136P

Presented at the

2021 European Society for Medical Oncology – Immuno-Oncology (ESMO-IO) Congress

Geneva, Switzerland • December 8–11, 2021

Phase I Dose Escalation Study of MCLA-145, a Bispecific Antibody Targeting CD137 and PD-L1 in Solid Tumors

Hans Prenen,[1*] Chrisann Kyi,[2] Griet Van Lancker,[3] Sandip Pravin Patel,[4] Diana Mittag,[5] Alison Weaver,[5] Kees Bol,[5] Vika Stalbovskaya,[5] Jennifer Pulini,[6] Gongfu Zhou,[6] Zhiwan Dong,[6] Ekaterine Asatiani,[7] F. Stephen Hodi[8]

[1]Antwerp University Hospital, Antwerp, Belgium; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]Ghent University Hospital, Ghent, Belgium; [4]UC San Diego, La Jolla, CA, USA; [5]Merus NV, Utrecht, Netherlands; [6]Incyte Corporation, Wilmington, DE, USA; [7]Incyte Biosciences International Sàrl, Morges, Switzerland; [8]Dana-Farber Cancer Institute, Harvard Cancer Center, Boston, MA, USA

* Presenting author (Hans.Prenen@uza.be)

Introduction

- MCLA-145 is a fully human common light chain IgG1-based bispecific antibody that binds CD137 and PD-L1[1]
 - MCLA-145 was Fc silenced to block nonspecific Fcγ receptor-mediated effects
- Preclinically, MCLA-145 induces CD137 signaling in the presence of PD-L1 and correlates with PD-L1 expression levels[1,2]
- Targeting CD137 and the PD-1/PD-L1 axis with MCLA-145 enhances antigen-mediated T-cell activation via CD137 costimulation and blocking inhibitory PD-L1 (**Figure 1**)[1]
- A bell-shaped dose-response curve was observed for MCLA-145 *in vitro* (**Figure 2**)
- MCLA-145 was shown to promote immunologic memory in *in vitro* T-cell priming assays,[1] consistent with literature demonstrating that CD137 costimulation promotes durable CD8 T-cell responses[3,4]
- In preclinical xenograft models, MCLA-145 demonstrated antitumor activity, with enrichment of CD8+ T cells in tumors following treatment[1]
- MCLA-145-CL01/MCLA-145-101 is a phase I multicenter trial investigating MCLA-145 in different solid tumors considered not amenable to surgery or other curative treatments or procedures
 - The trial includes dose escalation and expansion parts

Figure 1. MCLA-145 Mechanism of Action

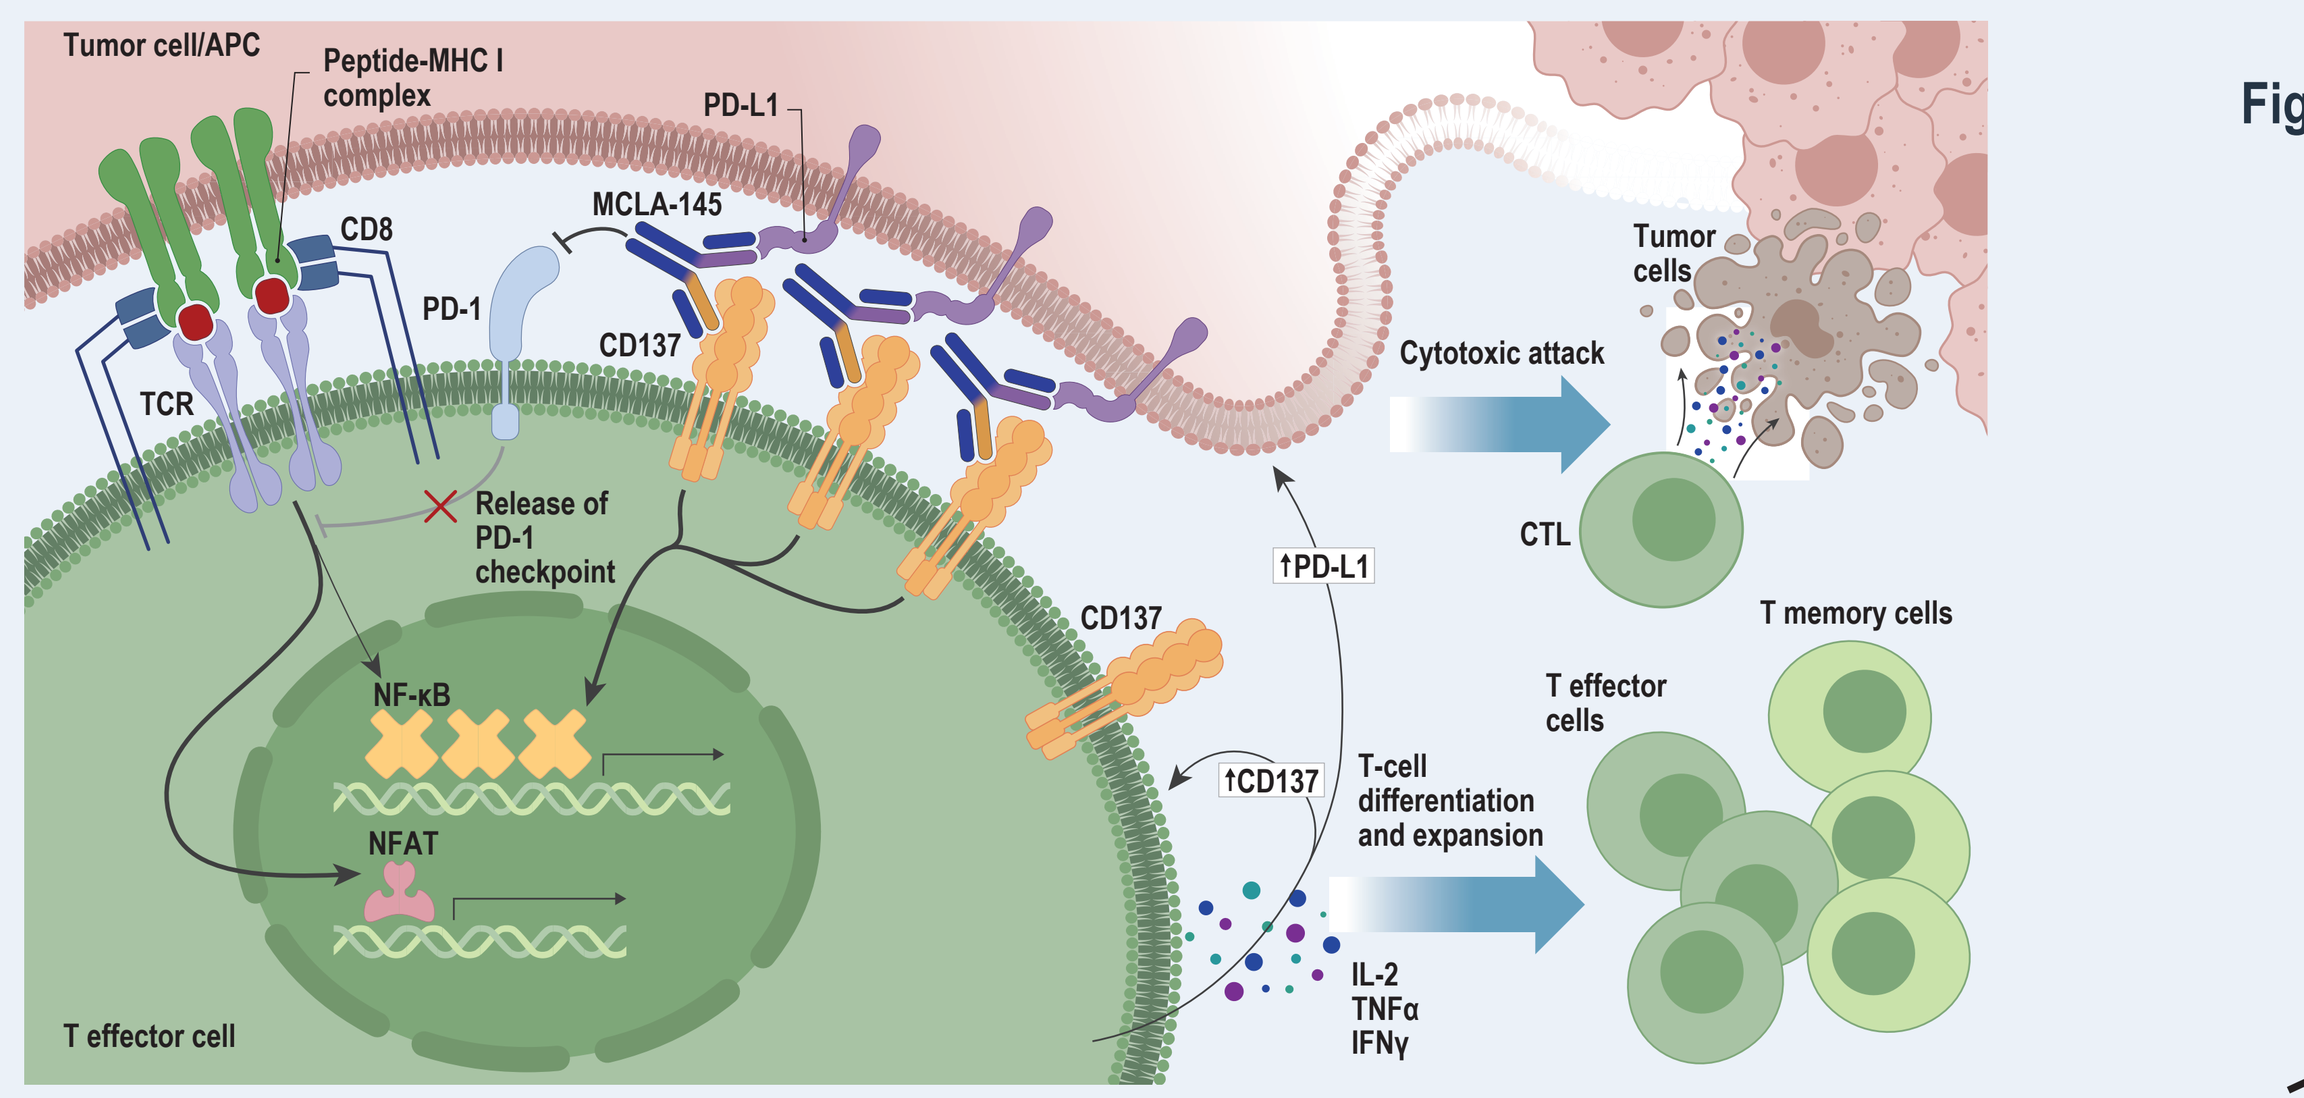

Tumor cell/APC
Peptide-MHC I complex
PD-L1
CD8
MCLA-145
PD-1
CD137
TCR
Release of PD-1 checkpoint
NF-κB
NFAT
T effector cell
CD137
↑PD-L1
↑CD137
Cytotoxic attack
Tumor cells
CTL
T memory cells
T effector cells
T-cell differentiation and expansion
IL-2
TNFα
IFNγ

Figure 2. Complex Formation by Flow Cytometry Demonstrated a Bell-Shaped Dose Response

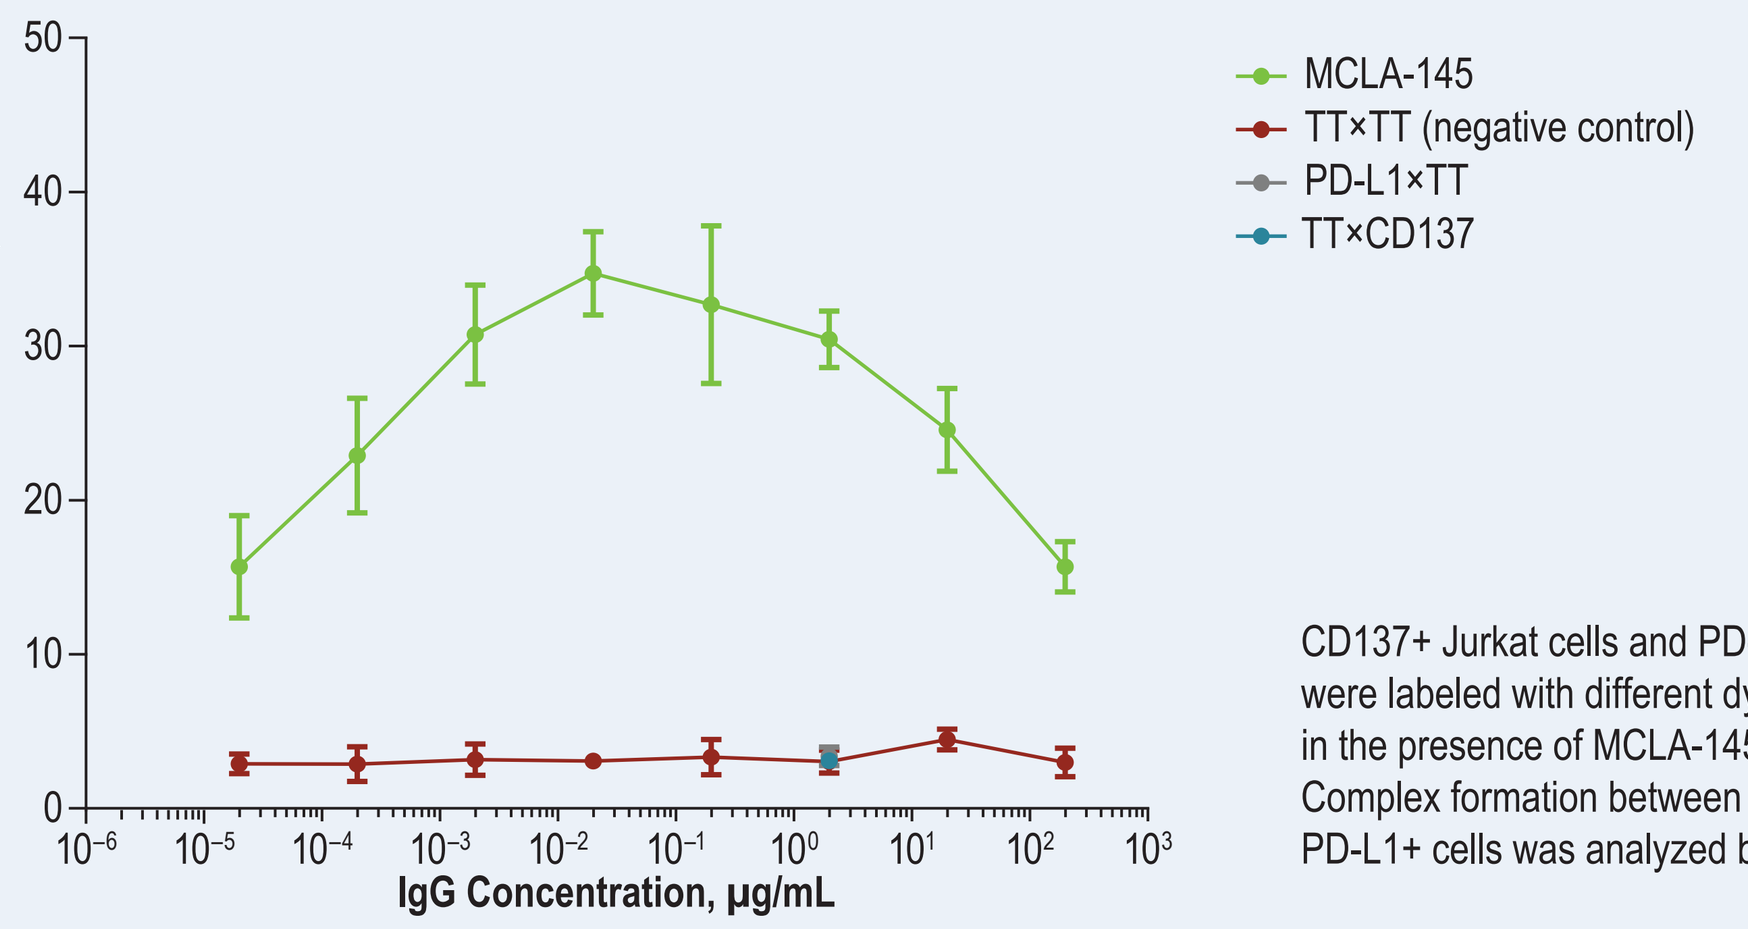

Double-Positive Events, %
IgG Concentration, μg/mL
MCLA-145
TT×TT (negative control)
PD-L1×TT
TT×CD137

CD137+ Jurkat cells and PD-L1+ CHO cells were labeled with different dyes and co-cultured in the presence of MCLA-145 or controls. Complex formation between CD137+ and PD-L1+ cells was analyzed by flow cytometry.

Purpose

- Evaluation of the safety and preliminary efficacy of MCLA-145 in advanced or metastatic solid tumors

Methods

Patients and Study Design

- This open-label phase I study (NCT03922204) is currently enrolling patients in the dose escalation part
- Adult patients (age ≥18 years) with histologically or cytologically confirmed advanced or recurrent/metastatic solid tumors known to respond to immunotherapy (limit of 1 prior anti–PD-1 immunotherapy regimen allowed), Eastern Cooperative Oncology Group (ECOG) status of 0–1, ≤4 prior systemic treatment regimens are eligible
- Key exclusion criteria are prior therapy with a CD137 (4-1BB) agonist, prior chimeric antigen receptor (CAR) T-cell therapy, prior grade ≥3 immune-mediated adverse events (AEs) with anti–PD-1 therapy, or prior anti–PD-1 therapy-associated liver toxicity of grade >1
- MCLA-145 is being administered intravenously (range, 0.4–75 mg) every 14 days (Q2W) in 28-day cycles; the study includes dose escalation and confirmation/safety expansion

Objectives

- The primary objective is to evaluate the safety, tolerability, and dose-limiting toxicity (DLT) of MCLA-145 and to determine maximum tolerated dose (MTD) and/or recommended dose for expansion (RDE)
- Secondary objectives include the following:
 - Evaluation of pharmacokinetics
 - Exploration of preliminary antitumor activity through assessment of objective response rate (ORR; complete response [CR] + partial response [PR]), disease control rate (DCR; CR + PR + stable disease [SD]), progression-free survival (PFS), and duration of response (DOR) per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1
- Exploratory objectives include evaluating the effect of MCLA-145 on tumor and immune cell infiltrate biomarkers as predictive markers of MCLA-145 activity

Statistical Analyses

- Dose escalation was guided by an adaptive Bayesian logistic regression model (BLRM) with overdose control
- Data are analyzed using descriptive statistics

Results

Dosing and Baseline Patient Characteristics

- As of 14 July 2021, 34 patients were enrolled and treated (**Figure 3**); median (range) age was 60.5 (27–81) years, and 55.9% were male (**Table 1**)

Figure 3. Dose Escalation

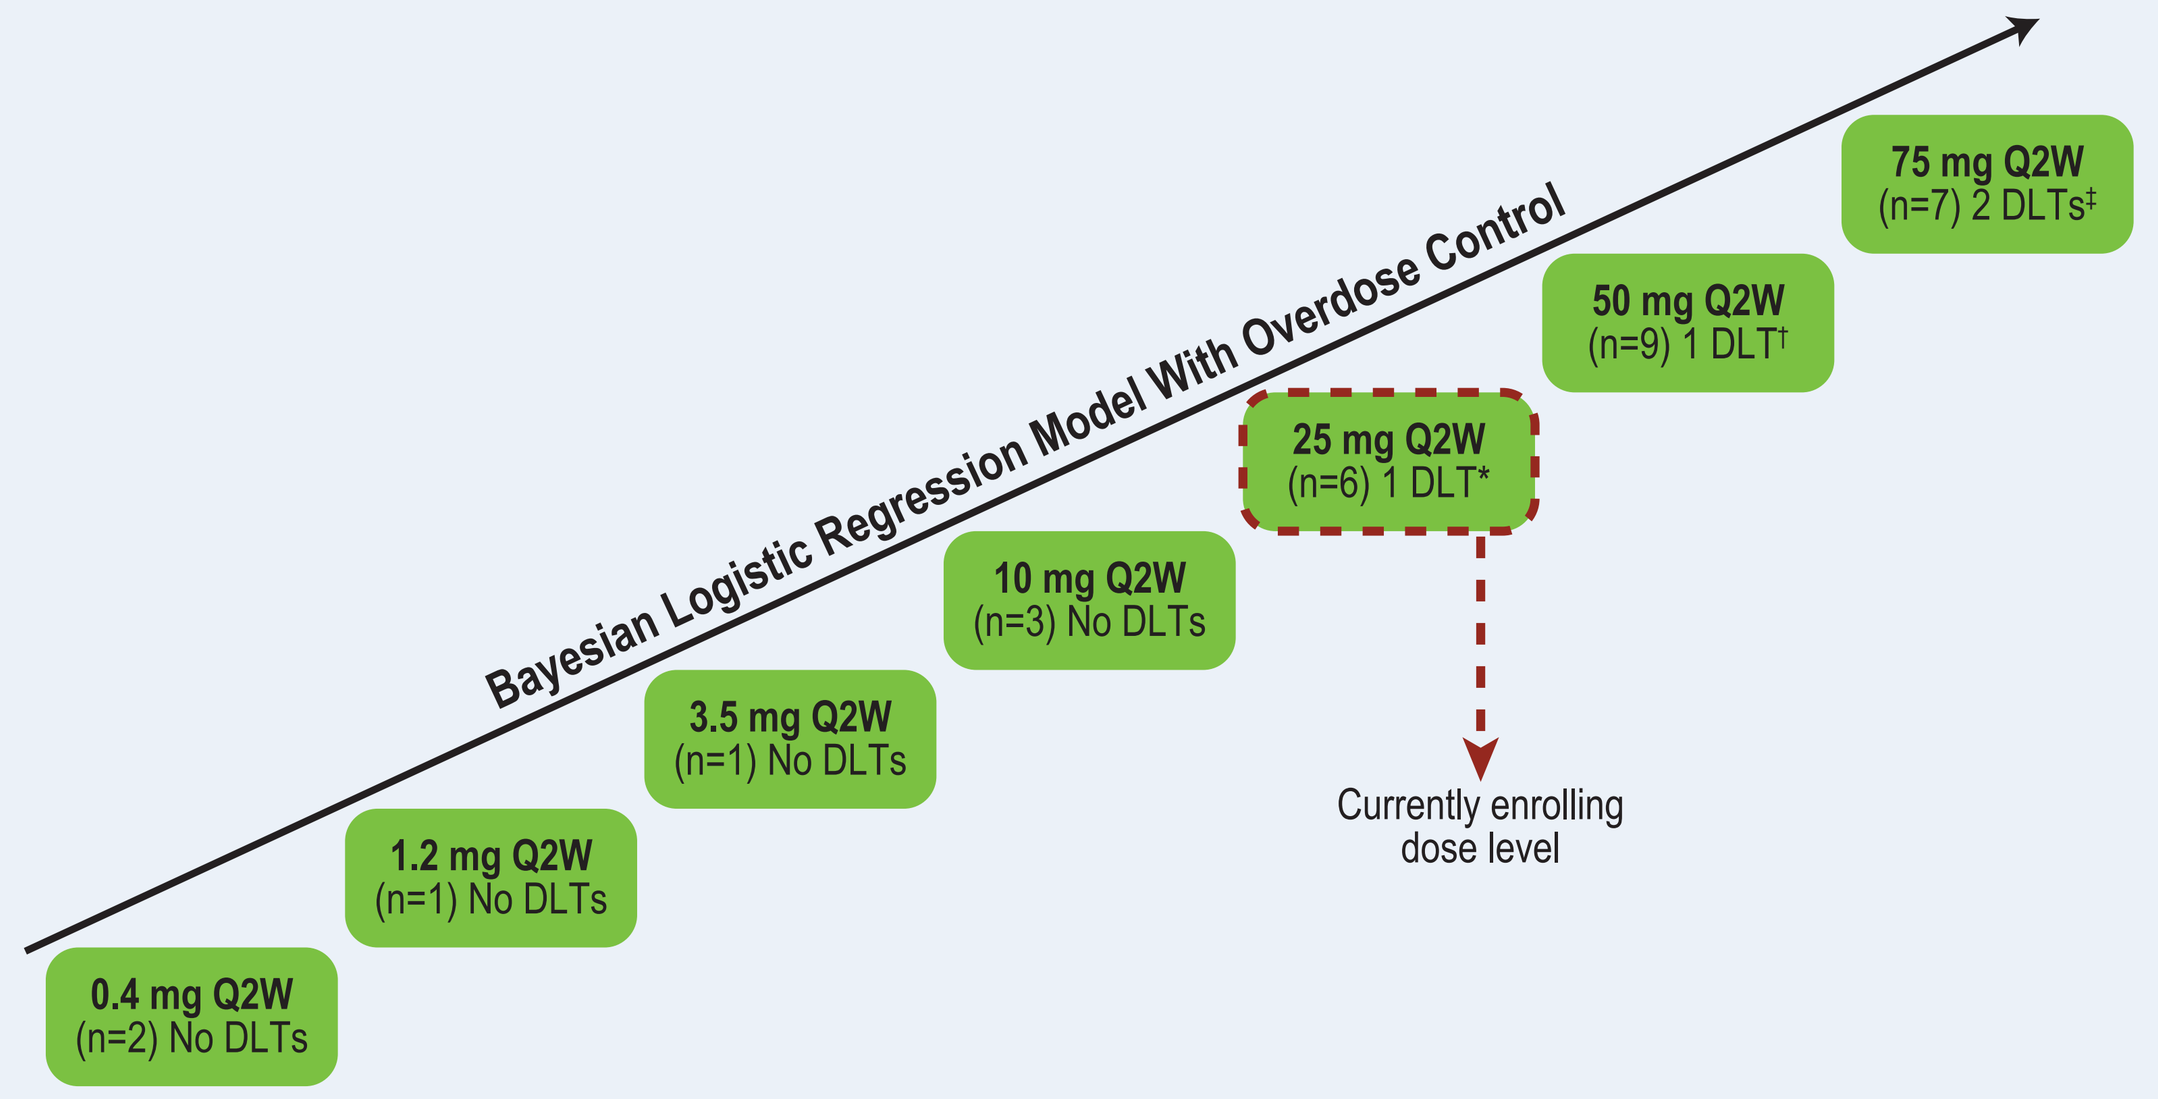

Bayesian Logistic Regression Model With Overdose Control
0.4 mg Q2W (n=2) No DLTs
1.2 mg Q2W (n=1) No DLTs
3.5 mg Q2W (n=1) No DLTs
10 mg Q2W (n=3) No DLTs
25 mg Q2W (n=6) 1 DLT*
50 mg Q2W (n=9) 1 DLT†
75 mg Q2W (n=7) 2 DLTs‡
Currently enrolling dose level

ALT, alanine aminotransferase; AST, aspartate aminotransferase; DLT, dose-limiting toxicity (DLT period is defined as 28 days from the first infusion); Q2W, every 14 days.
Intermediate dose levels from 25 to 75 mg have been explored in 5 additional patients not included in the figure above.
* Febrile neutropenia. † Myositis. ‡ ALT/AST increased, febrile neutropenia.

Table 1. Baseline Patient Demographics and Clinical Characteristics (Full Analysis Set)

Characteristic	Total (N=34)
Age, median (range), y	60.5 (27–81)
Male, n (%)	19 (55.9)
White, n (%)	30 (88.2)
ECOG status, n (%)	
0	16 (47.1)
1	18 (52.9)
Time since initial diagnosis, median (range), y	2.2 (0.2–10.1)
Most common tumor types,*† n (%)	
Colorectal cancer	5 (14.7)
Lung cancer	4 (11.8)
Renal cancer	4 (11.8)
Glioblastoma	4 (11.8)
Pancreatic cancer	3 (8.8)
Previous lines of therapy, n (%)	
0	0
1	5 (14.7)
2	10 (29.4)
≥3	19 (55.9)
Previous immunotherapy, n (%)	17 (50.0)
PD-L1 expression on tumor cells, n (%)	
0%	11 (32.4)
≥1%	7 (20.6)
Unknown / not evaluable	16 (47.1)
PD-L1 expression on tumor-associated immune cells, n (%)	
0%	4 (11.8)
≥1%	14 (41.2)
Unknown / not evaluable	16 (47.1)

ECOG, Eastern Cooperative Oncology Group; PD-L1, programmed cell death ligand 1.
* Occurring in >2 patients.
† Tumor type in n=2 patients was head and neck cancer.

Safety

- Median (range) duration of treatment with MCLA-145 was 6 (1–74) weeks
- Treatment-emergent AEs (TEAEs) occurred in 33 patients (97.1%); treatment-related TEAEs occurred in 23 patients (67.6%), most commonly fatigue (n=6, 17.6%) and decreased neutrophil count (n=6, 17.6%; **Table 2**)
- DLTs occurred in 4 patients (11.8%) and were managed as follows:
 - 25 mg: grade 3 febrile neutropenia (responsive to filgrastim)
 - 50 mg: grade 2 myositis (responsive to steroid treatment)
 - 75 mg: grade 3 febrile neutropenia (resolved without growth factor support)
 - 75 mg: grade 3 alanine aminotransferase (ALT) and aspartate aminotransferase (AST) increase (responsive to steroid treatment)

Table 2. Summary of Overall and Most Common* TEAEs

	Any TEAE (N=34)		Treatment-Related TEAE (N=34)	
Characteristic, n (%)	Any Grade	Grade ≥3	Any Grade	Grade ≥3
Any TEAE	33 (97.1)	22 (64.7)	23 (67.6)	10 (29.4)
Serious TEAE	16 (47.1)	14 (41.2)	4 (11.8)	4 (11.8)
Fatal TEAE	3 (8.8)	3 (8.8)	0	0
Most common TEAEs*				
Fatigue	14 (41.2)	0	6 (17.6)	0
Decreased appetite	11 (32.4)	1 (2.9)	2 (5.9)	0
Dyspnea	9 (26.5)	0	2 (5.9)	0
Nausea	7 (20.6)	1 (2.9)	1 (2.9)	0
Decreased neutrophil count	7 (20.6)	6 (17.6)	6 (17.6)	5 (14.7)
Diarrhea	6 (17.6)	0	1 (2.9)	0
Productive cough	6 (17.6)	0	0	0
Vomiting	6 (17.6)	2 (5.9)	2 (5.9)	0
Decreased lymphocyte count	5 (14.7)	5 (14.7)	4 (11.8)	4 (11.8)
Myalgia	5 (14.7)	0	3 (8.8)	0
Pyrexia	5 (14.7)	0	2 (5.9)	0
ALT increased	4 (11.8)	3 (8.8)	3 (8.8)	2 (5.9)
Arthralgia	4 (11.8)	0	0	0
Headache	4 (11.8)	1 (2.9)	1 (2.9)	0
Non-cardiac chest pain	4 (11.8)	1 (2.9)	0	0
Pruritus	4 (11.8)	0	3 (8.8)	0
Maculopapular rash	4 (11.8)	0	4 (11.8)	0
Decreased white blood cell count	4 (11.8)	4 (11.8)	4 (11.8)	4 (11.8)

ALT, alanine aminotransferase; TEAE, treatment-emergent adverse event.
* Occurring in ≥10% of patients.

- Laboratory ALT/AST elevations of any grade were observed in 15 unique patients (44.1%), with grade ≥3 ALT/AST elevations in 6 patients (17.6%), mainly seen at doses 50 to 75 mg
 - Laboratory grade ≥3 elevations included ALT elevations in 5 patients (14.7%) and AST elevations in 4 patients (11.8%)
 - No fatalities were observed, and all patients recovered

Pharmacokinetics/Pharmacodynamics

- Exposure was dose dependent, with mean terminal half-life increasing from 28 to 69 hours in the 10-mg to 75-mg dose range; MCLA-145 followed target-mediated drug disposition (**Figure 4**, **Table 3**)
- At doses of 10 mg and above, peripheral T-cell activation, including cytotoxic CD8+ T cells, was observed (**Figure 5**)
- Pharmacodynamic effects reached a plateau across 10 to 75 mg dose levels, which is consistent with signs of a bell-shaped dose-response curve observed *in vitro* (**Figure 2**)[1]
 - *In vitro*, maximum IL-2, IFNγ, and TNFα was released by activated T cells after 3 days of incubation with MCLA-145 at concentrations below 1 μg/mL
- This is in line with a predicted plateau for simultaneous binding of MCLA-145 to its targets PD-L1 and CD137 in tumors (trimers) by an integrated semi-mechanistic pharmacokinetic/pharmacodynamic model

Figure 4. Serum Concentrations of MCLA-145 After First Dose

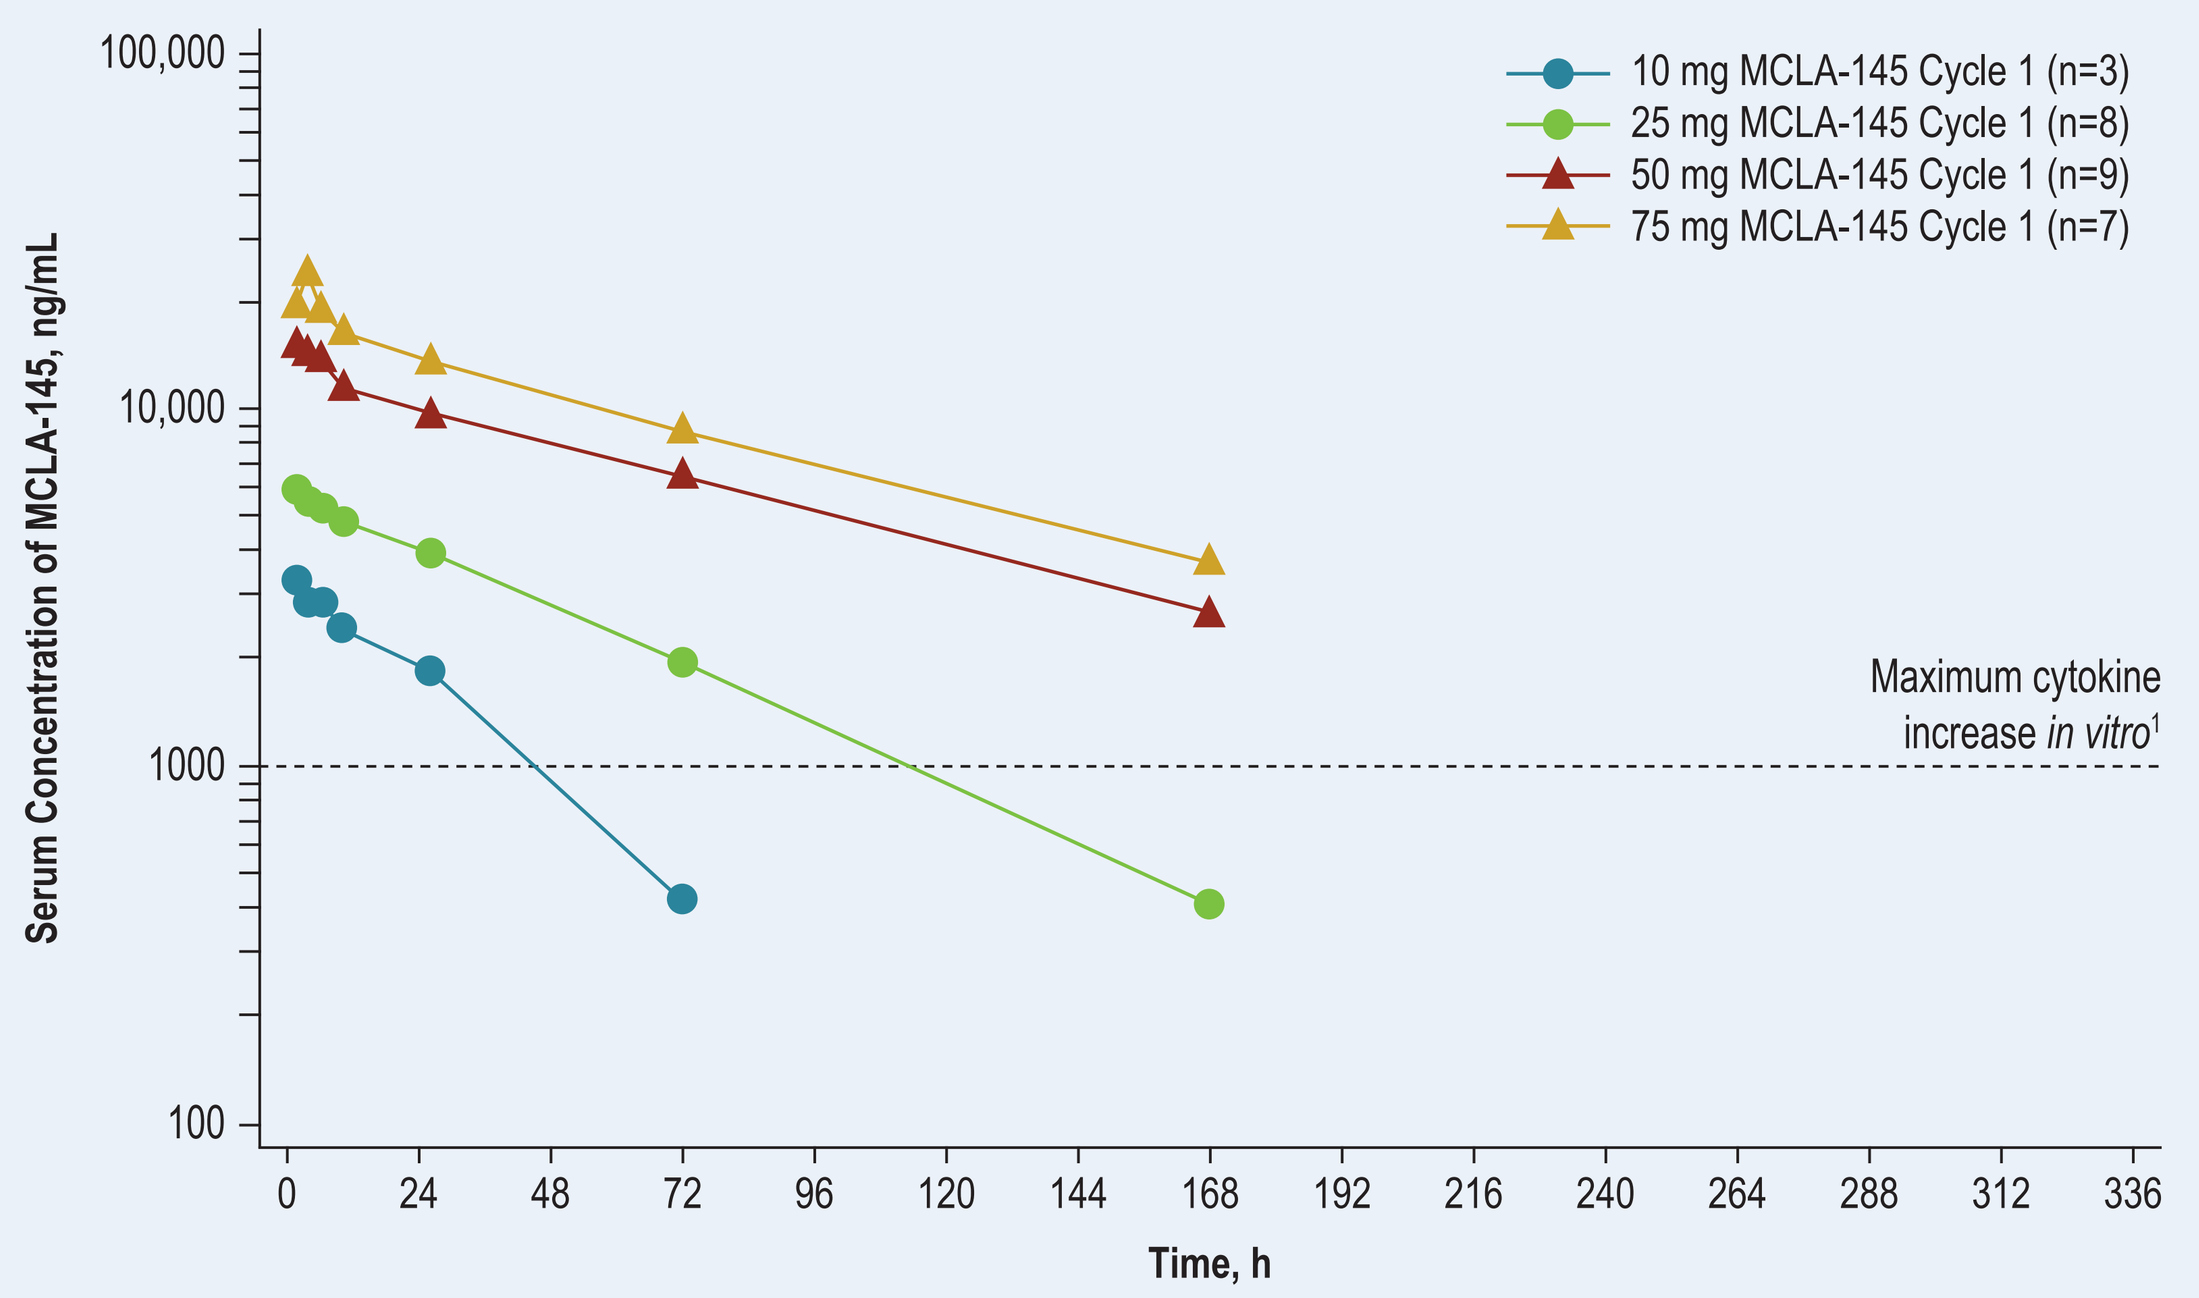

Serum Concentration of MCLA-145, ng/mL
Time, h
10 mg MCLA-145 Cycle 1 (n=3)
25 mg MCLA-145 Cycle 1 (n=8)
50 mg MCLA-145 Cycle 1 (n=9)
75 mg MCLA-145 Cycle 1 (n=7)
Maximum cytokine increase in vitro[1]

Table 3. Pharmacokinetic Parameters After First Dose

Dose, mg	n	C_{max}, μg/mL	AUC_{inf}, mg·h/mL	$t_{1/2}$, h
10	3	3.3	135	28
25*	8	6.3	388	40
50	9	15.7	1511	64
75	7	26.7	1998	69

AUC_{inf}, area under the curve from 0 to infinity; C_{max}, maximum concentration; $t_{1/2}$, half-life.
* 25 mg data includes patients from the intermediate dosing cohort.

Figure 5. Peripheral Blood T-Cell Proliferation

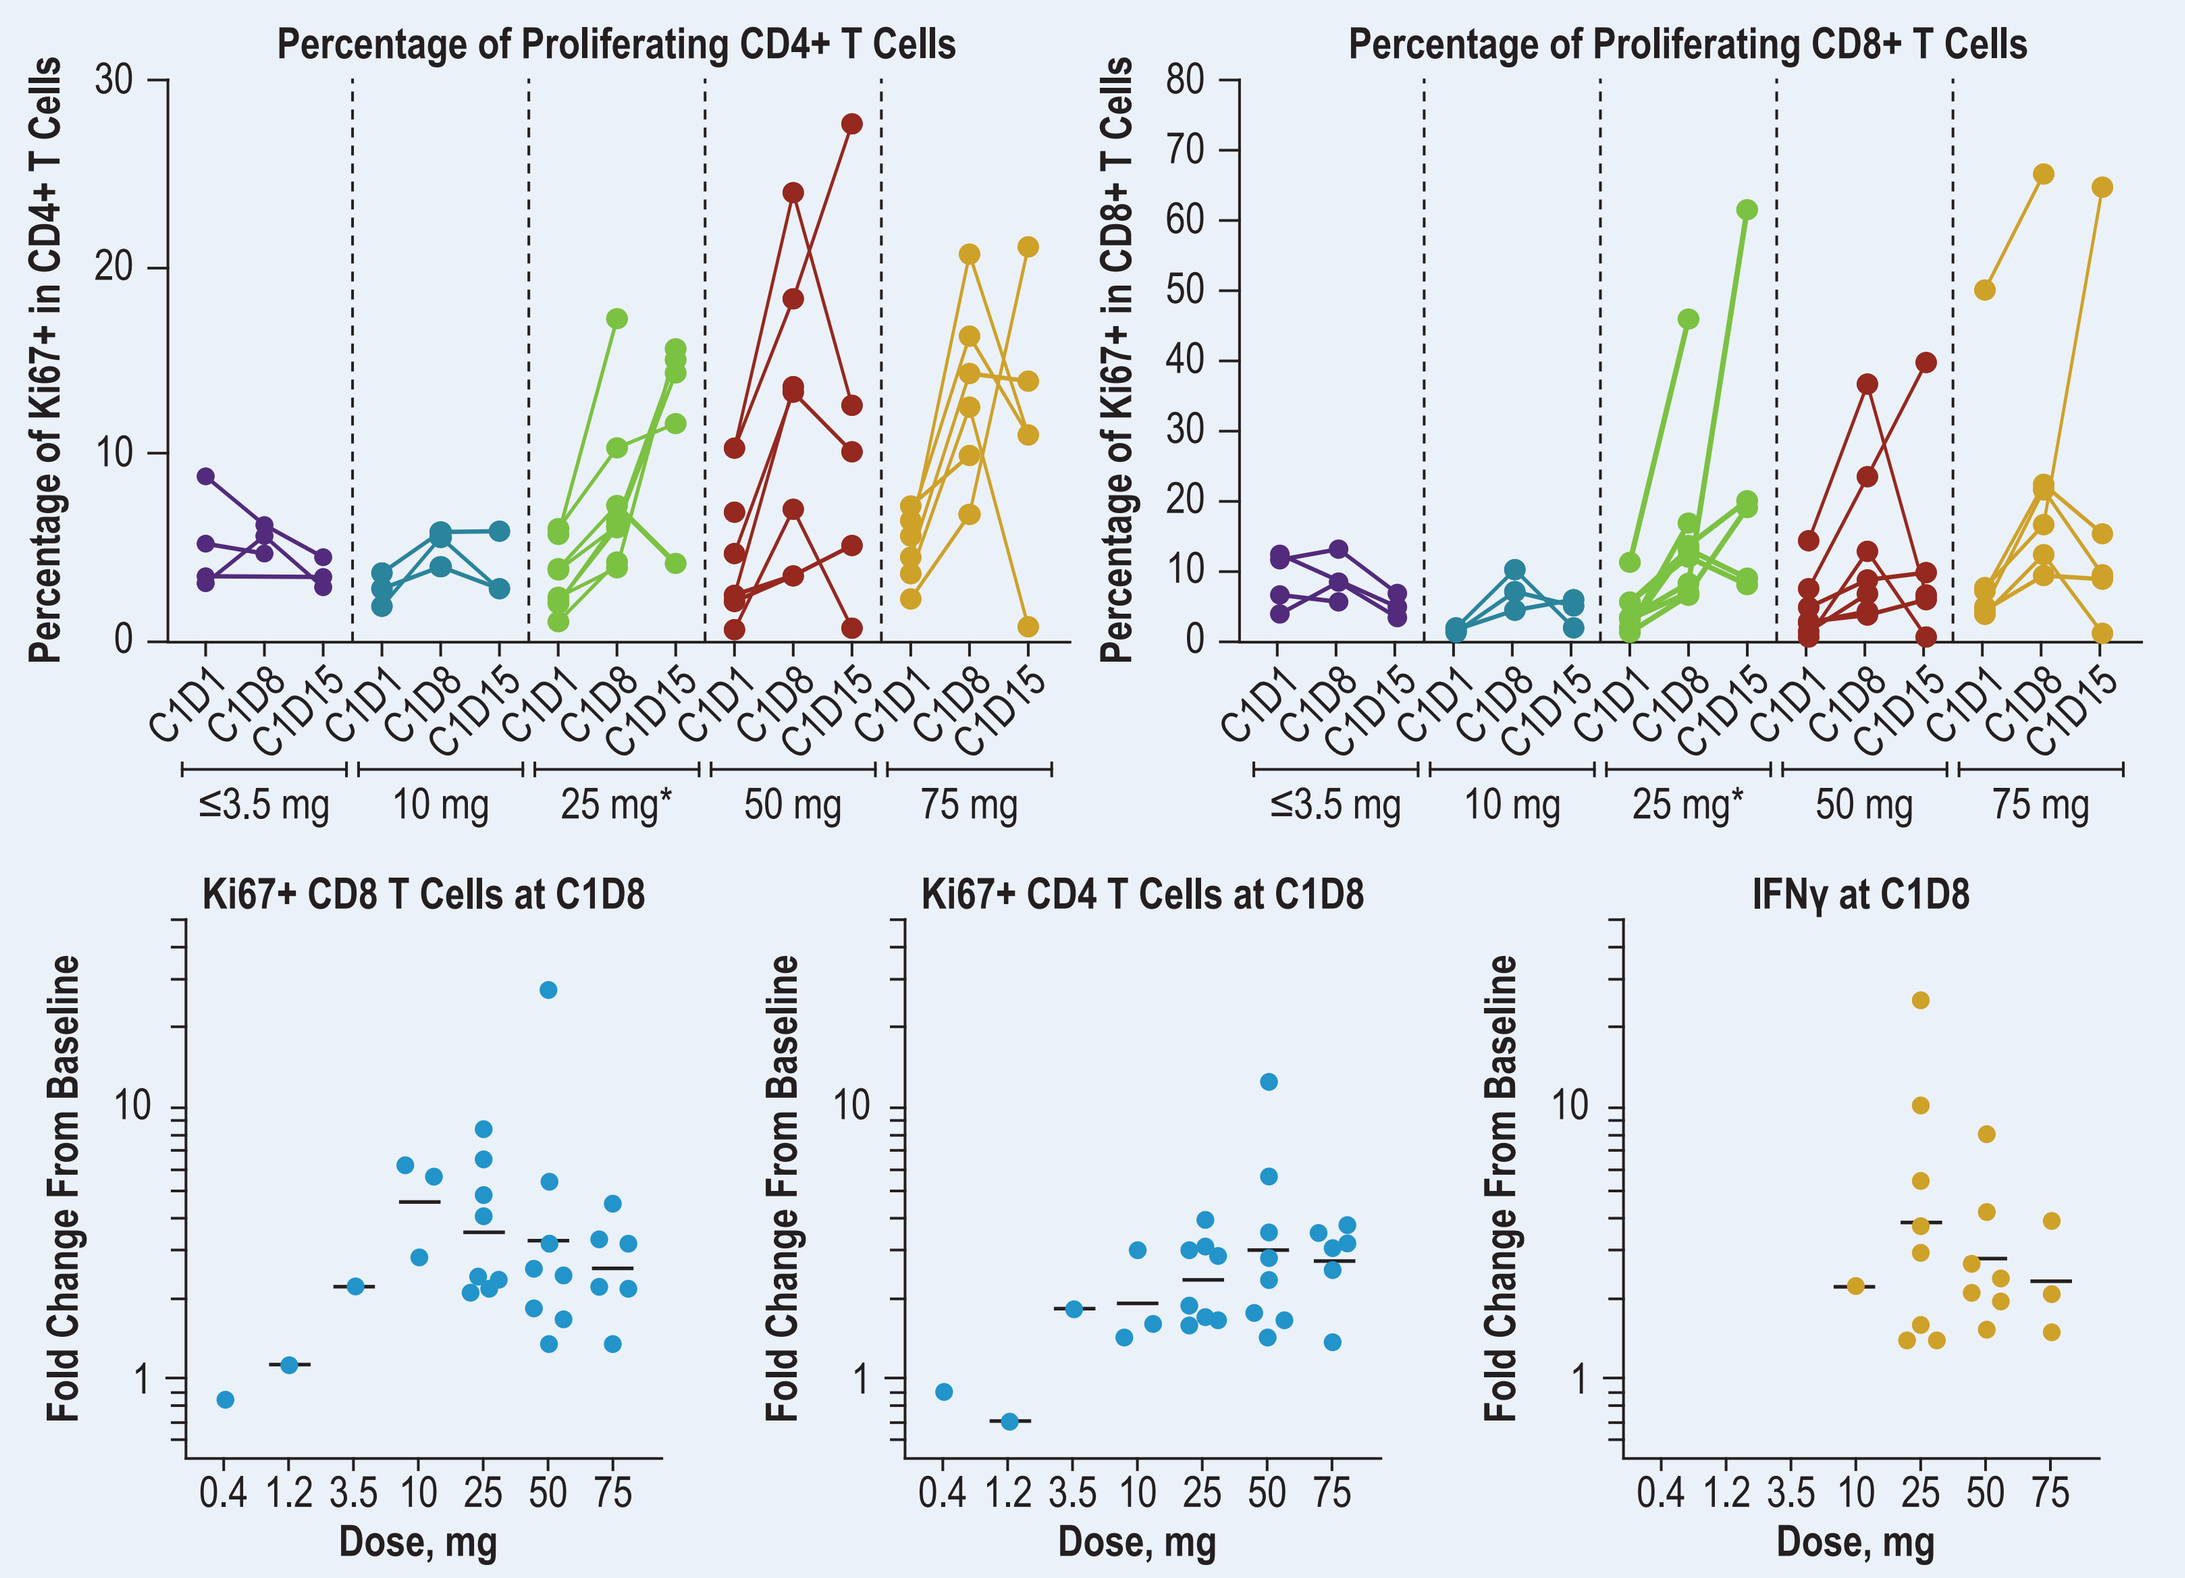

Percentage of Proliferating CD4+ T Cells
Percentage of Proliferating CD8+ T Cells
Percentage of Ki67+ in CD4+ T Cells
Percentage of Ki67+ in CD8+ T Cells
≤3.5 mg
10 mg
25 mg*
50 mg
75 mg
Ki67+ CD8 T Cells at C1D8
Ki67+ CD4 T Cells at C1D8
IFNγ at C1D8
Fold Change From Baseline
Dose, mg

C, cycle; D, day.
* 25 mg data includes patients from the intermediate dosing cohort.
Horizontal lines=Geometric mean per dose level.
Limited data for IFNγ fold change due to levels below detection limit for 13/33 patients.

Signs of Antitumor Activity

- Among patients treated at doses between 25 and 75 mg (n=27), 5 of 12 patients with available data had documented PD-L1 tumor expression ≥1%, including the cervical cancer patient with a PR (Patient 1)
- Preliminary evidence of antitumor activity has been observed at doses ≥25 mg
 - **Patient 1 (MCLA-145, 50 mg, PD-L1+)** with papillary cervical cancer diagnosed in 2014 who enrolled in the study following surgery, radiation therapy, and chemotherapy
 - Achieved a partial response (35% decrease) per RECIST v1.1 as determined by the investigator after discontinuation from MCLA-145 therapy due to elevated ALT/AST
 - **Patient 2 (MCLA-145, 25 mg, PD-L1 status unknown)** with glioblastoma diagnosed in 2010, who received prior surgery, radiation therapy, and treatment with temozolomide
 - Showed shrinkage of brain lesion present at screening; latest response assessment continues to show stable brain lesion and is fluid-attenuated inversion recovery (FLAIR) stable
 - Treatment ongoing, >21 months

Tumor Biomarkers

- Paired biopsies were available for 8 patients; changes in tumor CD8 and PD-L1 levels pre- and post-dose are shown in **Figure 6**

Figure 6. Tumor Biomarkers in 8 Patients With Available Paired Biopsies

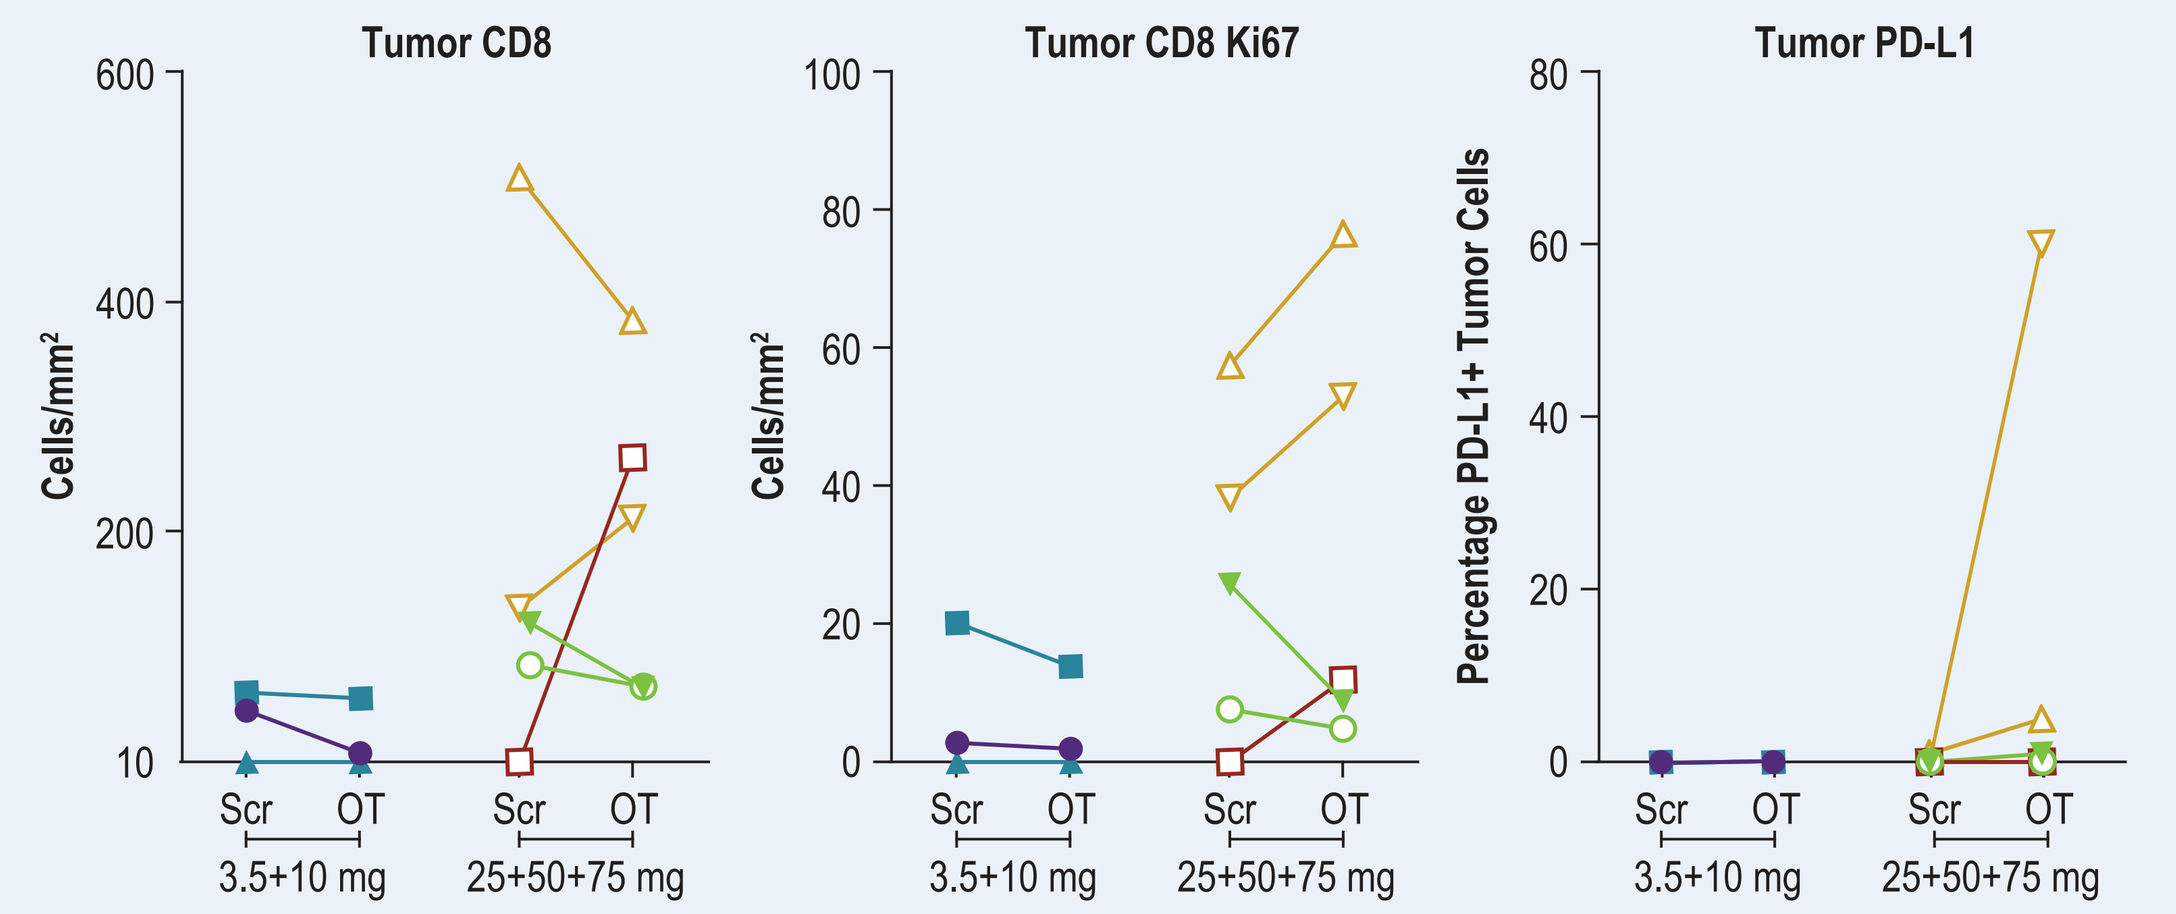

Tumor CD8
Tumor CD8 Ki67
Tumor PD-L1
Cells/mm²
Percentage PD-L1+ Tumor Cells
Scr
OT
3.5+10 mg
25+50+75 mg

3.5 mg, purple; 10 mg, blue; 25 mg, green; 50 mg, red; 75 mg, orange.
Scr, screening; OT, on treatment.

Conclusions

- **Thirty-four patients have been treated with MCLA-145 at dose levels ranging from 0.4 to 75 mg Q2W**
- **Reported AEs are consistent with the mechanism of action and can be managed with drug interruption and/or administration of steroids in some patients**
- **Preliminary evidence of antitumor activity has been observed at doses ≥25 mg**
- **Peripheral blood shows robust T-cell activation following treatment with MCLA-145, including activation of cytotoxic CD8+ cells and IFNγ, and reached a plateau across the MCLA-145 10 to 75 mg Q2W dosing range**
- **Increase in tumor CD8+ T cells and PD-L1 expression has been observed in some patients**
- **Patients continue to be enrolled at 25 mg Q2W. Further evaluation of optimal dose and efficacy in PD-L1+ tumors is planned. Full blockade of PD-L1 may be required**

Disclosures

HP has received honoraria from Roche, Bayer, Amgen, Ipsen, Pfizer, Novartis, Sanofi, Merck, Vifor Pharma, and Lilly. CK has received research funding from Bristol Myers Squibb, Merus, and Gritstone Oncology. GVL has no competing interest to declare. SPP has received scientific advisory income from Amgen, AstraZeneca, Bristol Myers Squibb, Eli Lilly, Genentech, Illumina, Merck, Rakuten, Paradigm, and Tempus. DM, AW, KB, and VS are employees and shareholders of Merus NV. JP, GZ, ZD, and EA are employees and shareholders of Incyte Corporation. FSH has served as a consultant advisor for Aduro, Bristol Myers Squibb, Eisai, EMD Serono, Genentech/Roche, Idera, Merck, Novartis, Pieris Pharmaceutical, Sanofi, and Takeda; holds equity in Apricity, Bicara, Pionyr, and Torque; and has served on advisory boards for Apricity, Bicara, Bioentre, Checkpoint Therapeutics, Compass Therapeutics, Iovance, Pionyr, Surface, and Torque.

Acknowledgments

Support for this study was provided by Incyte Corporation (Wilmington, DE, USA) and Merus NV (Utrecht, Netherlands). We thank Paul Tacken (Merus) for contributions to the analysis and presentation. Medical writing assistance was provided by Wendy van der Spuy, PhD, an employee of ICON (North Wales, PA, USA), and was funded by Incyte Corporation.

References

1. Geuijen C, et al. *Nat Commun*. 2021;12(1):4445. **2.** Plyte S, et al. *J Immunother Cancer*. 2019;7(Suppl 1):282 (abstract P489). **3.** Willoughby JE, et al. *J Immunol*. 2014;193(1):244. **4.** Eskiocak U, et al. *JCI Insight*. 2020;5(5):e133647.

To download a copy of this poster, scan code.

Copies of this e-poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.