptor dimerization. Left, on reporter cell lines ligand binding induces dimerization of chimeric receptors and the formation of a fully reconstituted active β-galactosidase enzyme. Enzyme activity is detected by chemiluminescence. Middle and right panel, inhibitory effect of MCLA-129 on EGF (13 ng/ml) induced EGFR and HGF (190 ng/ml) induced c-MET dimerization in the presence of MCLA-129 or negative control antibody.

MCLA-129 INHIBITS EGFR AND c-MET PHOSPHORYLATION

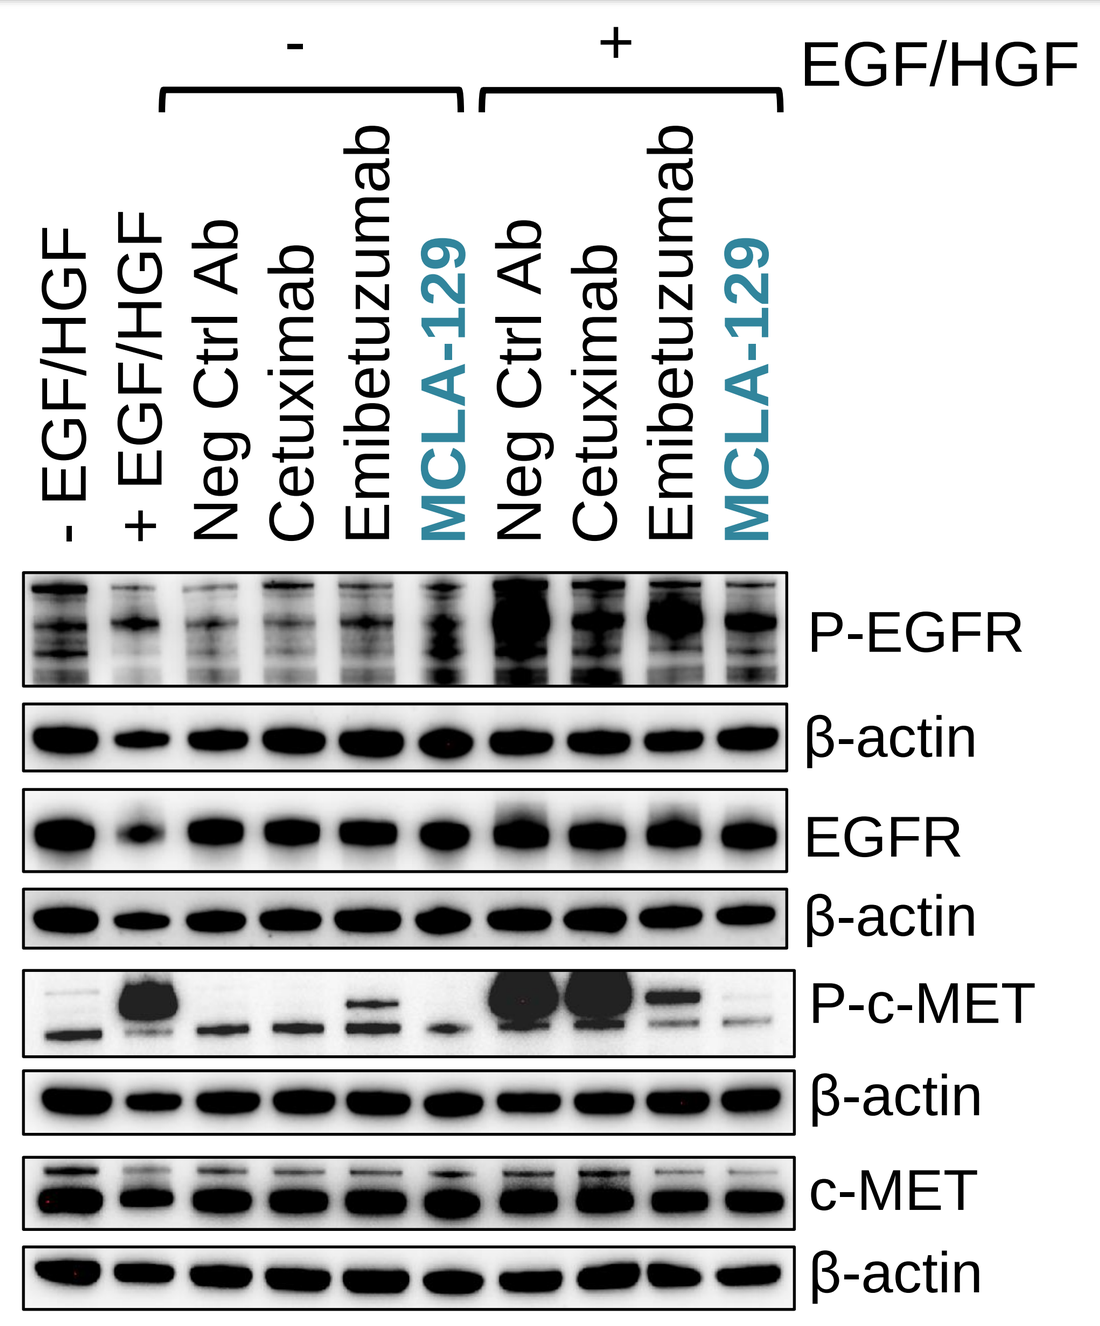
EGF/HGF
- EGF/HGF
+ EGF/HGF
Neg Ctrl Ab
Cetuximab
Emibetuzumab
MCLA-129
P-EGFR
β-actin
EGFR
P-c-MET
c-MET

- Ligand-dependent phosphorylation of EGFR and c-MET was assessed *in vitro* in NCI-H1650 (*EGFR* $^{del\ E746\text{-}A750}$, *EGFR* ^{amplification}) NSCLC cells
- MCLA-129 inhibits phosphorylation of EGFR and c-MET

Figure 3 | Ligand-dependent phosphorylation of EGFR and c-MET assessed by immunoblotting. Serum-starved NCI-H1650 were incubated with EGF (50 ng/ml) and HGF 100 ng/ml) in the presence of MCLA-129, negative control antibody and reference antibodies anti-EGFR cetuximab and anti-c-MET Emibetuzumab analog (each 10 µg/ml).

MCLA-129 PROMOTES ADCC AND ADCP OF NSCLC CELLS

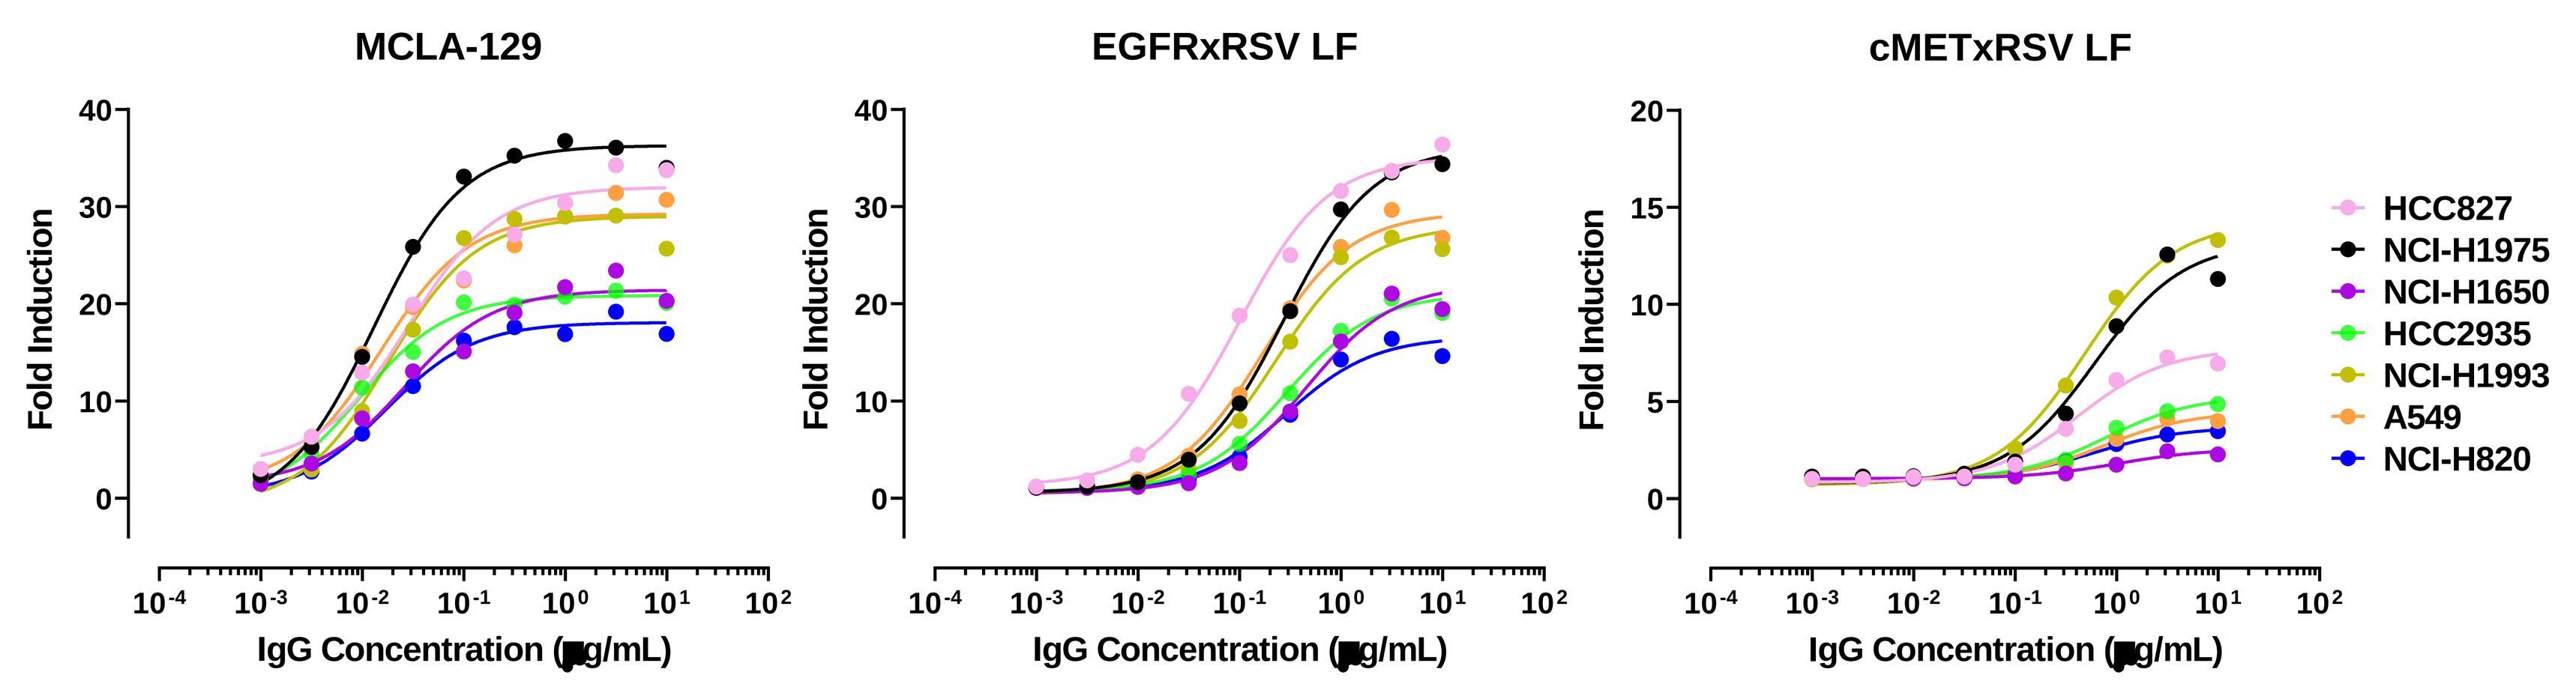
MCLA-129
EGFRxRSV LF
cMETxRSV LF
Fold Induction
IgG Concentration (µg/mL)
HCC827
NCI-H1975
NCI-H1650
HCC2935
NCI-H1993
A549
NCI-H820

Figure 4 | ADCC induced by MCLA-129 and monovalent c-MET and EGFR binding antibodies having the same Fab domains as MCLA-129 against NSCLC cell lines using high affinity FcγRIIIa 158V-variant effector cells.

- NSCLC cell lines varying in their EGFR and c-MET expression levels and EGFR mutational status representing several patient populations were tested
- Both the EGFR and the c-MET Fab arms contribute to Antibody-Dependent Cell-Mediated Cytotoxicity (ADCC) enhanced by avidity binding
- MCLA-129 displays ADCC towards all NSCLC cell lines tested
- MCLA-129 induces ADCC using effector cells expressing either high or low affinity FcγRIIIa variant

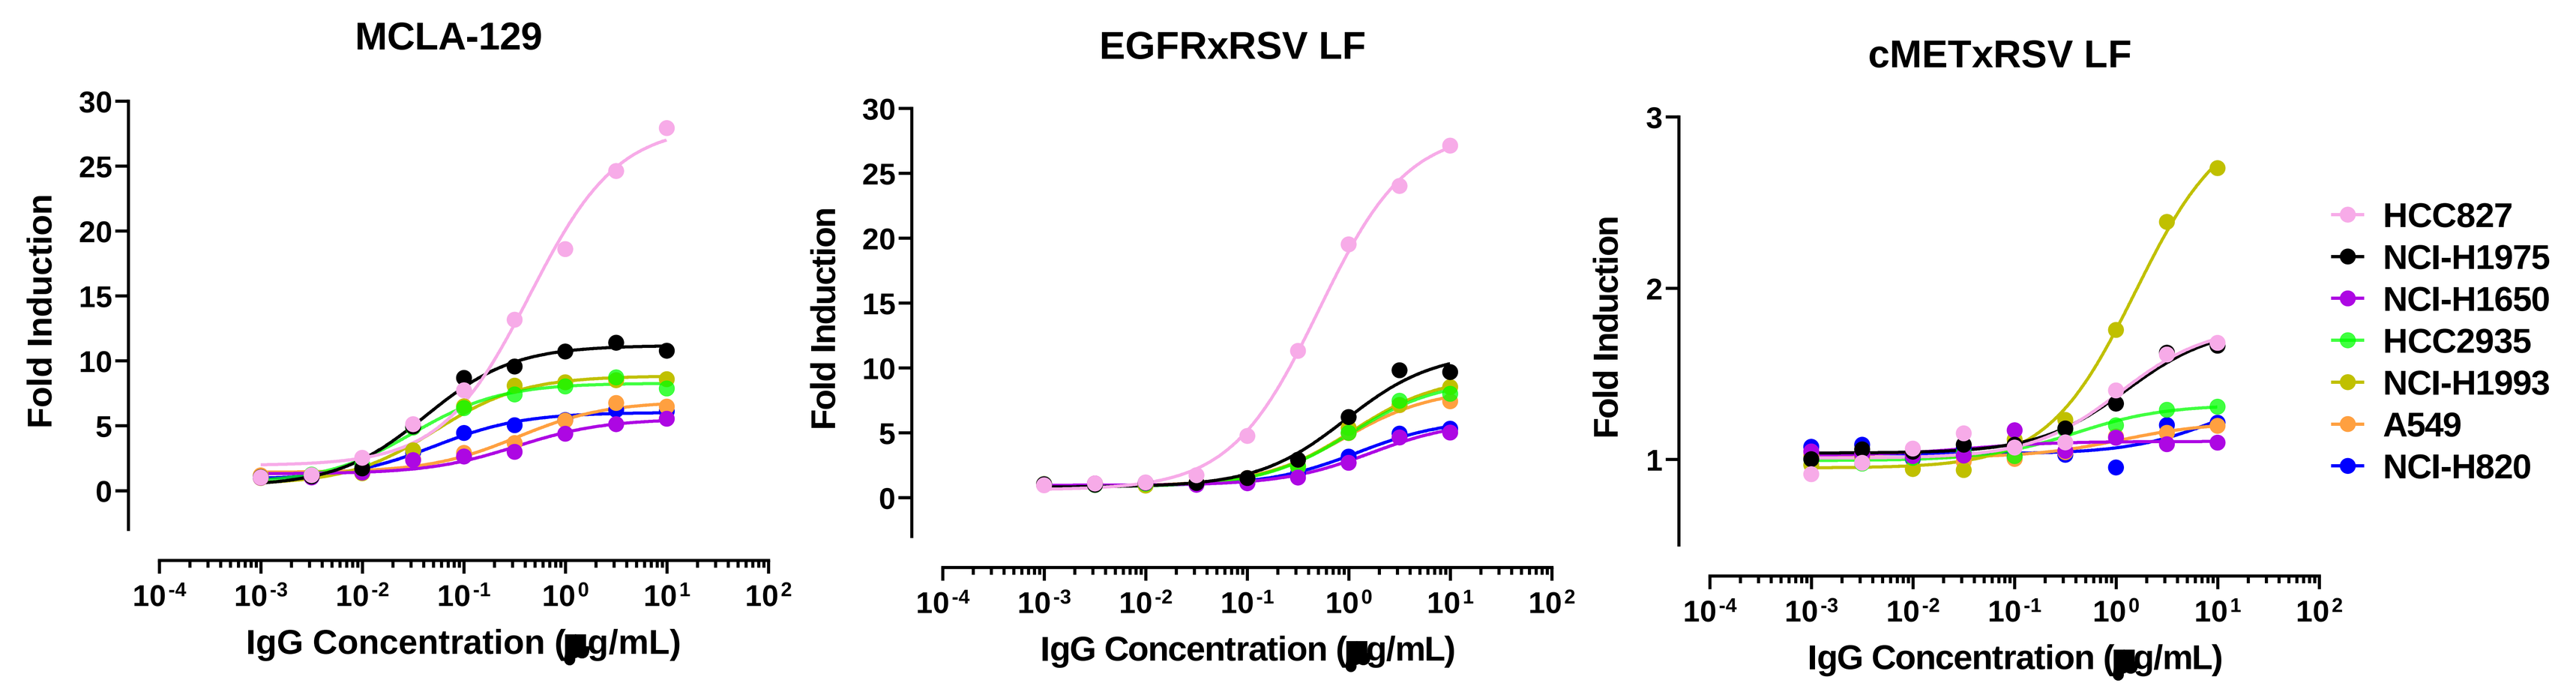
MCLA-129
EGFRxRSV LF
cMETxRSV LF
Fold Induction
IgG Concentration (µg/mL)
HCC827
NCI-H1975
NCI-H1650
HCC2935
NCI-H1993
A549
NCI-H820

Figure 5 | ADCC induced by MCLA-129 and monovalent c-MET and EGFR binding antibodies having the same Fab domains as MCLA-129 against NSCLC cell lines using low affinity FcγRIIIa 158F-variant effector cells.

- MCLA-129 induces Antibody-Dependent Cell-Mediated Phagocytosis (ADCP) of NSCLC cells

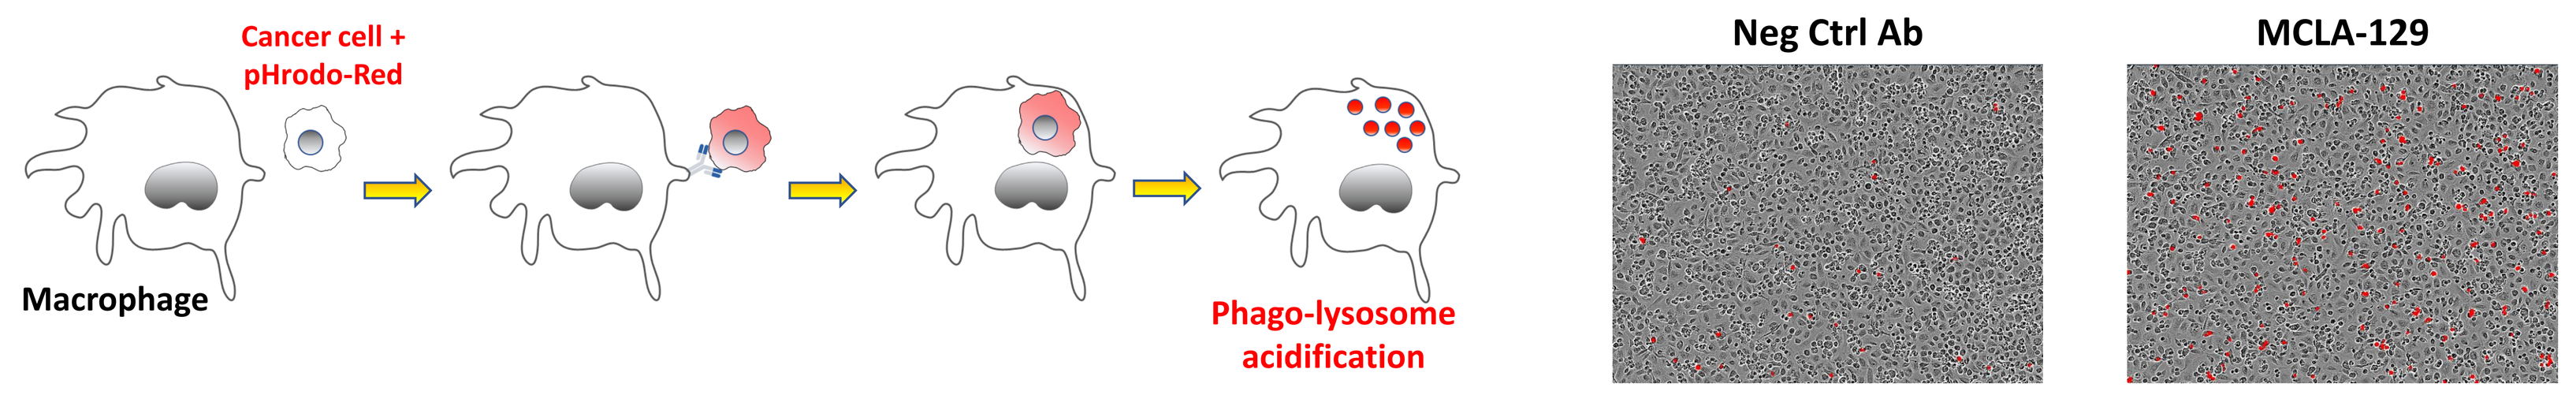
Cancer cell + pHrodo-Red
Macrophage
Phago-lysosome acidification
Neg Ctrl Ab
MCLA-129

Figure 6 | ADCP induced by MCLA-129 against NSCLC cell line HCC827. pH-sensitive pHrodo dye labelled HCC827 target cells were incubated with MCLA-129 or negative control antibody and primary macrophage effector cells. ADCP was assessed on the Incucyte® system (pHrodo-Red fluorescence > 25 RCU).

MCLA-129 INHIBITS EGFR EXON20INS PDX TUMOR GROWTH

- Patient-derived NSCLC LXFE2478 xenograft with EGFR exon 20 insertion
- Insertion of 9 nucleotides affecting the EGFR tyrosine kinase domain (M766_A767insASV)
- Implanted in nude mice having normal activity of the Fc receptor-expressing NK and myeloid cells
- MCLA-129 treatment leads to dose-dependent tumor shrinkage

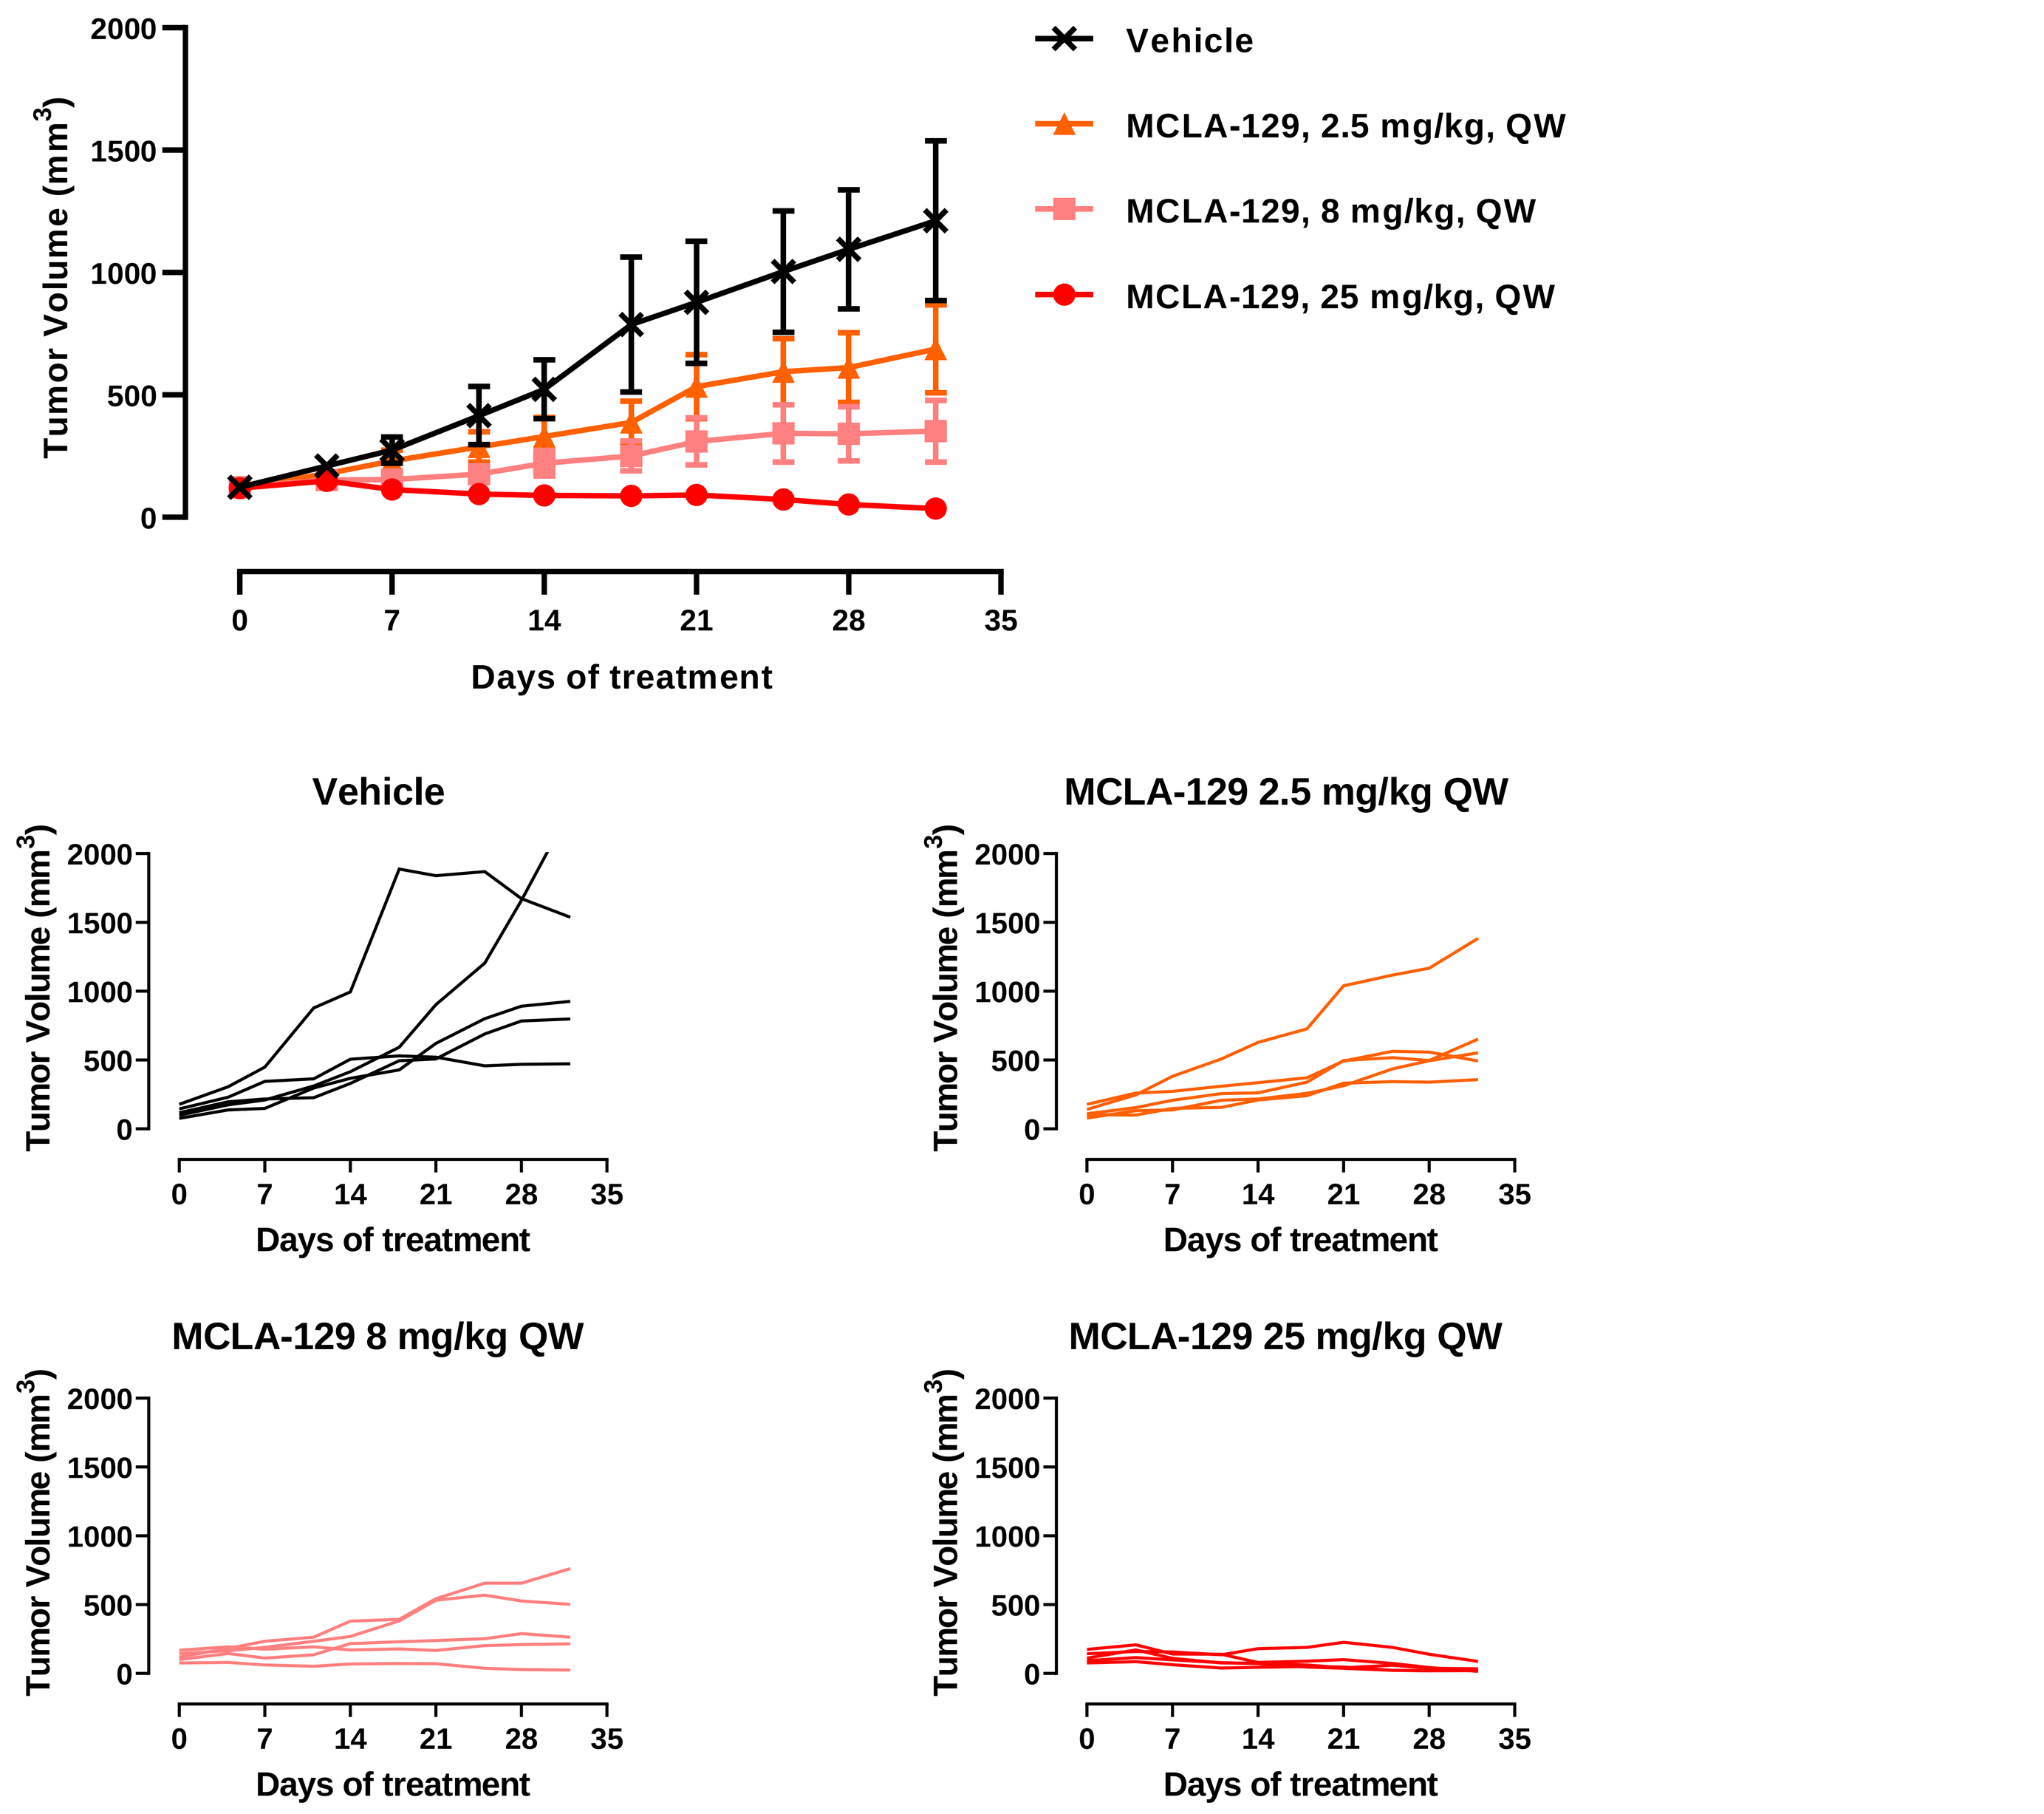
Vehicle
MCLA-129, 2.5 mg/kg, QW
MCLA-129, 8 mg/kg, QW
MCLA-129, 25 mg/kg, QW
Tumor Volume (mm³)
Days of treatment
MCLA-129 2.5 mg/kg QW
MCLA-129 8 mg/kg QW
MCLA-129 25 mg/kg QW

Figure 7 | Activity of MCLA-129 on *EGFR exon20ins* PDX tumor growth in ADCC-competent mice. *EGFR exon20ins* PDX engrafted into nude mice. MCLA-129 was administered at 2.5, 8 or 25 mg/kg i.p. weekly to mice with established tumors. Upper panel, mean tumor volume ± SEM is shown. Lower panels, individual tumor volume is shown.

CONCLUSIONS

- MCLA-129 is a Fc-enhanced common light chain bispecific human IgG1 Biclonics® antibody specifically targeting the receptor tyrosine kinases EGFR and c-MET
- The epitope for MCLA-129 overlaps with EGF binding site
- MCLA-129 inhibits ligand-induced EGFR and c-MET receptor dimerization and phosphorylation
- MCLA-129 promotes Antibody-Dependent Cell-Mediated Cytotoxicity (ADCC) and Antibody-Dependent Cell-Mediated Phagocytosis (ADCP) of NSCLC cells
- MCLA-129 significantly inhibits growth of a patient-derived EGFR exon20ins tumor in a preclinical xenograft model
- These data provide support for the ongoing phase 1/2 study of MCLA-129 in patients with solid tumors, including NSCLC with EGFR exon20ins (Study MCLA-129-CL01, NCT04868877)

Contact Information

David de Gorter
T +31 85 016 2500
E d.degorter@merus.nl

References

1. Tsuji et al. Oncotarget. 2017;8(42): 71805-71816
2. Engelman et al. Science. 2007;316(5827): 1039-1043