Memorial Sloan Kettering Cancer Center

The HER2×HER3 bi-specific antibody Zenocutuzumab is effective at blocking growth of tumors driven by NRG1 gene fusions

Igor Odintsov[a], MD, Inna Khodos[a], MSc, Madelyn Espinosa-Cotton[a], PhD, Allan J.W. Lui[a], MD, Marissa Mattar[a], BSc, Alison M. Schram[a], MD, Marie N. O'Connor[b], PhD, Cecile A. W. Geuijen[b] PhD, Ron C.J. Schackmann[b], PhD, Jeroen J. Lammerts van Bueren[b], PhD, Elisa De Stanchina[a], PhD, Marc Ladanyi[a], MD, and Romel Somwar[a], PhD

[a]Memorial Sloan Kettering Cancer Center, New York, NY, USA

[b]Merus, Utrecht, The Netherlands

Abstract

Fusions involving the neuregulin 1 gene (NRG1) occur at low frequency in pancreatic, lung, and other cancers. NRG1 fusion oncoproteins bind to HER3, leading to heterodimerization with HER2 and potent activation of downstream signaling mainly via the PI3K-AKT pathway. Zenocutuzumab (Zeno, MCLA-128), an ADCC-enhanced anti-HER2×HER3 bi-specific antibody, uniquely 'docks' on HER2, to position the antibody and subsequently 'block' NRG1 from interacting with HER3, effectively preventing HER2:HER3 heterodimerization and downstream signaling. Our goal in this study was to evaluate the efficacy of Zeno in preclinical models of NRG1 fusion-positive cancers.

We tested Zeno in a panel of isogenic and patient-derived cell line and xenograft (PDX) models of lung, breast and pancreatic cancers. Cell lines either expressed an NRG1 fusion endogenously (MDA-MB-175-VII, DOC4-NRG1) or by lentiviral transfer of cDNAs (ATP1B1-NRG1 and SLC3A2-NRG1 in H6c7 pancreatic ductal cell line; CD74-NRG1 and VAMP2-NRG1 in immortalized human bronchial epithelial cells; and DOC4-NRG1 in MCF7 breast cancer cells). PDX models were generated from NSCLC samples harboring CD74-NRG1 (ST3204) or SLC3A2-NRG1 (LUAD-0061AS3) fusions and from a high grade serous ovarian cancer harboring a CLU-NRG1 fusion (OV-10-0050). Zeno treatment of NRG1 fusion-expressing breast, pancreatic, and lung cancer cell lines resulted in dose-dependent reduction of growth, and abrogated phosphorylation of HER3, HER4, AKT, p70S6 kinase and STAT3 in all cell lines tested. Phosphorylation of HER2, EGFR and MEK/ERK was inhibited, albeit with some variation, in a cell line-specific manner. Growth of isogenic control cell lines without NRG1 fusion was not significantly altered by Zeno treatment. In breast and lung cancer cell lines, Zeno treatment down-regulated cyclin D1 level and induced expression of the negative cell cycle regulators P21 or P27. Evidence of apoptosis activation (cleaved PARP, expression of BIM and PUMA) was also observed in cells exposed to Zeno. Treatment of mice bearing LUAD-0061AS3, ST3204 and OV-10-0050 PDX tumors (2.5, 8, 25 mg/kg, QW) caused a dose-dependent inhibition of tumor growth, with tumor shrinkage observed at the highest dose which is equivalent to the current human dose (750 mg Q2W). Finally, we assessed the ability of Zeno to induce antibody-dependent cellular cytotoxicity (ADCC) using a chromium release assay and peripheral blood mononuclear cells. Zeno induced significant cytotoxicity in MDA-MB-175-VII cells while a non-ADCC enhanced, non-specific IgG had no effect.

Here we show that Zeno effectively blocks the growth of NRG1 fusion-positive cell lines and xenograft models of tumors arising from lung, pancreas and other organs. *In vitro*, Zeno has no significant effect on parental cells and only inhibits viability induced by expression of a NRG1 fusion. *In vivo*, Zeno demonstrated significant efficacy at clinically relevant dose levels illustrating potent therapeutic activity. These results support the continued development of Zeno to treat patients with this molecularly defined subset of cancers.

Preclinical disease models with *NRG1* fusion

Model	*NRG1* alteration	Tissue of origin	Patient-derived/isogenic	Cell line/xenograft	Source
MDA-MB-175-VII	*DOC4-NRG1* fusion	Breast	Patient-derived	Cell line	ATCC
HCC-95	*NRG1* amplification	Lung	Patient-derived	Cell line	ATCC
HBEC-CD74-NRG1	*CD74-NRG1* fusion, cDNA	Lung	Isogenic*	Cell line	MSKCC
HBEC-VAMP2-NRG1	*VAMP2-NRG1* fusion, cDNA	Lung	Isogenic*	Cell line	MSKCC
H6C7-ATP1B1-NRG1	*ATP1B1-NRG1* fusion, cDNA	Pancreas	Isogenic**	Cell line	MSKCC
H6C7-SLC3A2-NRG1	*SLC3A2-NRG1* fusion, cDNA	Pancreas	Isogenic**	Cell line/xenograft	MSKCC
LUAD-0061AS3	*SLC3A2-NRG1* fusion	Lung	Patient-derived	Cell line/xenograft	MSKCC
OV-10-0050	*CLU-NRG1* fusion	Ovarian	Patient-derived	Xenograft	WuXi AppTec
ST3204	*CD74-NRG1* fusion	Lung	Patient-derived	Xenograft	XenoSTART

Table 1. List of NRG1 fusion-positive models used in this study. * Human bronchial epithelial cells (HBEC, obtained from Dr. John Minna (UT Texas Southwestern)) were immortalized using overexpression of TERT, CDK4 and dominant negative p53. The resulting cell line (HBEC-DNP53) was transduced with *NRG1* fusion cDNAs.

**** Human pancreatic ductal epithelial cells were immortalized by ectopic expression of viral E6/E7 proteins. The resulting cell line (H6C7, obtained from Kerafast (NY)) was transduced with NRG1 fusions.**

Zeno is effective at inhibiting growth and downstream signaling in NRG1-dependent cell lines

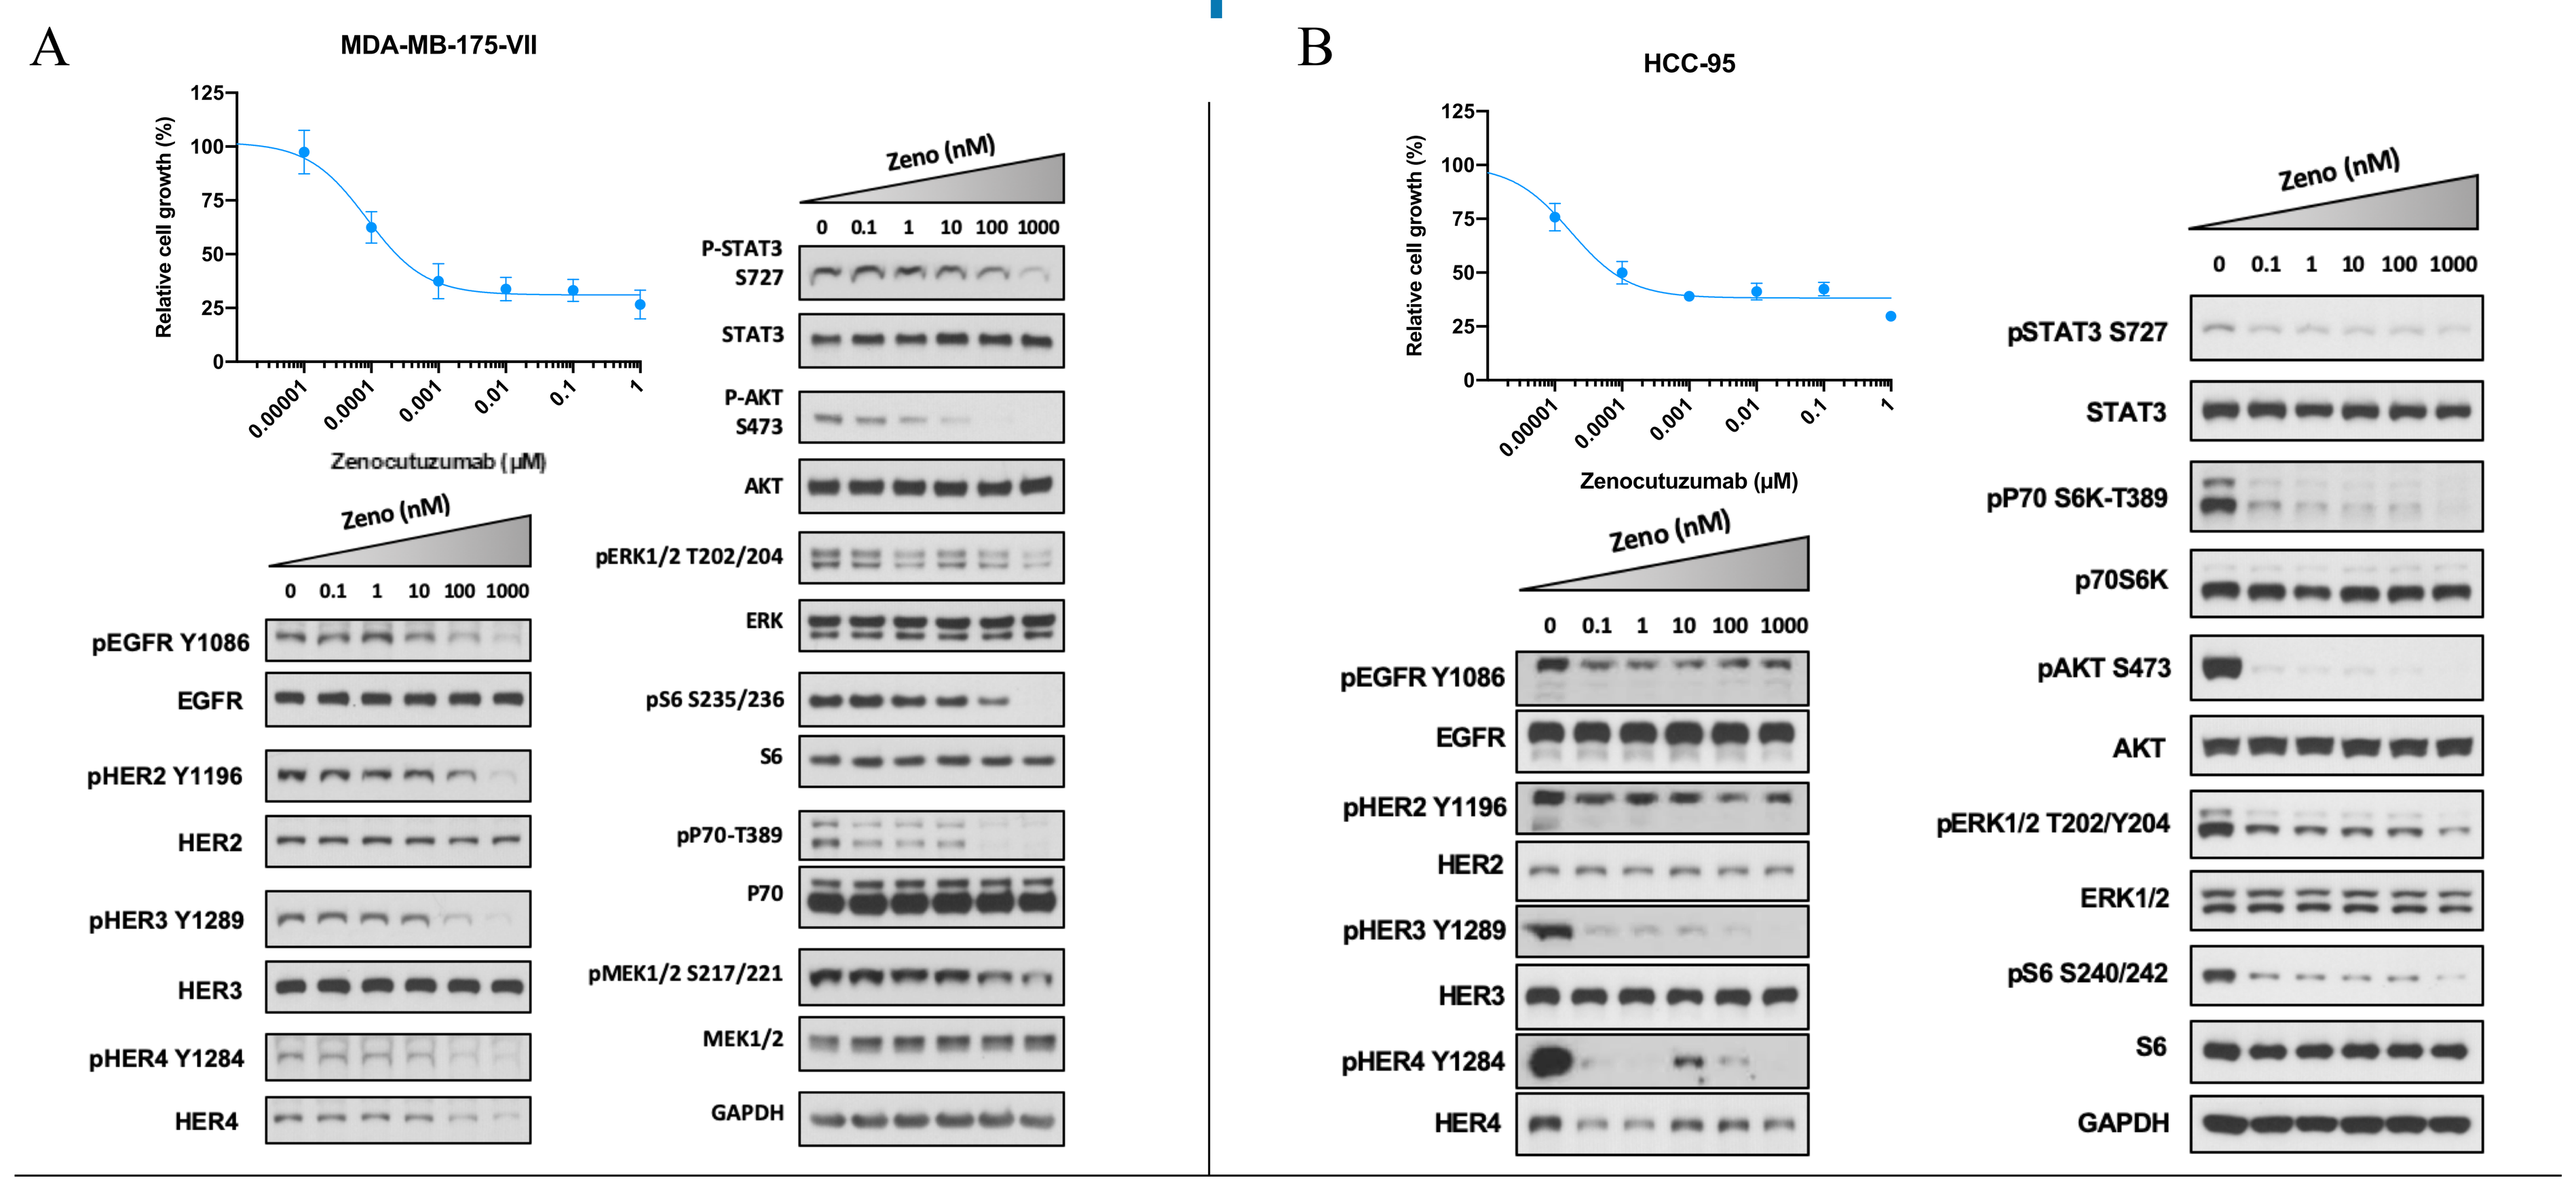
A
MDA-MB-175-VII
B
HCC-95
Relative cell growth (%)
Zenocutuzumab (µM)
Zeno (nM)
0 0.1 1 10 100 1000
pEGFR Y1086
EGFR
pHER2 Y1196
HER2
pHER3 Y1289
HER3
pHER4 Y1284
HER4
P-STAT3 S727
STAT3
P-AKT S473
AKT
pERK1/2 T202/204
ERK
pS6 S235/236
S6
pP70-T389
P70
pMEK1/2 S217/221
MEK1/2
GAPDH
pSTAT3 S727
pP70 S6K-T389
p70S6K
pAKT S473
pERK1/2 T202/Y204
ERK1/2
pS6 S240/242

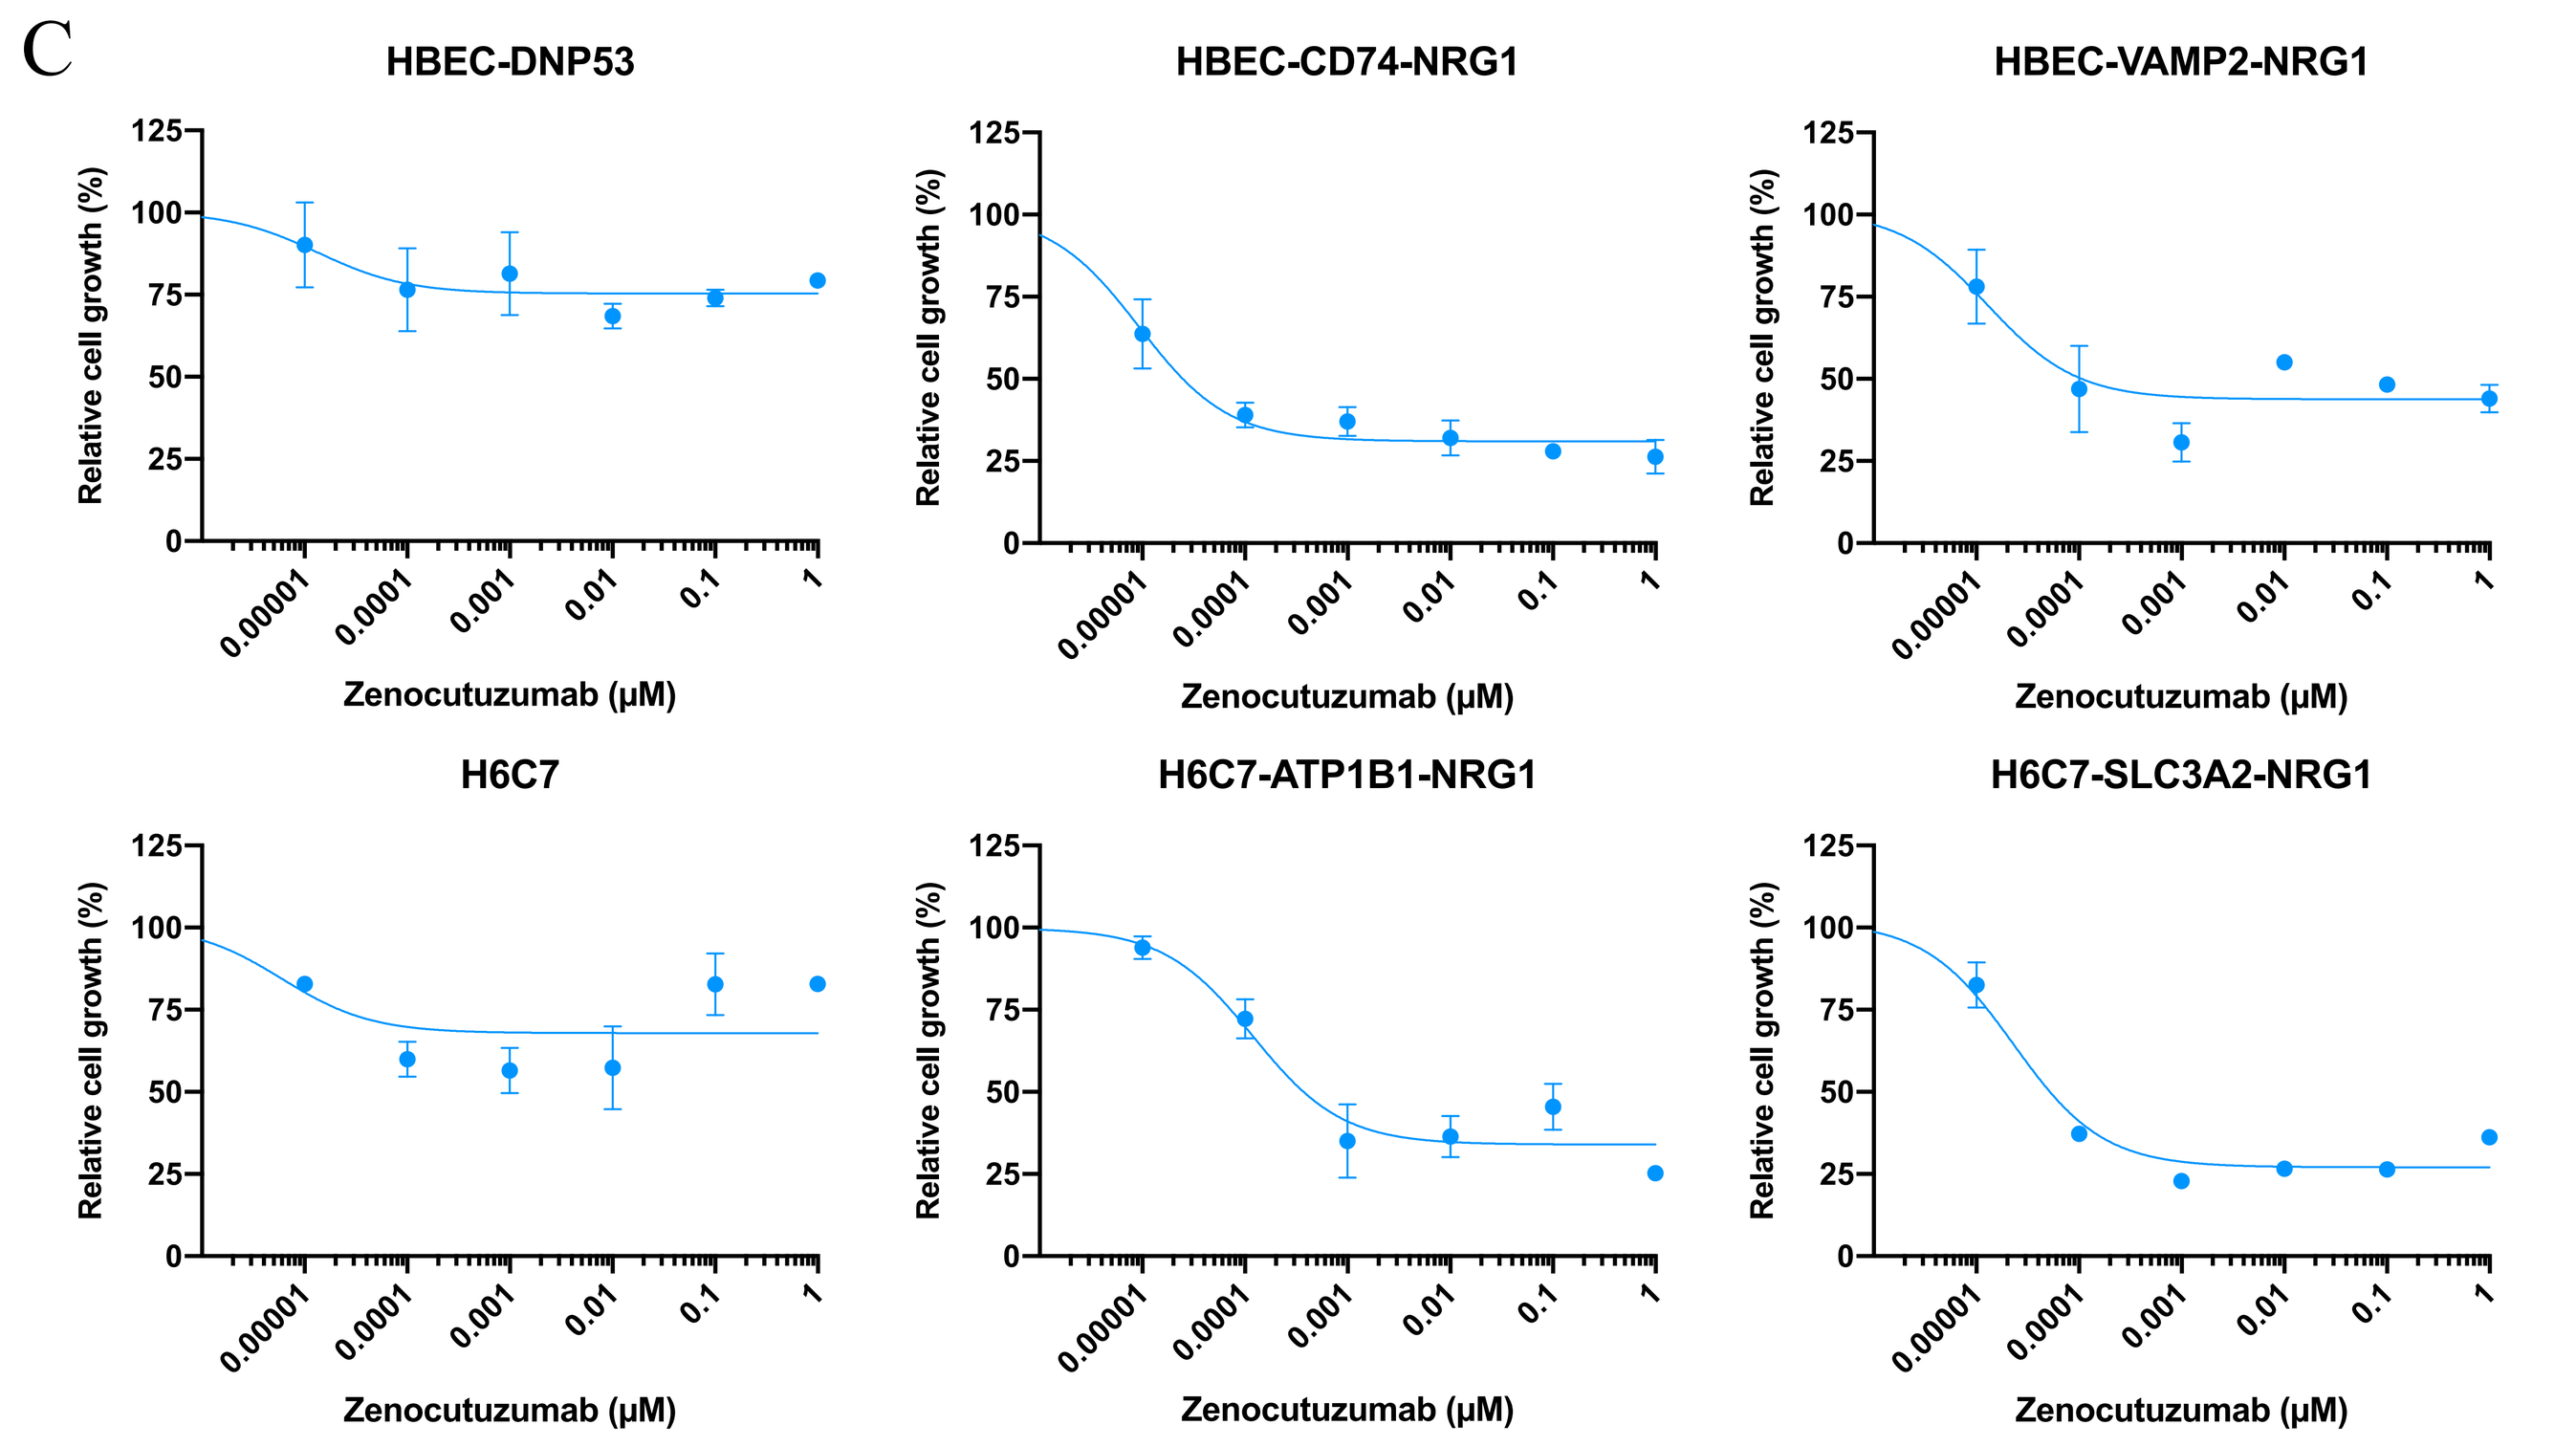
C
HBEC-DNP53
HBEC-CD74-NRG1
HBEC-VAMP2-NRG1
H6C7
H6C7-ATP1B1-NRG1
H6C7-SLC3A2-NRG1
Relative cell growth (%)
Zenocutuzumab (µM)

Figure 1. A and B: Cells were treated for 96 hours (graphs) or 3 hours with the indicated concentrations of Zeno. Viability assay (graphs) or Western blotting was then performed. C: Cell lines expressing various NRG1 fusions, and their isogenic controls were treated with the indicated concentrations of Zeno after which viability was determined.

Zeno promotes changes in apoptotic and cell cycle pathways and induces ADCC-dependent cell lysis

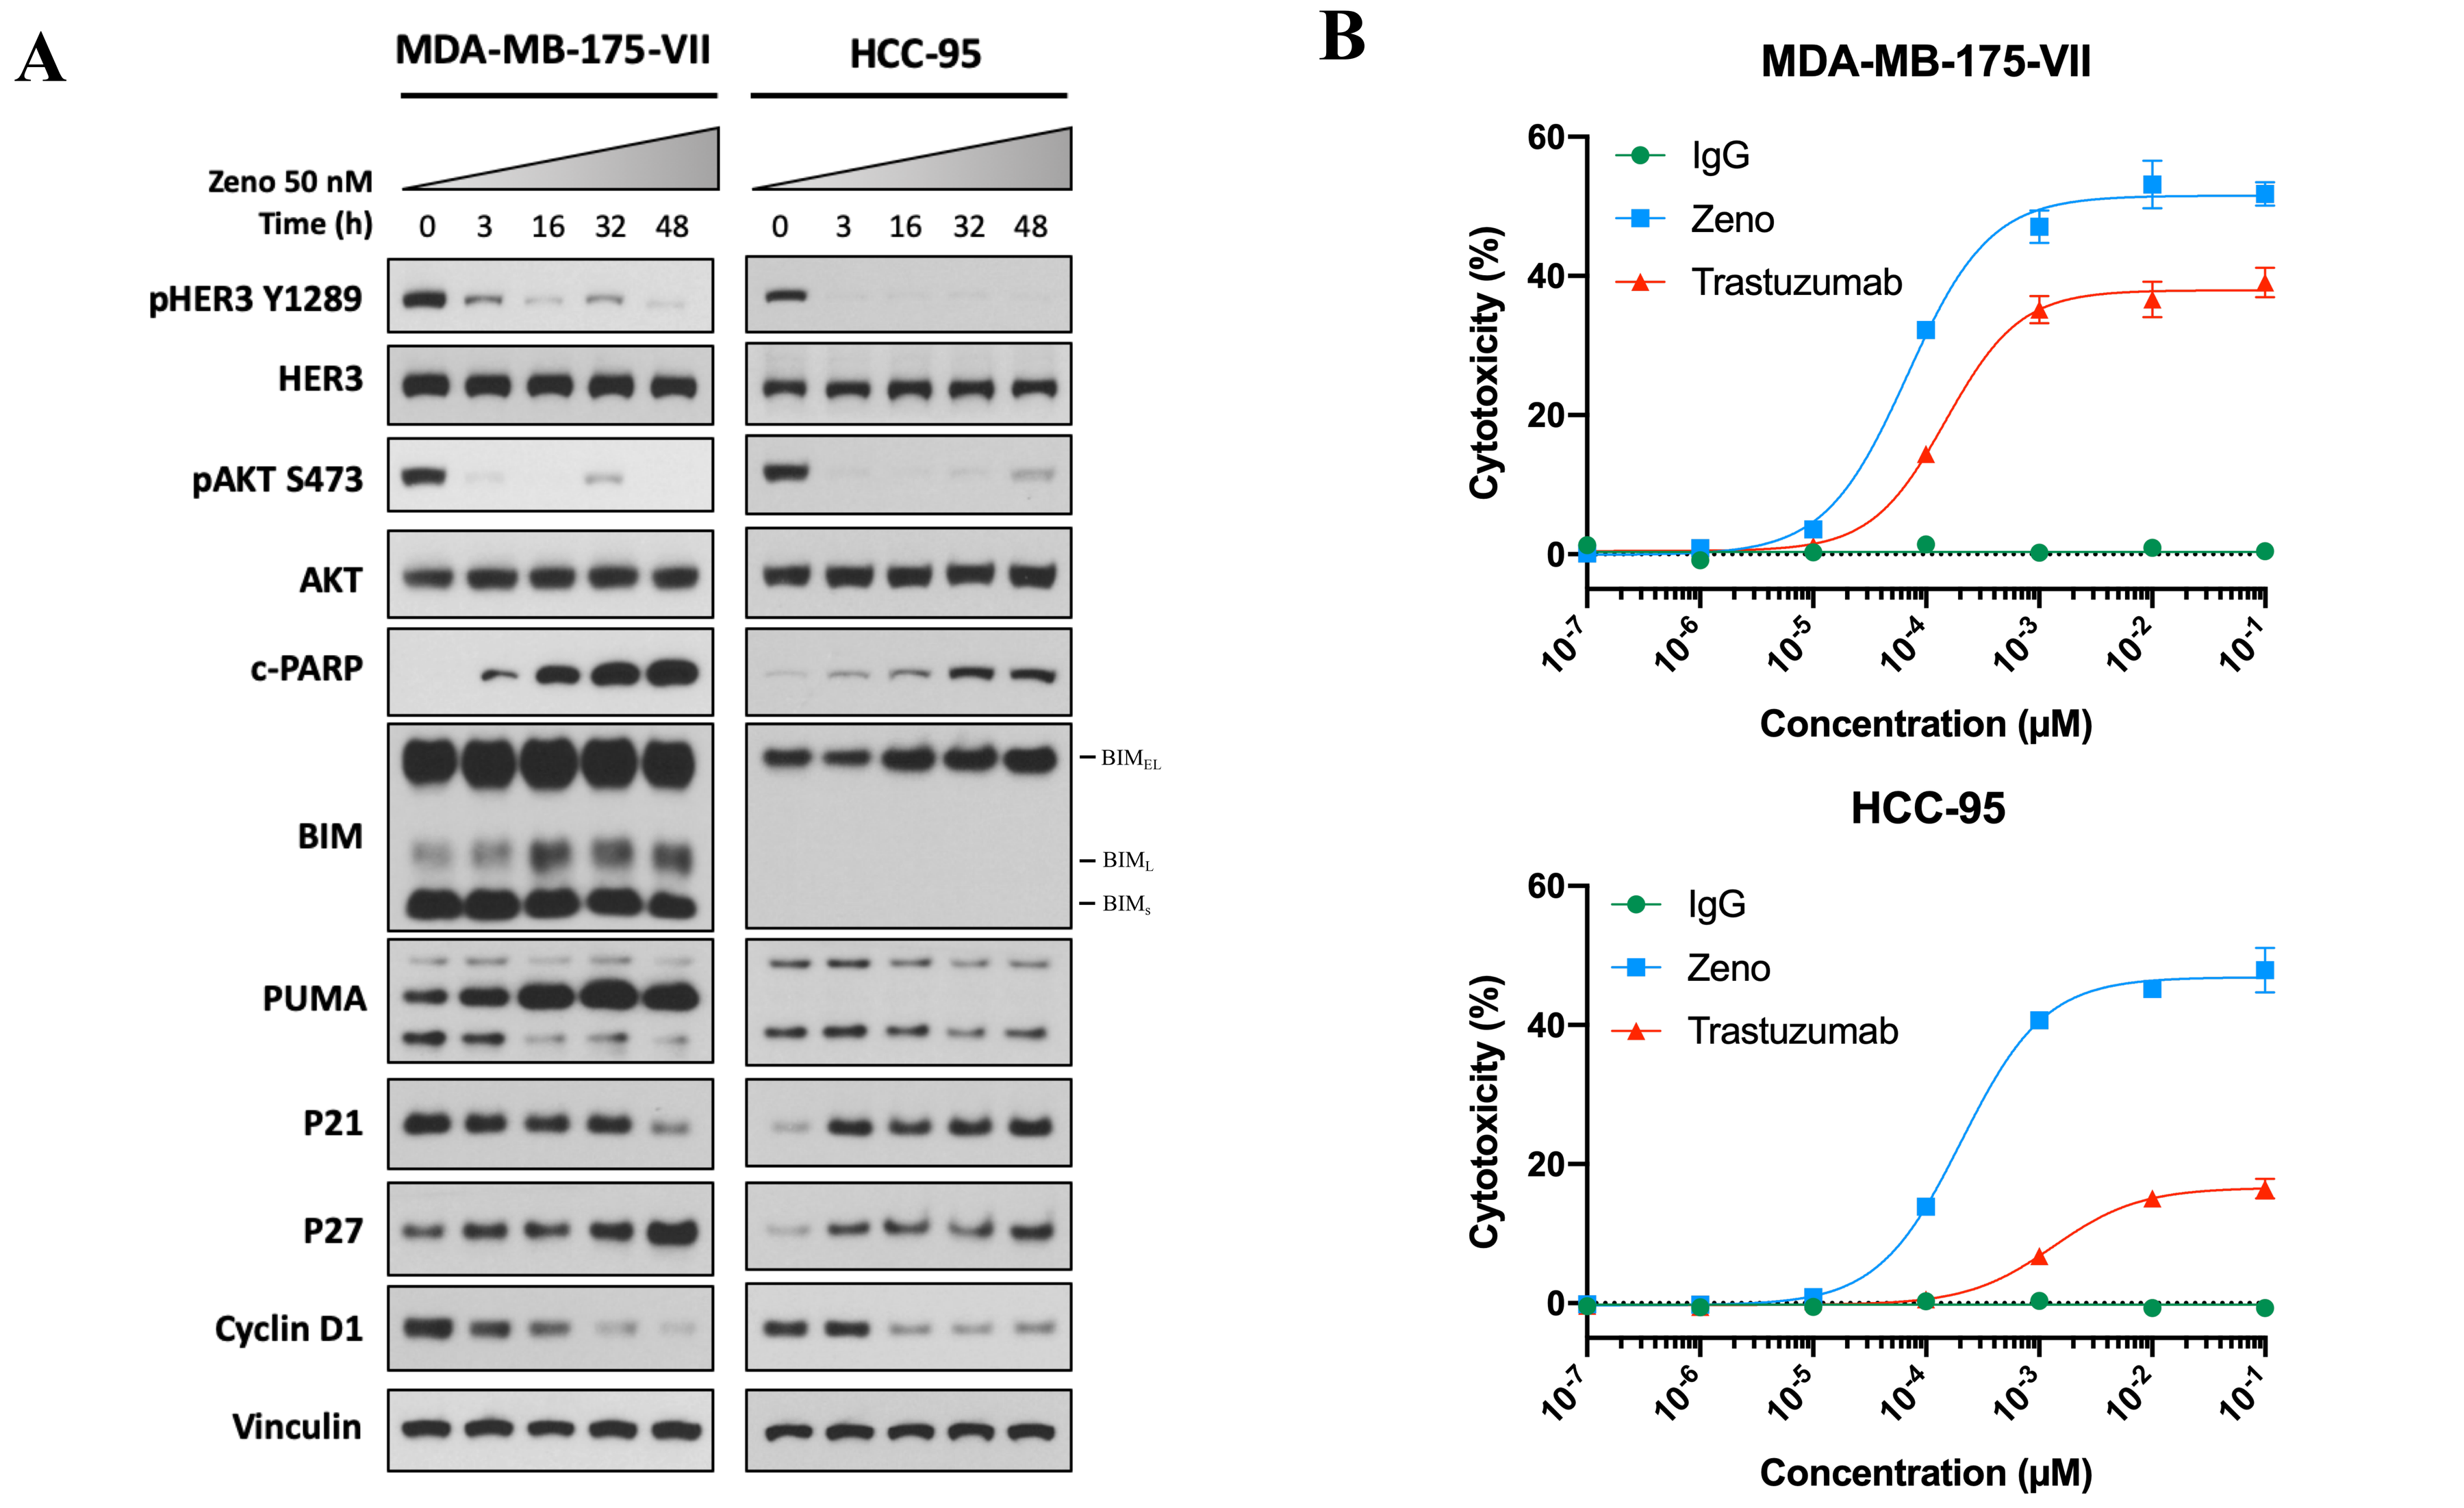
A
MDA-MB-175-VII
HCC-95
Zeno 50 nM
Time (h)
0 3 16 32 48
pHER3 Y1289
HER3
pAKT S473
AKT
c-PARP
BIM
PUMA
P21
P27
Cyclin D1
Vinculin
B
MDA-MB-175-VII
HCC-95
IgG
Zeno
Trastuzumab
Cytotoxicity (%)
Concentration (µM)

Figure 2. A: Cells were treated with Zeno for the indicated time periods and protein expression was determined by Western blotting.

B: ^{51}Chromium-release assay was performed to evaluate ADCC-dependent activity of Zeno in comparison to trastuzumab. IgG – nonspecific IgG antibody.

Zeno effectively blocks growth of NRG1 fusion-positive PDX models

LUAD-0061AS3, *SLC3A2-NRG1*, lung cancer

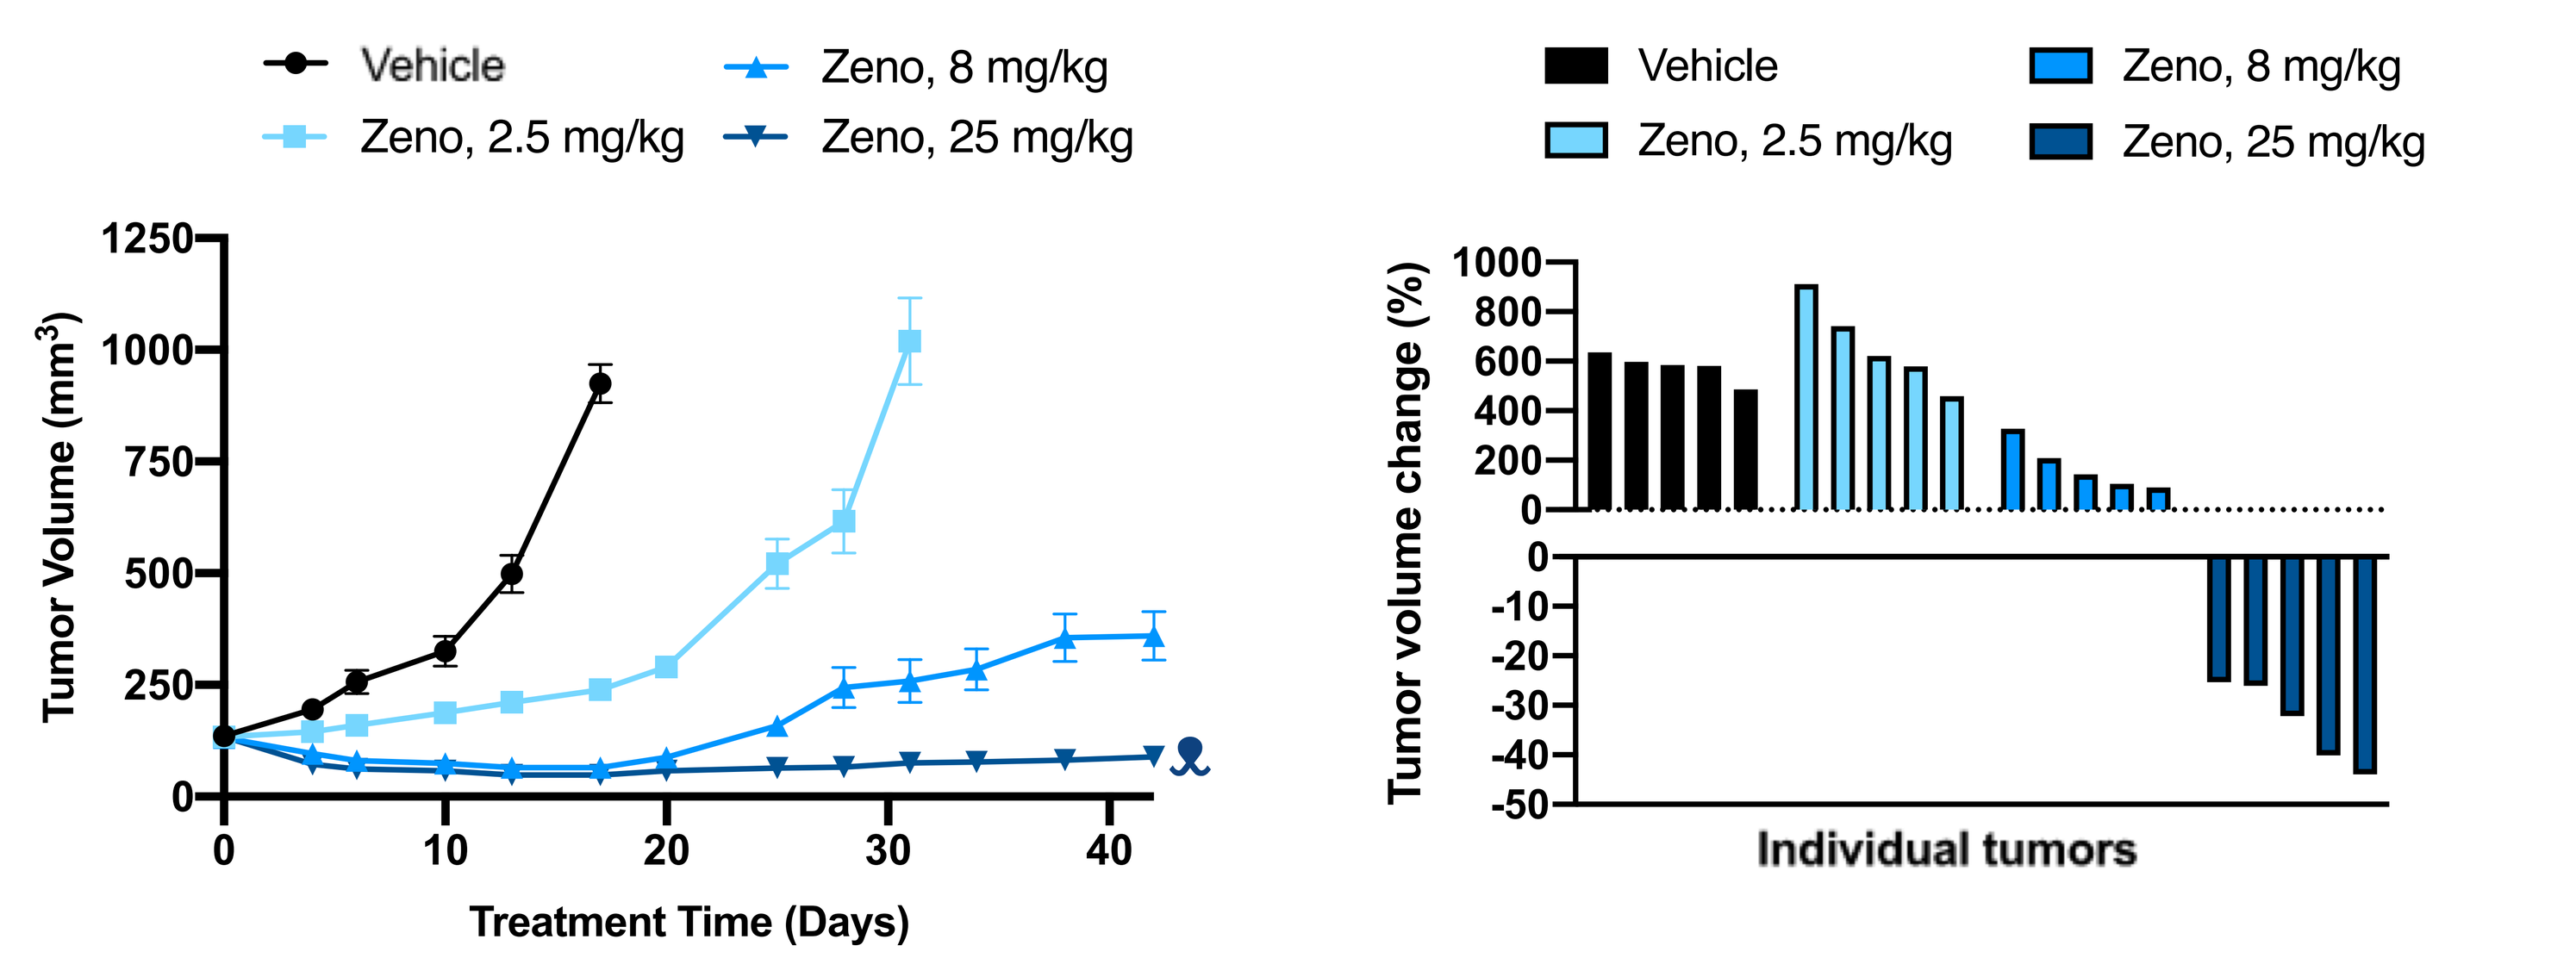
Vehicle
Zeno, 2.5 mg/kg
Zeno, 8 mg/kg
Zeno, 25 mg/kg
Tumor Volume (mm^3)
Treatment Time (Days)
Tumor volume change (%)
Individual tumors

OV-10-0050, *CLU-NRG1*, ovarian cancer

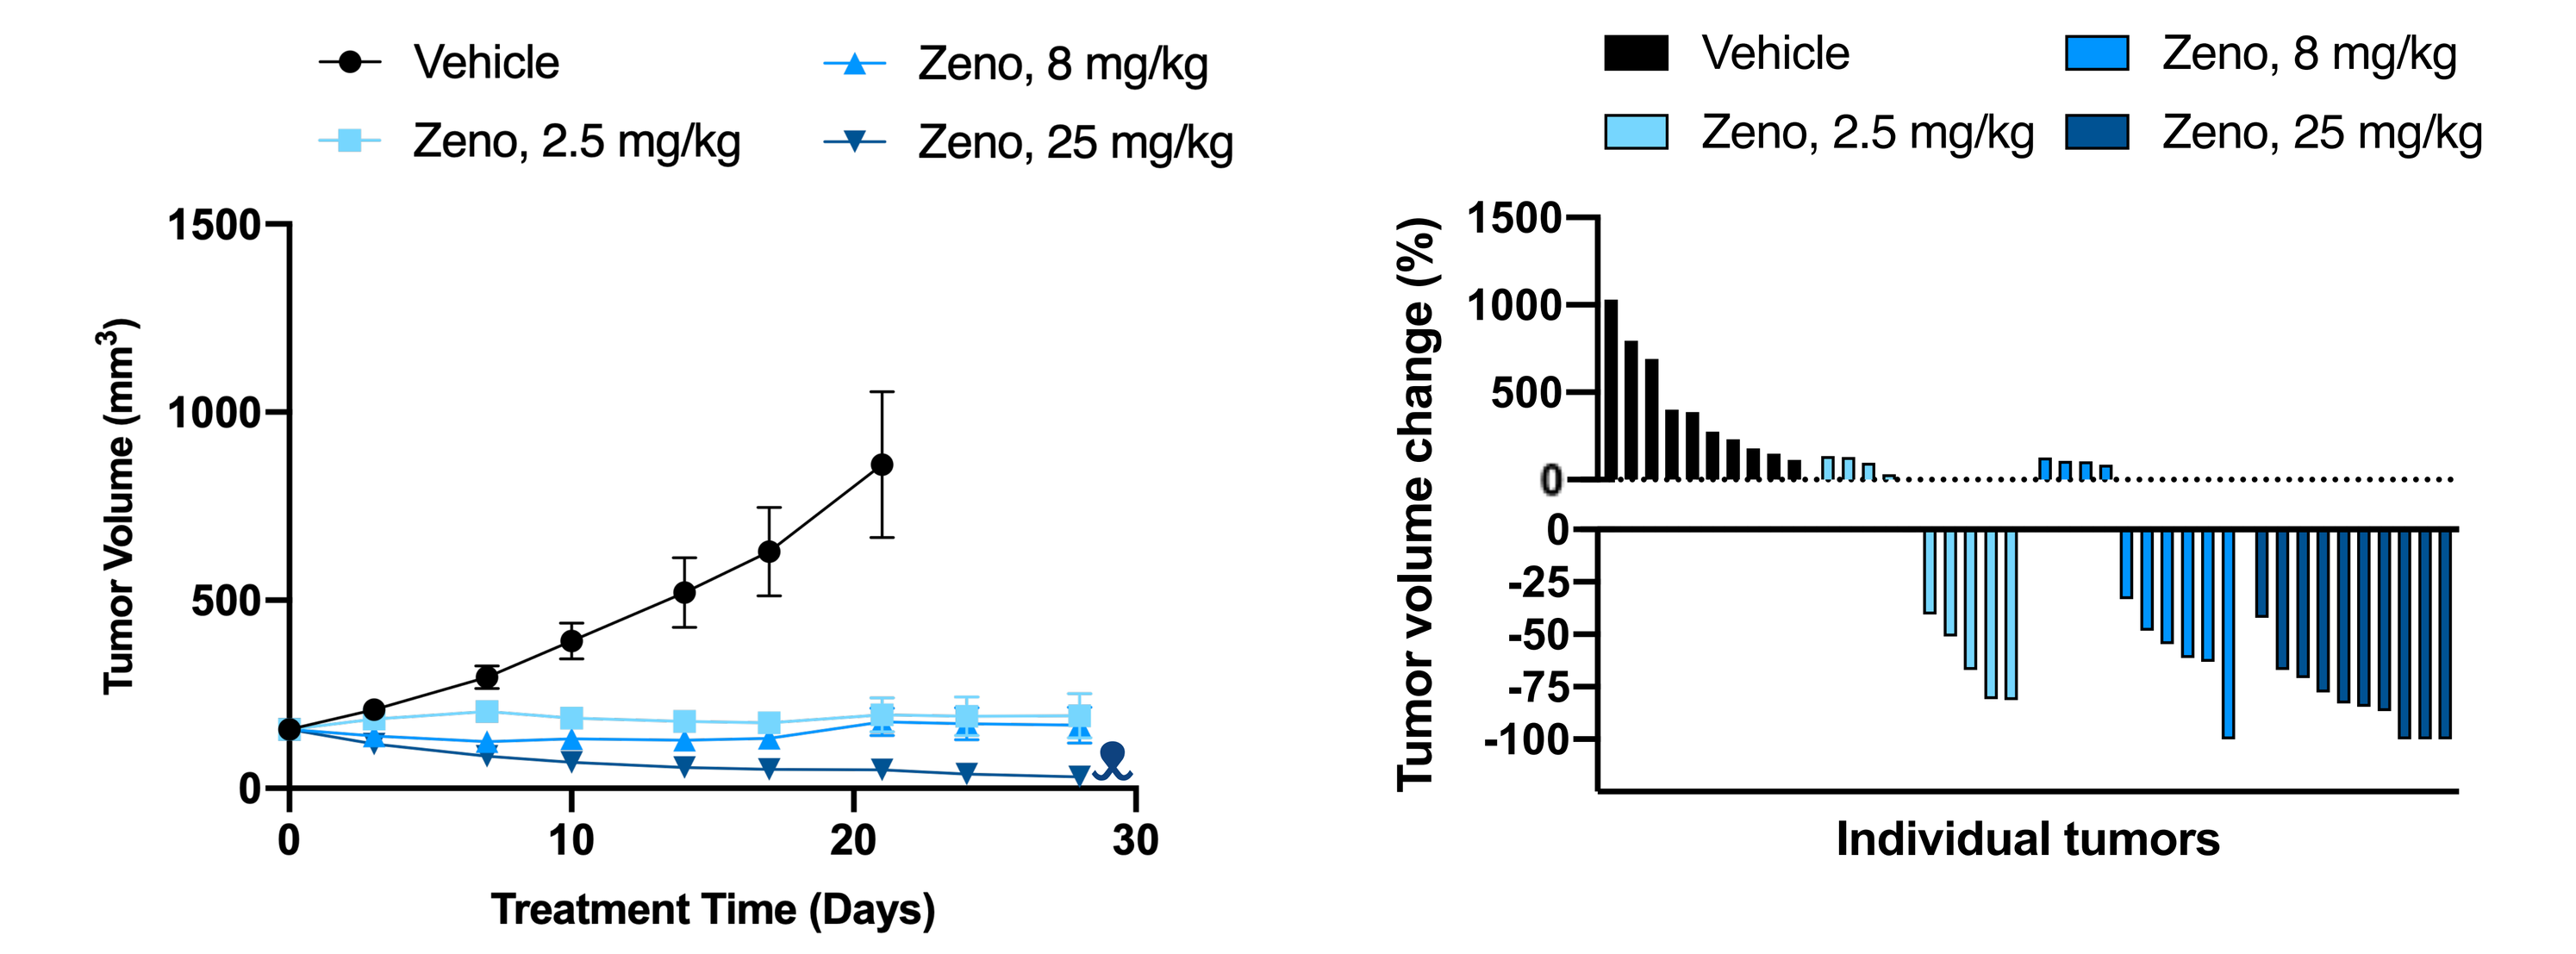
Vehicle
Zeno, 2.5 mg/kg
Zeno, 8 mg/kg
Zeno, 25 mg/kg
Tumor Volume (mm^3)
Treatment Time (Days)
Tumor volume change (%)
Individual tumors

ST3204, *CD74-NRG1*, lung cancer

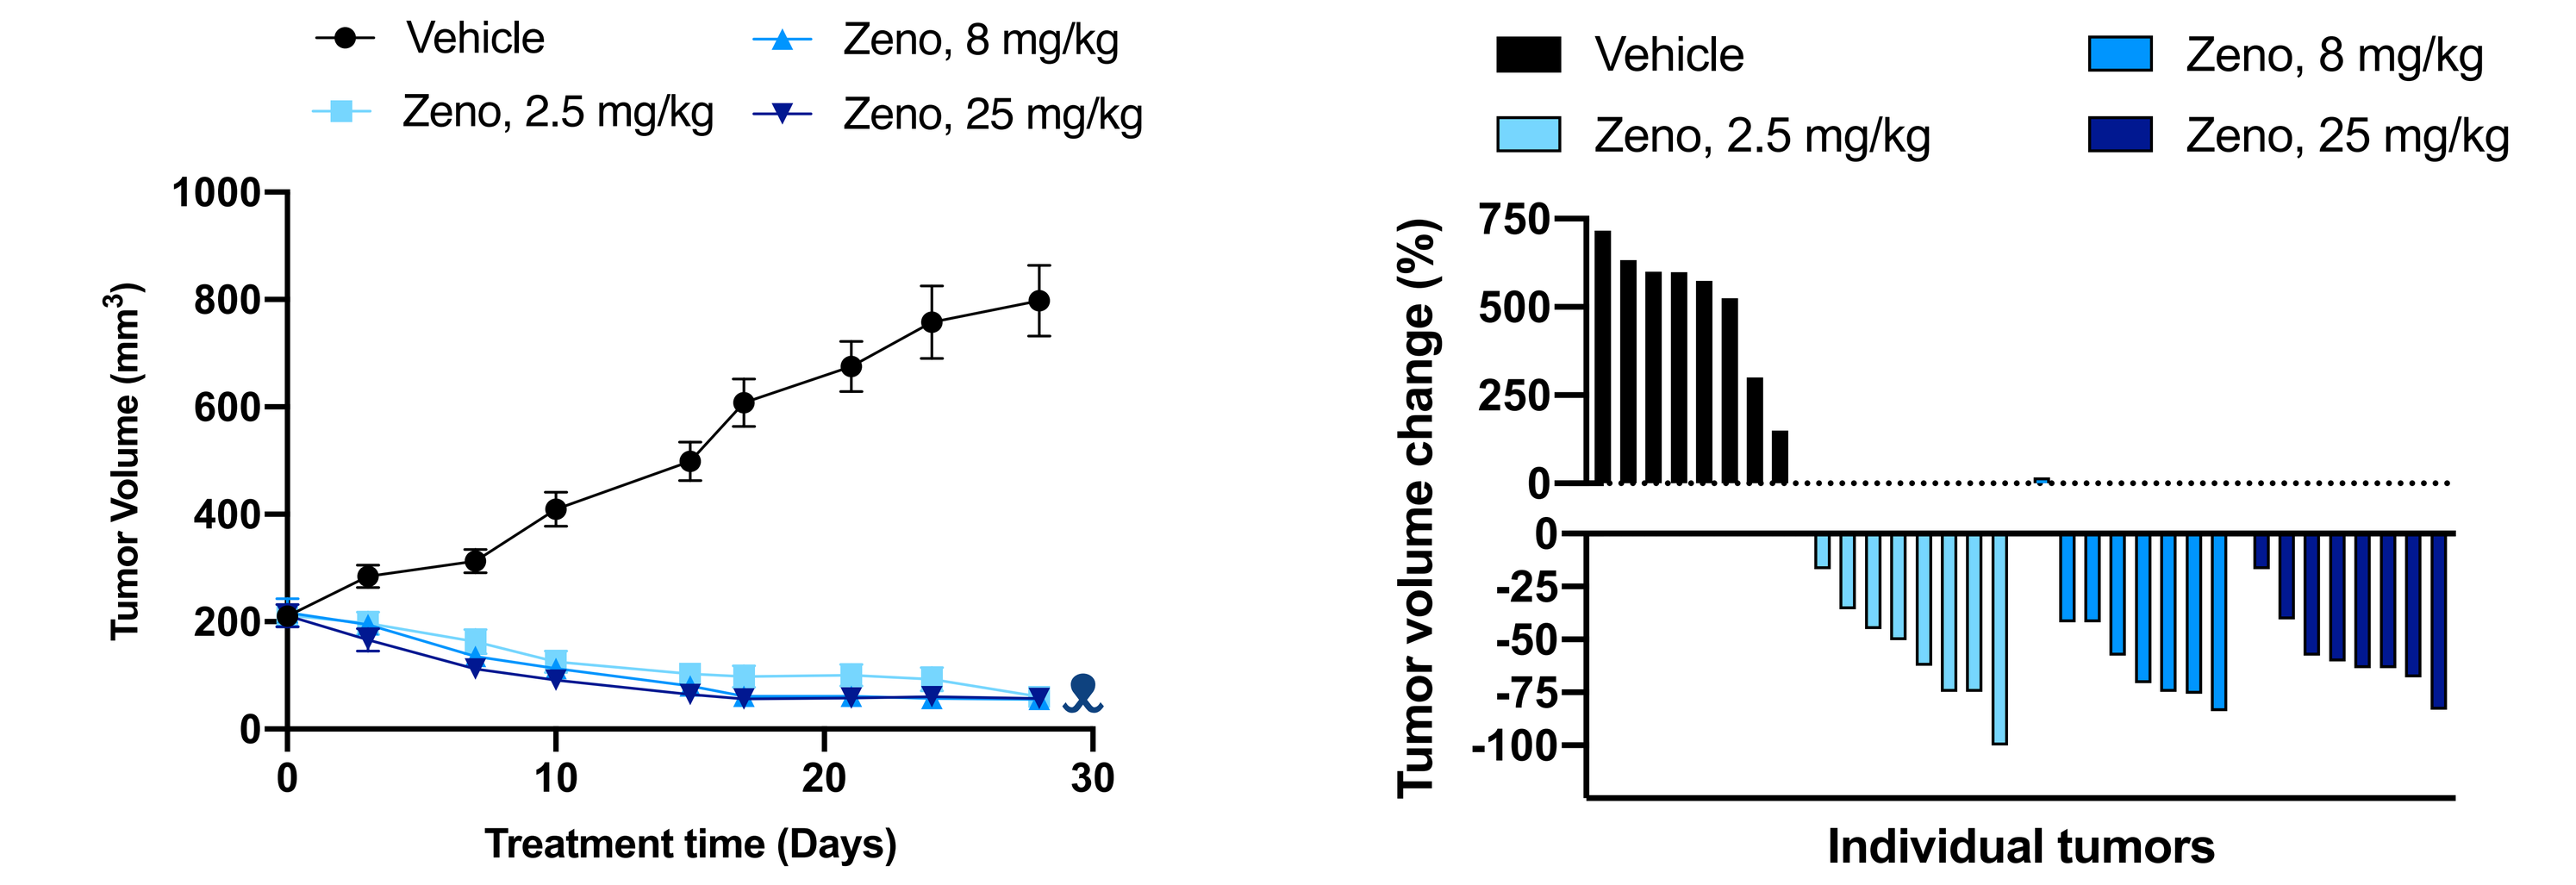
Vehicle
Zeno, 2.5 mg/kg
Zeno, 8 mg/kg
Zeno, 25 mg/kg
Tumor Volume (mm^3)
Treatment time (Days)
Tumor volume change (%)
Individual tumors

Figure 3. Left panels: Animals bearing the indicated PDX tumors (subcutaneous implantation) were treated with Zeno. Tumor volume was measured twice weekly. There were 5-10 mice per group and the data represent the mean ± SEM. Right panels: Tumor volume change (%) of individual tumors at the last day of measurement. : 25mg/kg Zeno in mice is comparable to the current clinical dose used in humans (750 mg Q2W).

Summary and Conclusion

- **Zeno blocks growth and causes death of NRG1-fusion positive cell lines.**
- **Zeno blocks HER3 phosphorylation and downstream signaling.**
- **Zeno induces expression of markers of apoptosis and cell cycle arrest and inhibits expression of cyclin D1.**
- **Treatment of NRG1-fusion positive patient-derived xenografts induces tumor shrinkage and durable tumor regression in multiple cancer types.**
- **These results further support the clinical development of Zeno by Merus as therapy for NRG1 fusion-driven cancers in the Phase 1/2 eNRGy trial (NCT02912949).**

Acknowledgment

This study was partially funded by grants from Merus and Cycle for Survival. *In vivo* studies with OV-10-0050 and ST3204 PDX models were performed by WuXi AppTec and XenoSTART, respectively.