AACR-NCI-EORTC Meeting 2019
Boston, MA, USA
October 26-30, 2019

#LB-C07

Preclinical evaluation of MCLA-129: a bispecific antibody targeting c-MET and EGFR

Cecile Geuijen[1], Mario di Matteo[2,3], Sarah Trusso Cafarello[2,3], Tristan Gallenne[1], Roy Nijhuis[1], Therese Visser[1], Willem Bartelink[1], Carina Bartelink-Clements[1], Vanessa Zondag-van der Zande[1], Eric Rovers[1], Karin de Cortie[1], Linda Kaldenberg-Hendriks[1], Berina Eppink[1], Rinse Klooster[1], John de Kruif[1], Massimiliano Mazzone[2,3], Mark Throsby[1]

[1]Merus N.V. Utrecht, The Netherlands; [2]Laboratory of Tumor Inflammation and Angiogenesis, Center for Cancer Biology, VIB, Leuven, Belgium; [3]Laboratory of Tumor Inflammation and Angiogenesis, Center for Cancer Biology, Department of Oncology, KU Leuven, Belgium.

BACKGROUND & RATIONALE

- EGFR and c-MET activate the same signalling pathways to drive proliferation, survival and invasion.
- MET/HGF amplification has a potential role in resistance to EGFR targeted treatment (e.g. NSCLC, GBM)[1].
- In NSCLC, resistance to TKI inhibition of mutant EGFR is associated with c-MET signalling[2].
- In patients treated with EGFR-TKIs, high circulating HGF predicted poor prognosis.

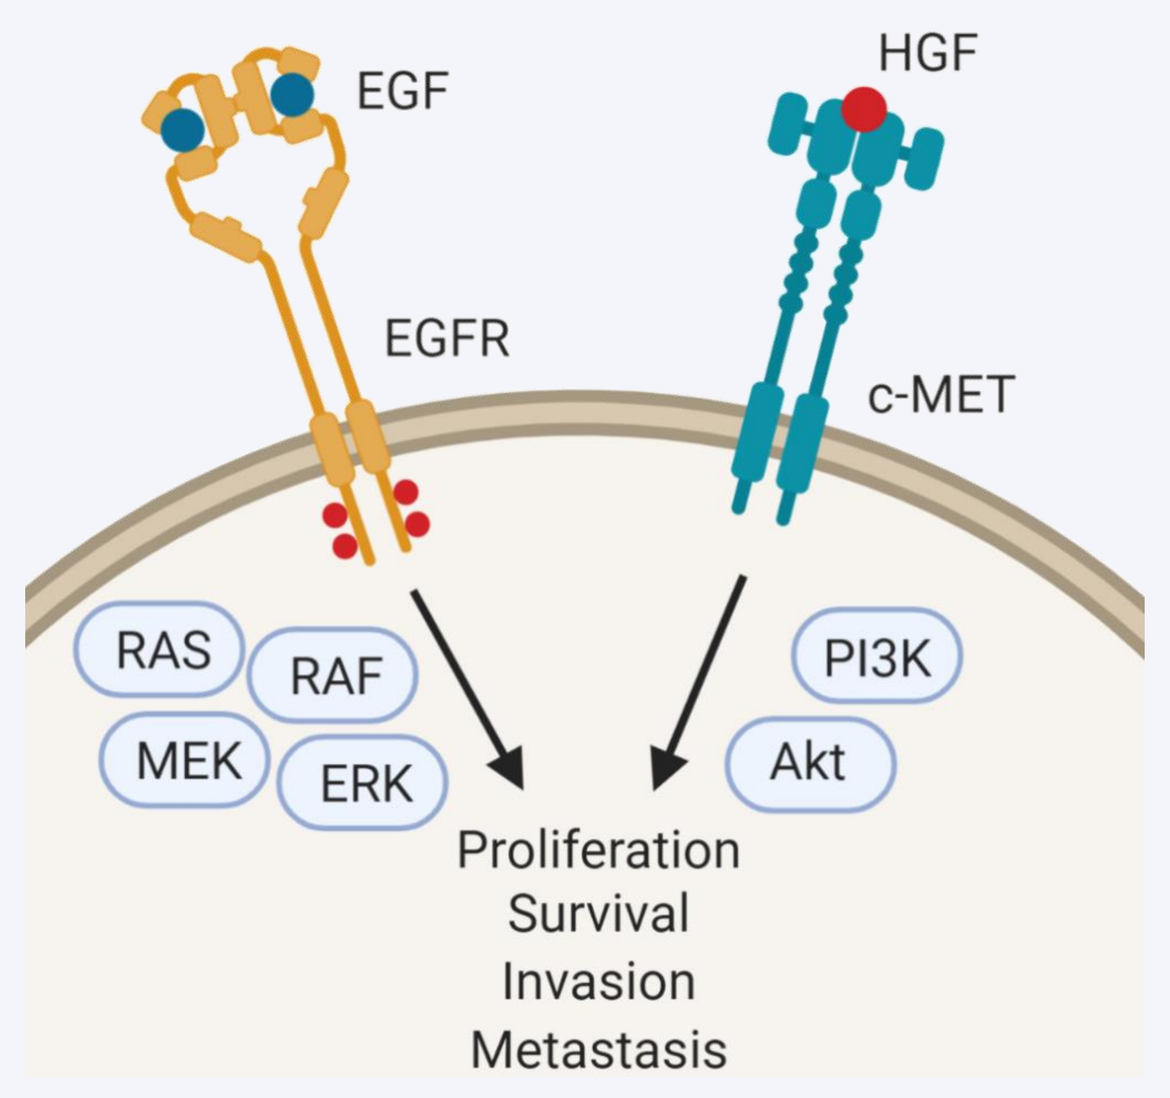
EGF
HGF
EGFR
c-MET
RAS
RAF
MEK
ERK
PI3K
Akt
Proliferation
Survival
Invasion
Metastasis

THE BICLONICS® PLATFORM

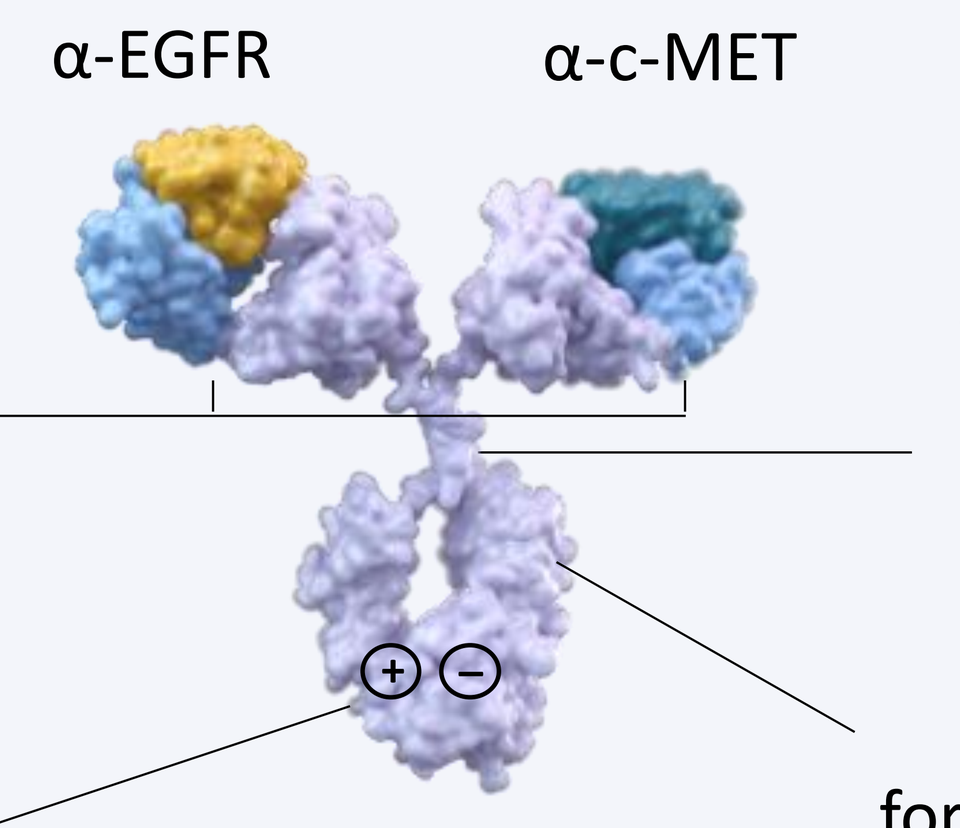
α-EGFR
α-c-MET

Common Light Chain for 'unforced', natural pairing with 2 different heavy chains

IgG Format for efficient manufacturing and predictable *in vivo* behavior

Electrostatic attraction to efficiently drive formation of Biclonics®

Fc Modifications for improved functionality (ADCC or silencing)

UNBIASED SCREENING

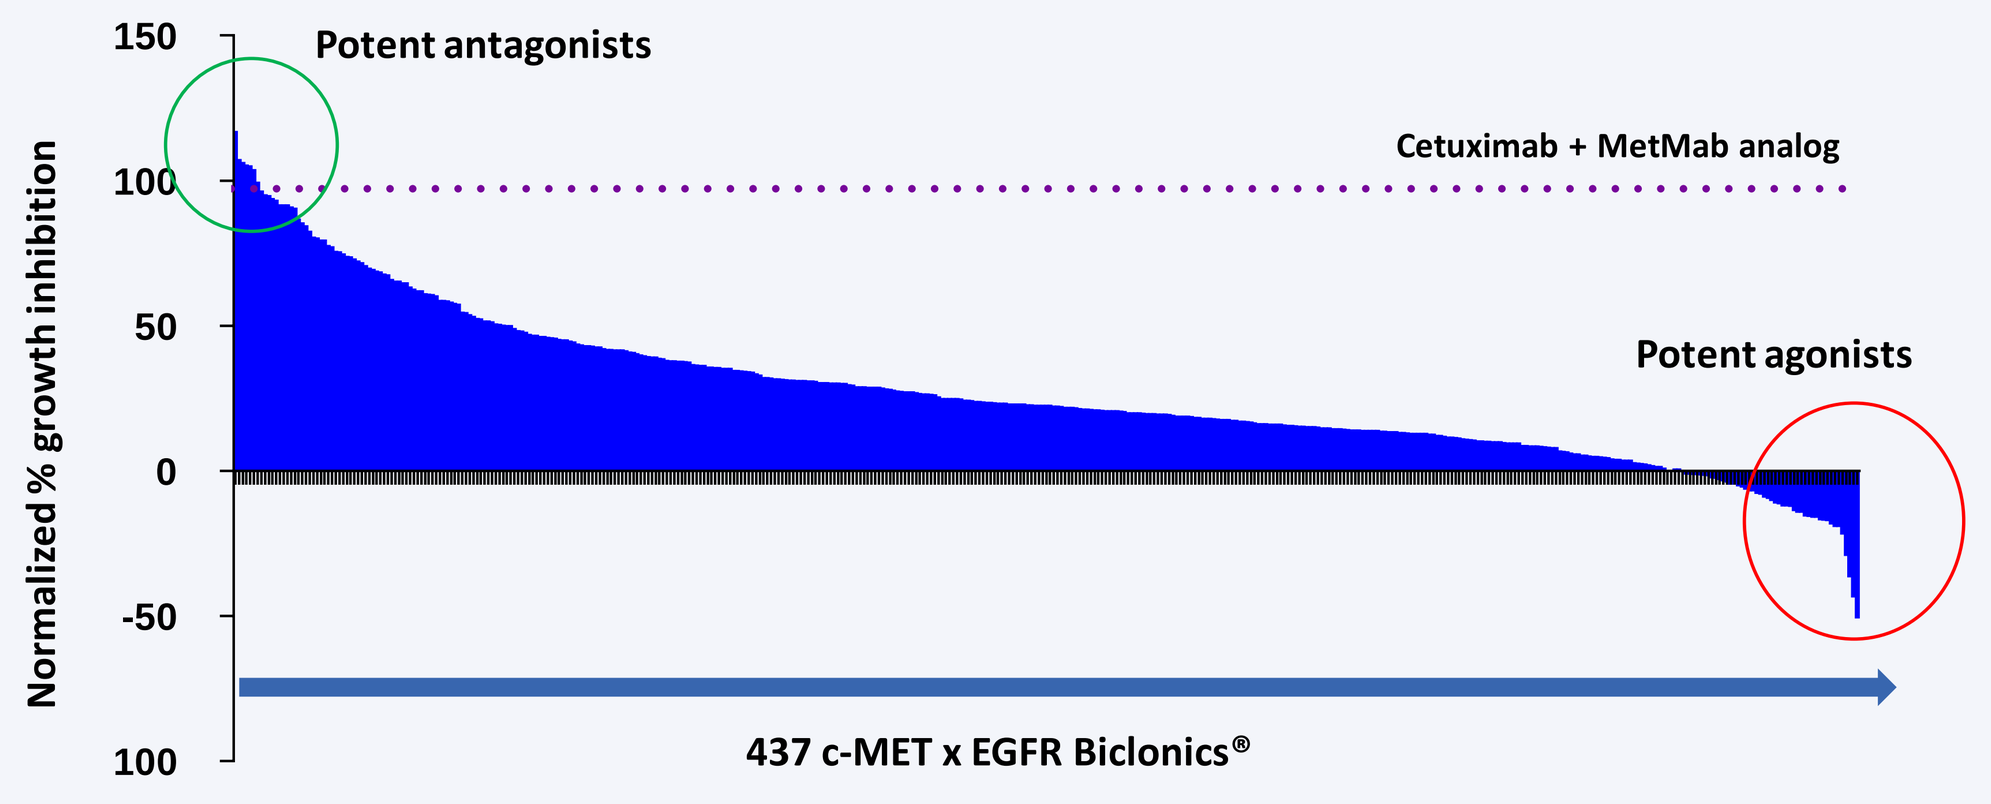
Potent antagonists
Cetuximab + MetMab analog
Potent agonists
Normalized % growth inhibition
437 c-MET x EGFR Biclonics®
N87 cells
Starvation medium + EGF/HGF
•Test antibodies
•Cetuximab + MetMab analog
Alamar Blue readout for proliferation

MCLA-129 SELECTION & CHARACTERISTICS

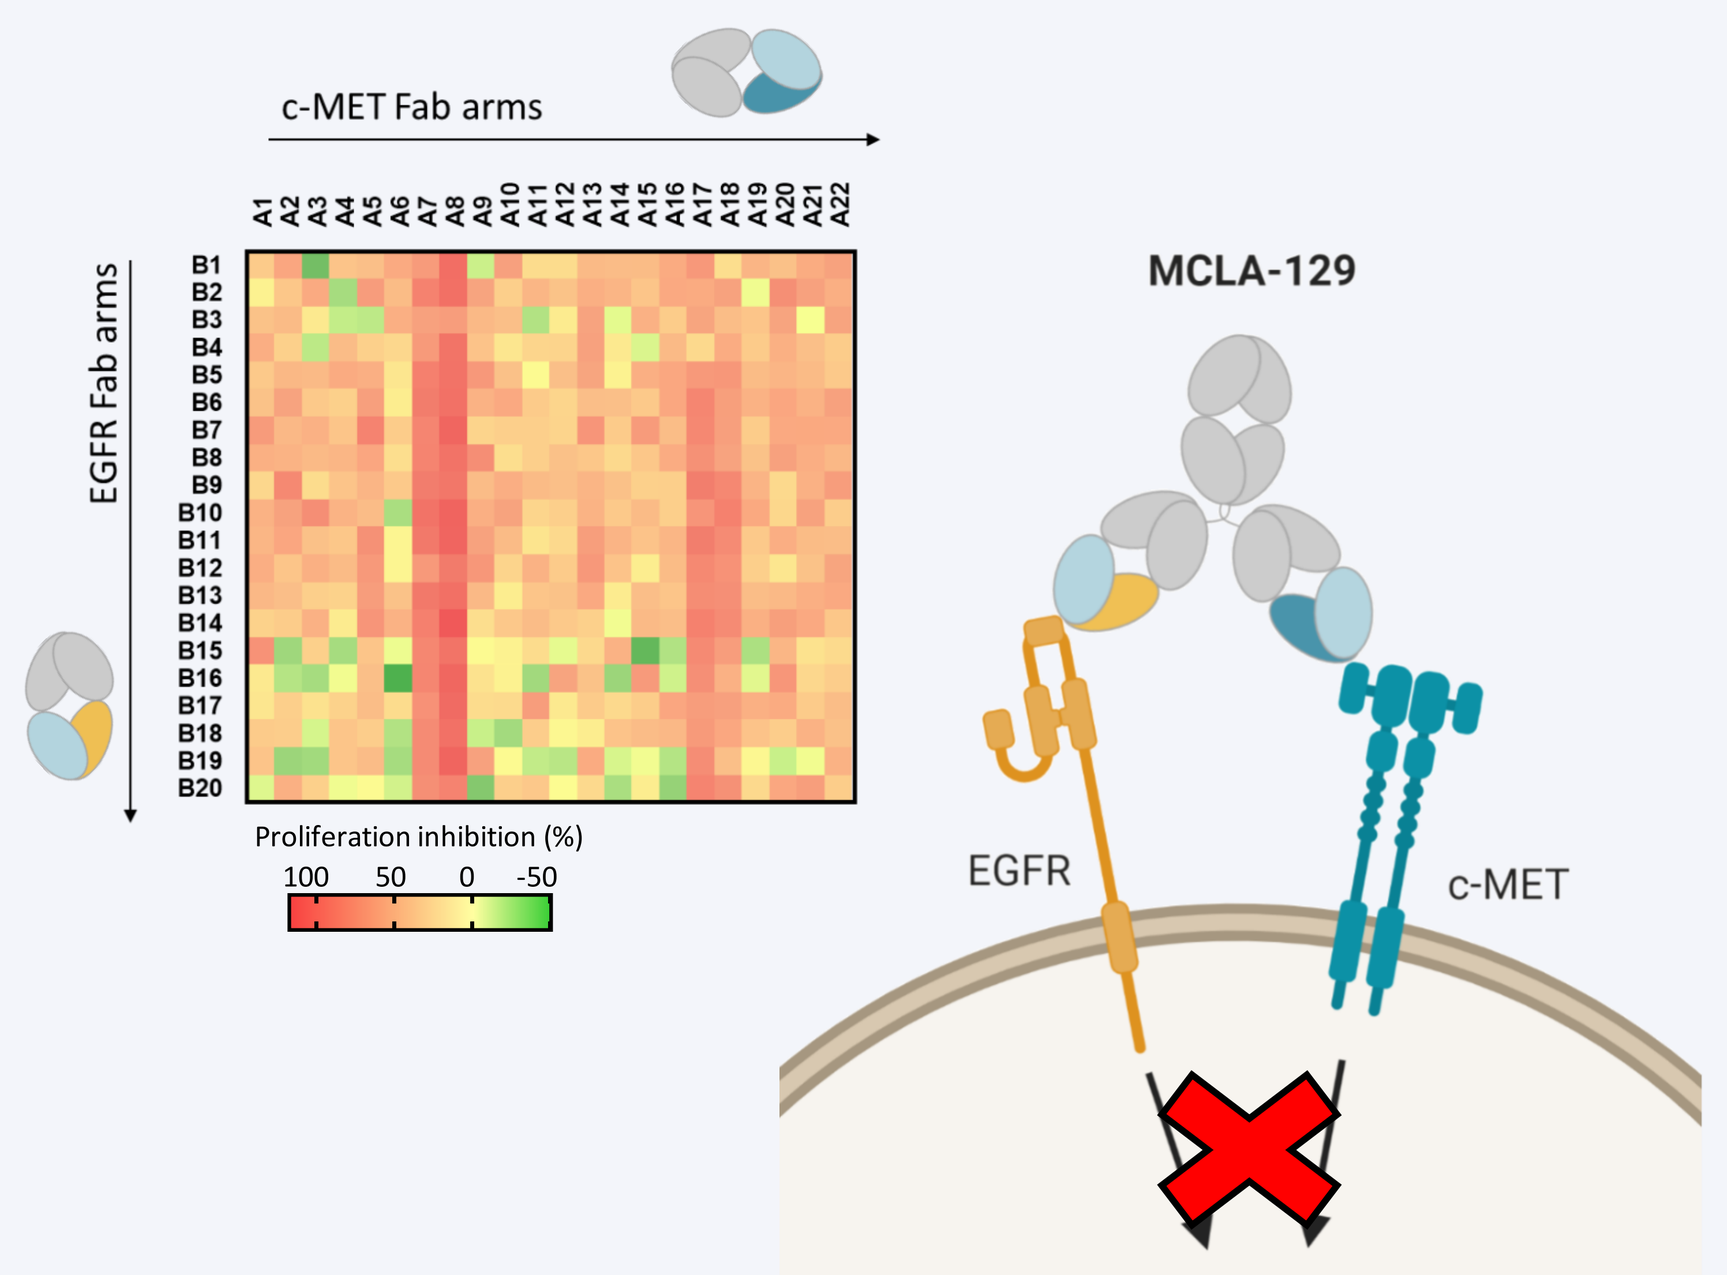
c-MET Fab arms
EGFR Fab arms
Proliferation inhibition (%)
100 50 0 -50
MCLA-129
EGFR
c-MET

- Specific combinations of c-MET and EGFR Fab arms in the Biclonics® format result in either potent antagonistic or agonistic activity.
- MCLA-129 selected from a panel of 8 Biclonics® based on potency of c-MET inhibition.
- MCLA-129 competes with the ligand binding domains of EGFR and c-MET.

c-METxEGFR BICLONICS® REVERSE HGF RESISTANCE

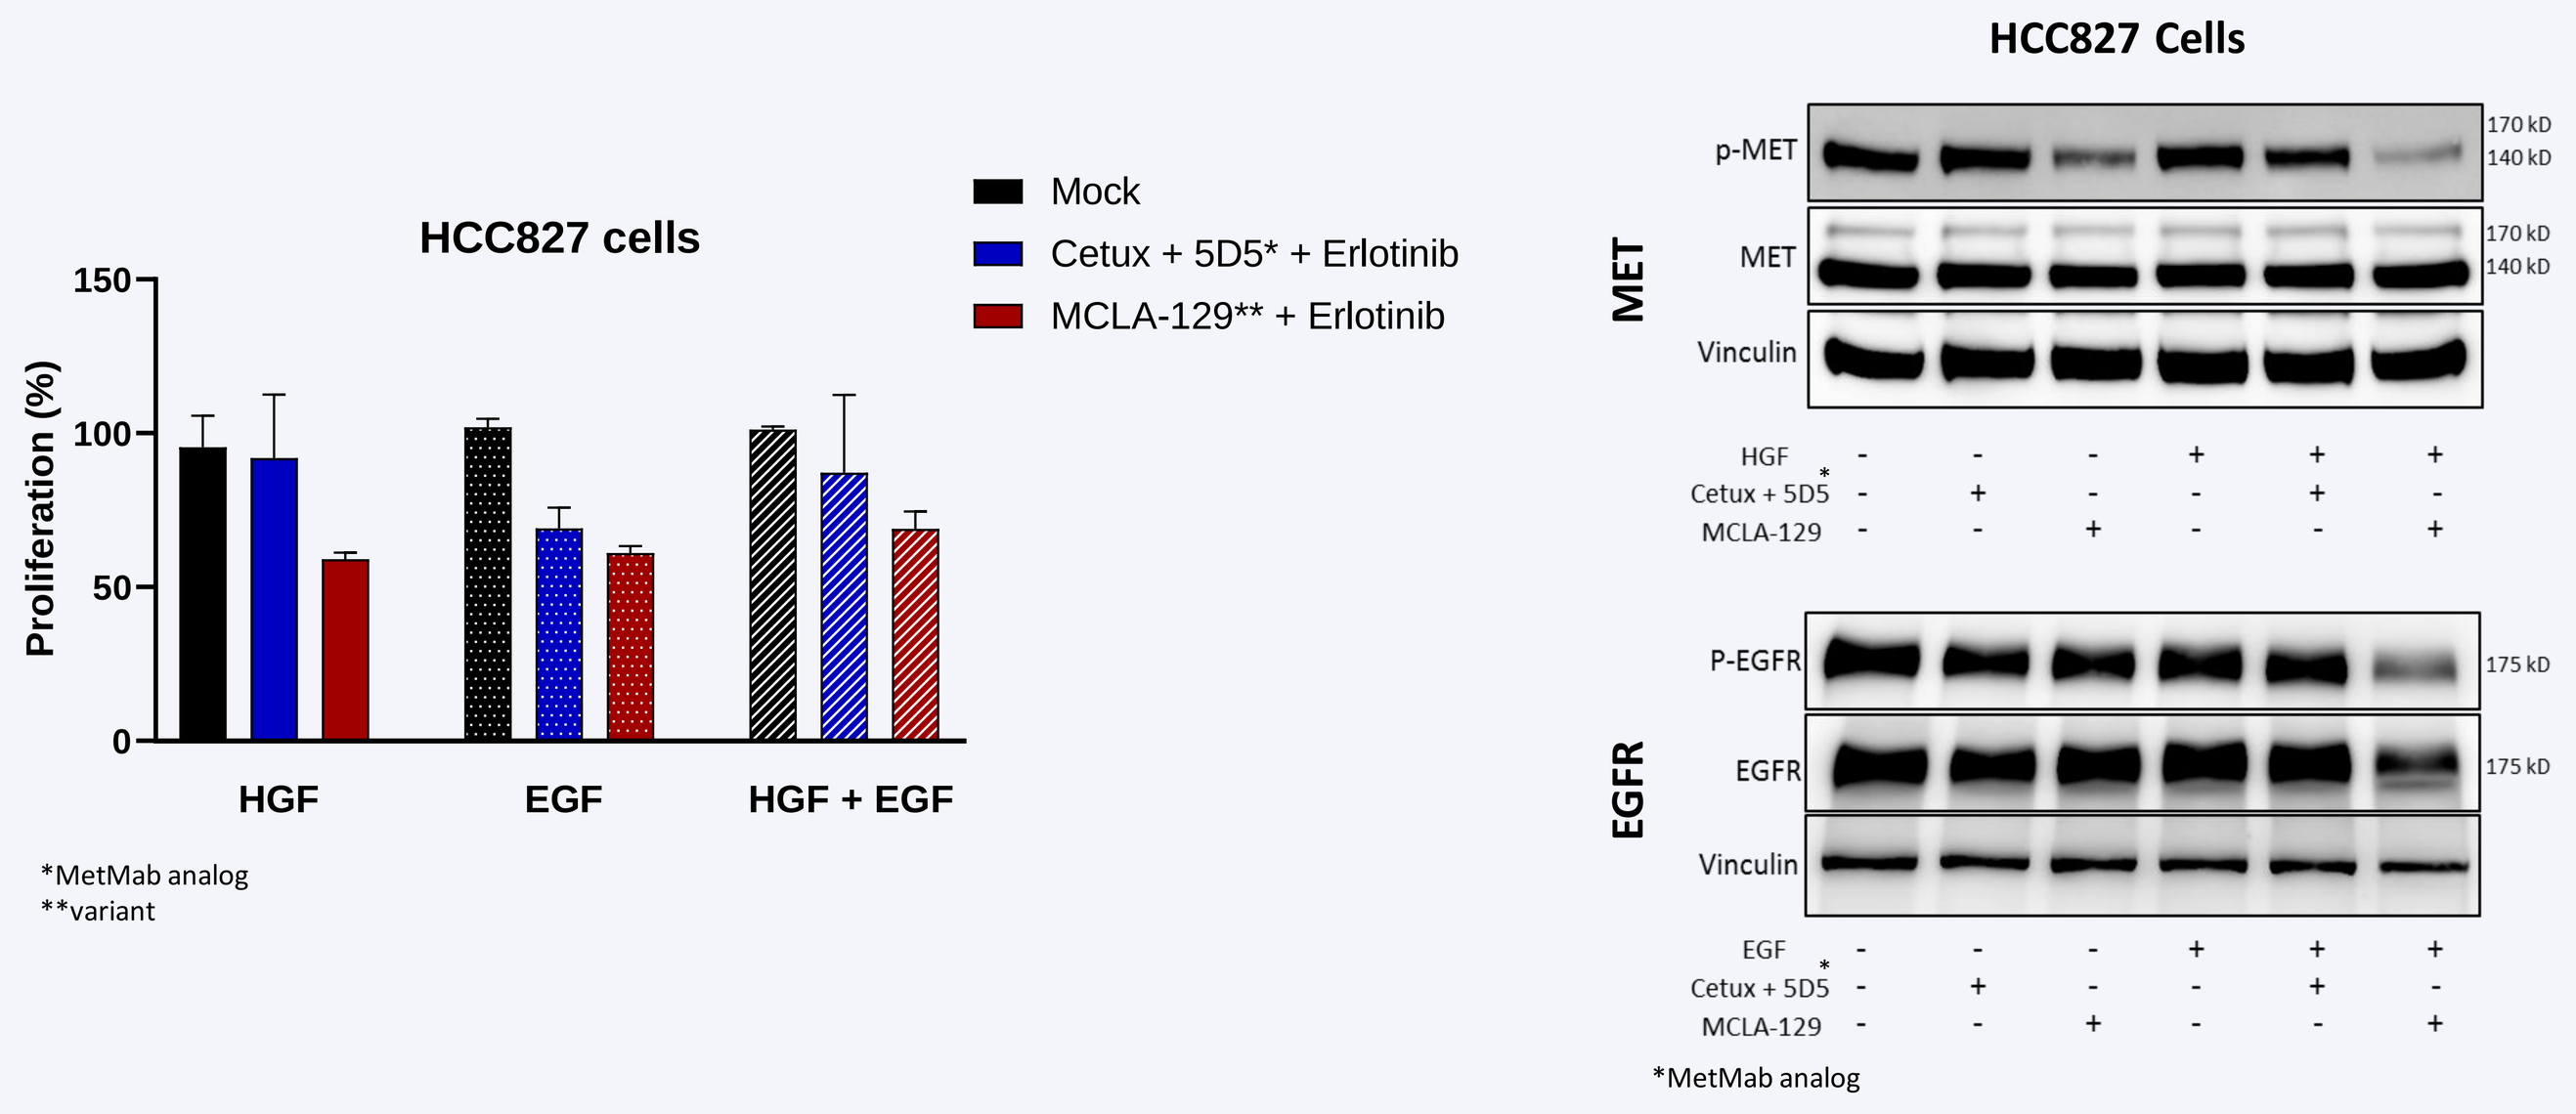
HCC827 cells
Mock
Cetux + 5D5* + Erlotinib
MCLA-129** + Erlotinib
Proliferation (%)
HGF
EGF
HGF + EGF
*MetMab analog
**variant
HCC827 Cells
p-MET
MET
Vinculin
P-EGFR
EGFR
Vinculin
*MetMab analog

- Combination of EGF and HGF reverses TKI inhibition in both NSCLC cell lines.
- MCLA-129 restores sensitivity of NSCLC cell lines to Erlotinib.
- MCLA-129 inhibits HGF and EGF induced phosphorylation of receptors.
- MCLA-129 is more potent in HCC827 cells than Cetuximab + MetMab analog (5D5).

INFLUENCE OF RECEPTOR DENSITY ON ADCC

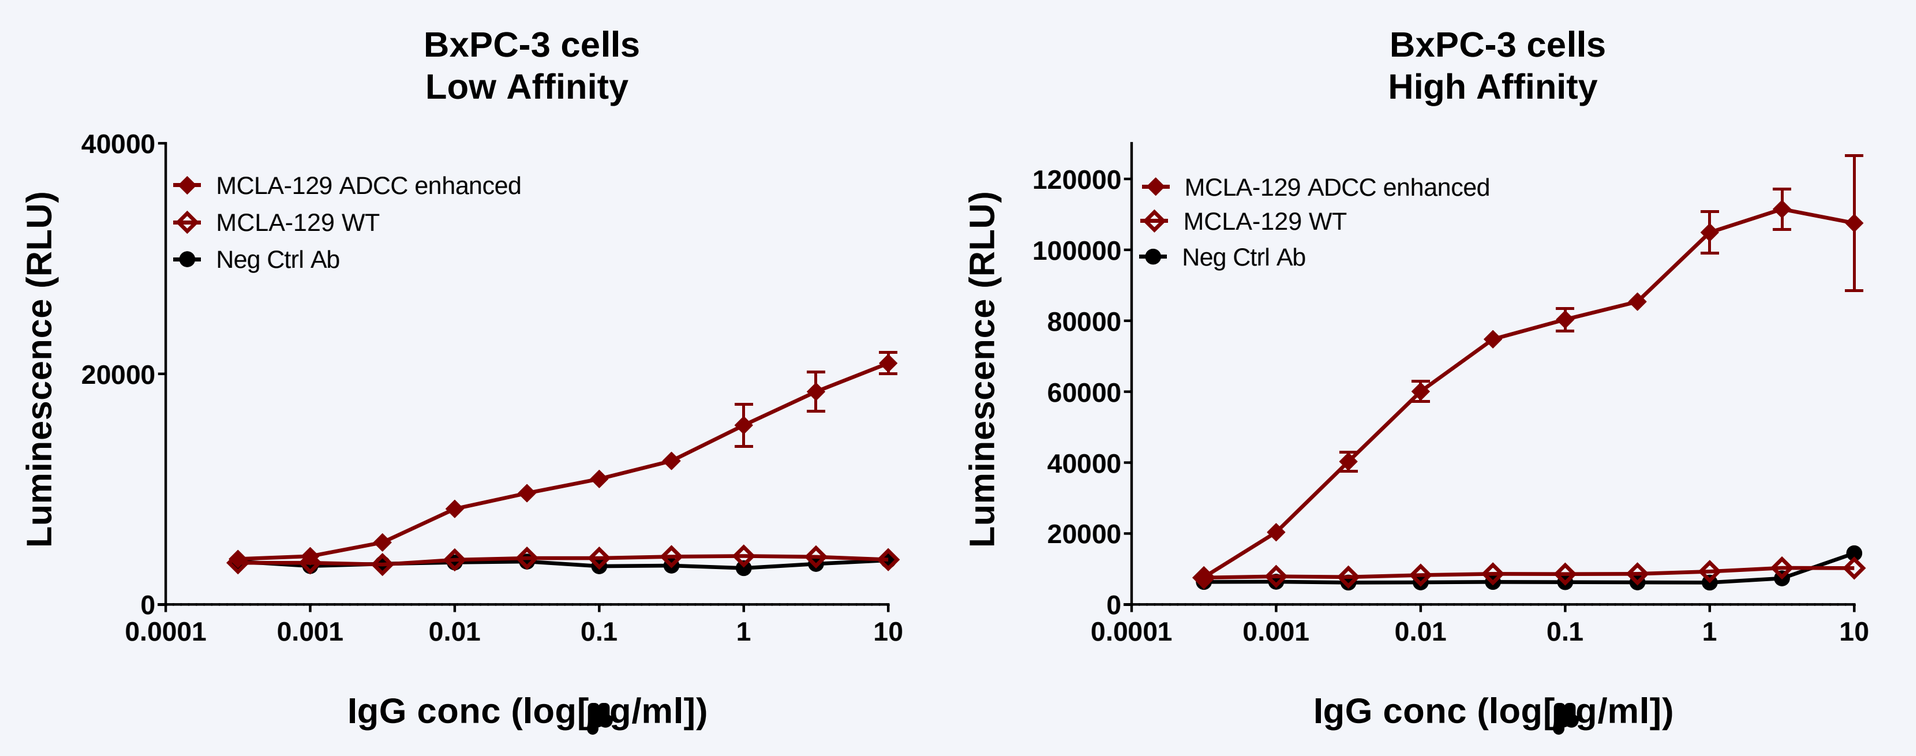
BxPC-3 cells
Low Affinity
BxPC-3 cells
High Affinity
MCLA-129 ADCC enhanced
MCLA-129 WT
Neg Ctrl Ab
Luminescence (RLU)
IgG conc (log[µg/ml])

- Afucosylation of MCLA-129 is required for ADCC activity.
- Cells expressing both EGFR and c-MET, i.e. BxPC-3, are preferentially targeted by ADCC.

MCLA-129 INHIBITS TKI RESISTANT NSCLC

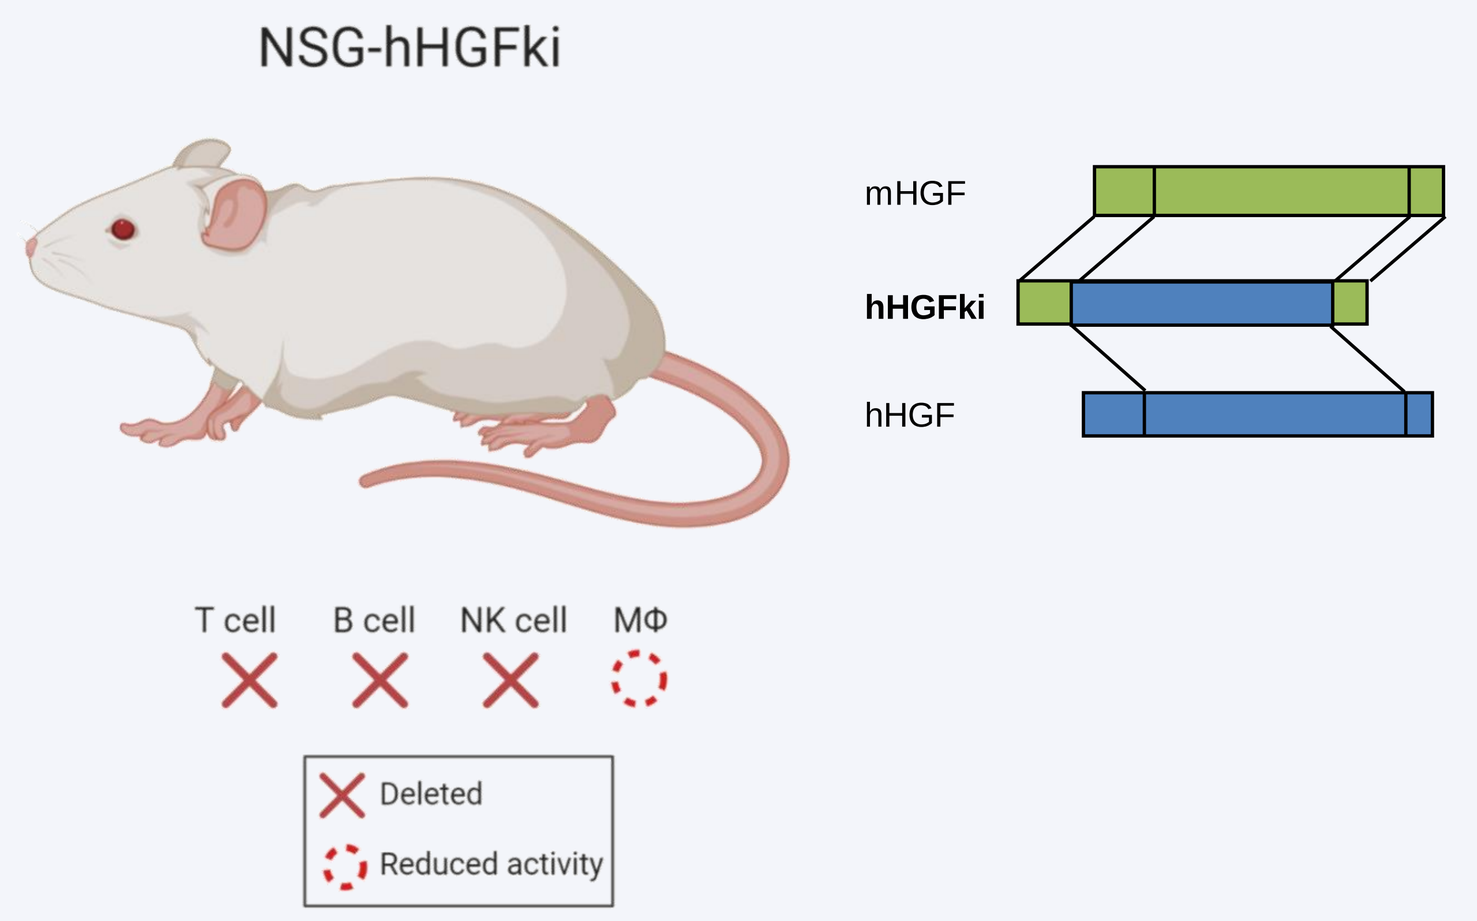
NSG-hHGFki
mHGF
hHGFki
hHGF
T cell
B cell
NK cell
MΦ
Deleted
Reduced activity

- HCC827 tumor cells were engrafted into immunodeficient NOD SCID gamma (NSG) human hepatocyte growth factor knock-in (hHGFki) mice, that express human HGF in place of endogenous mouse HGF (NSG-hHGFki mice, stk#014553)[3].
- The mice were treated with: Erlotinib (6 mg/kg) once daily (QD) and antibodies/ antibodies combinations (25mg/kg) weekly.

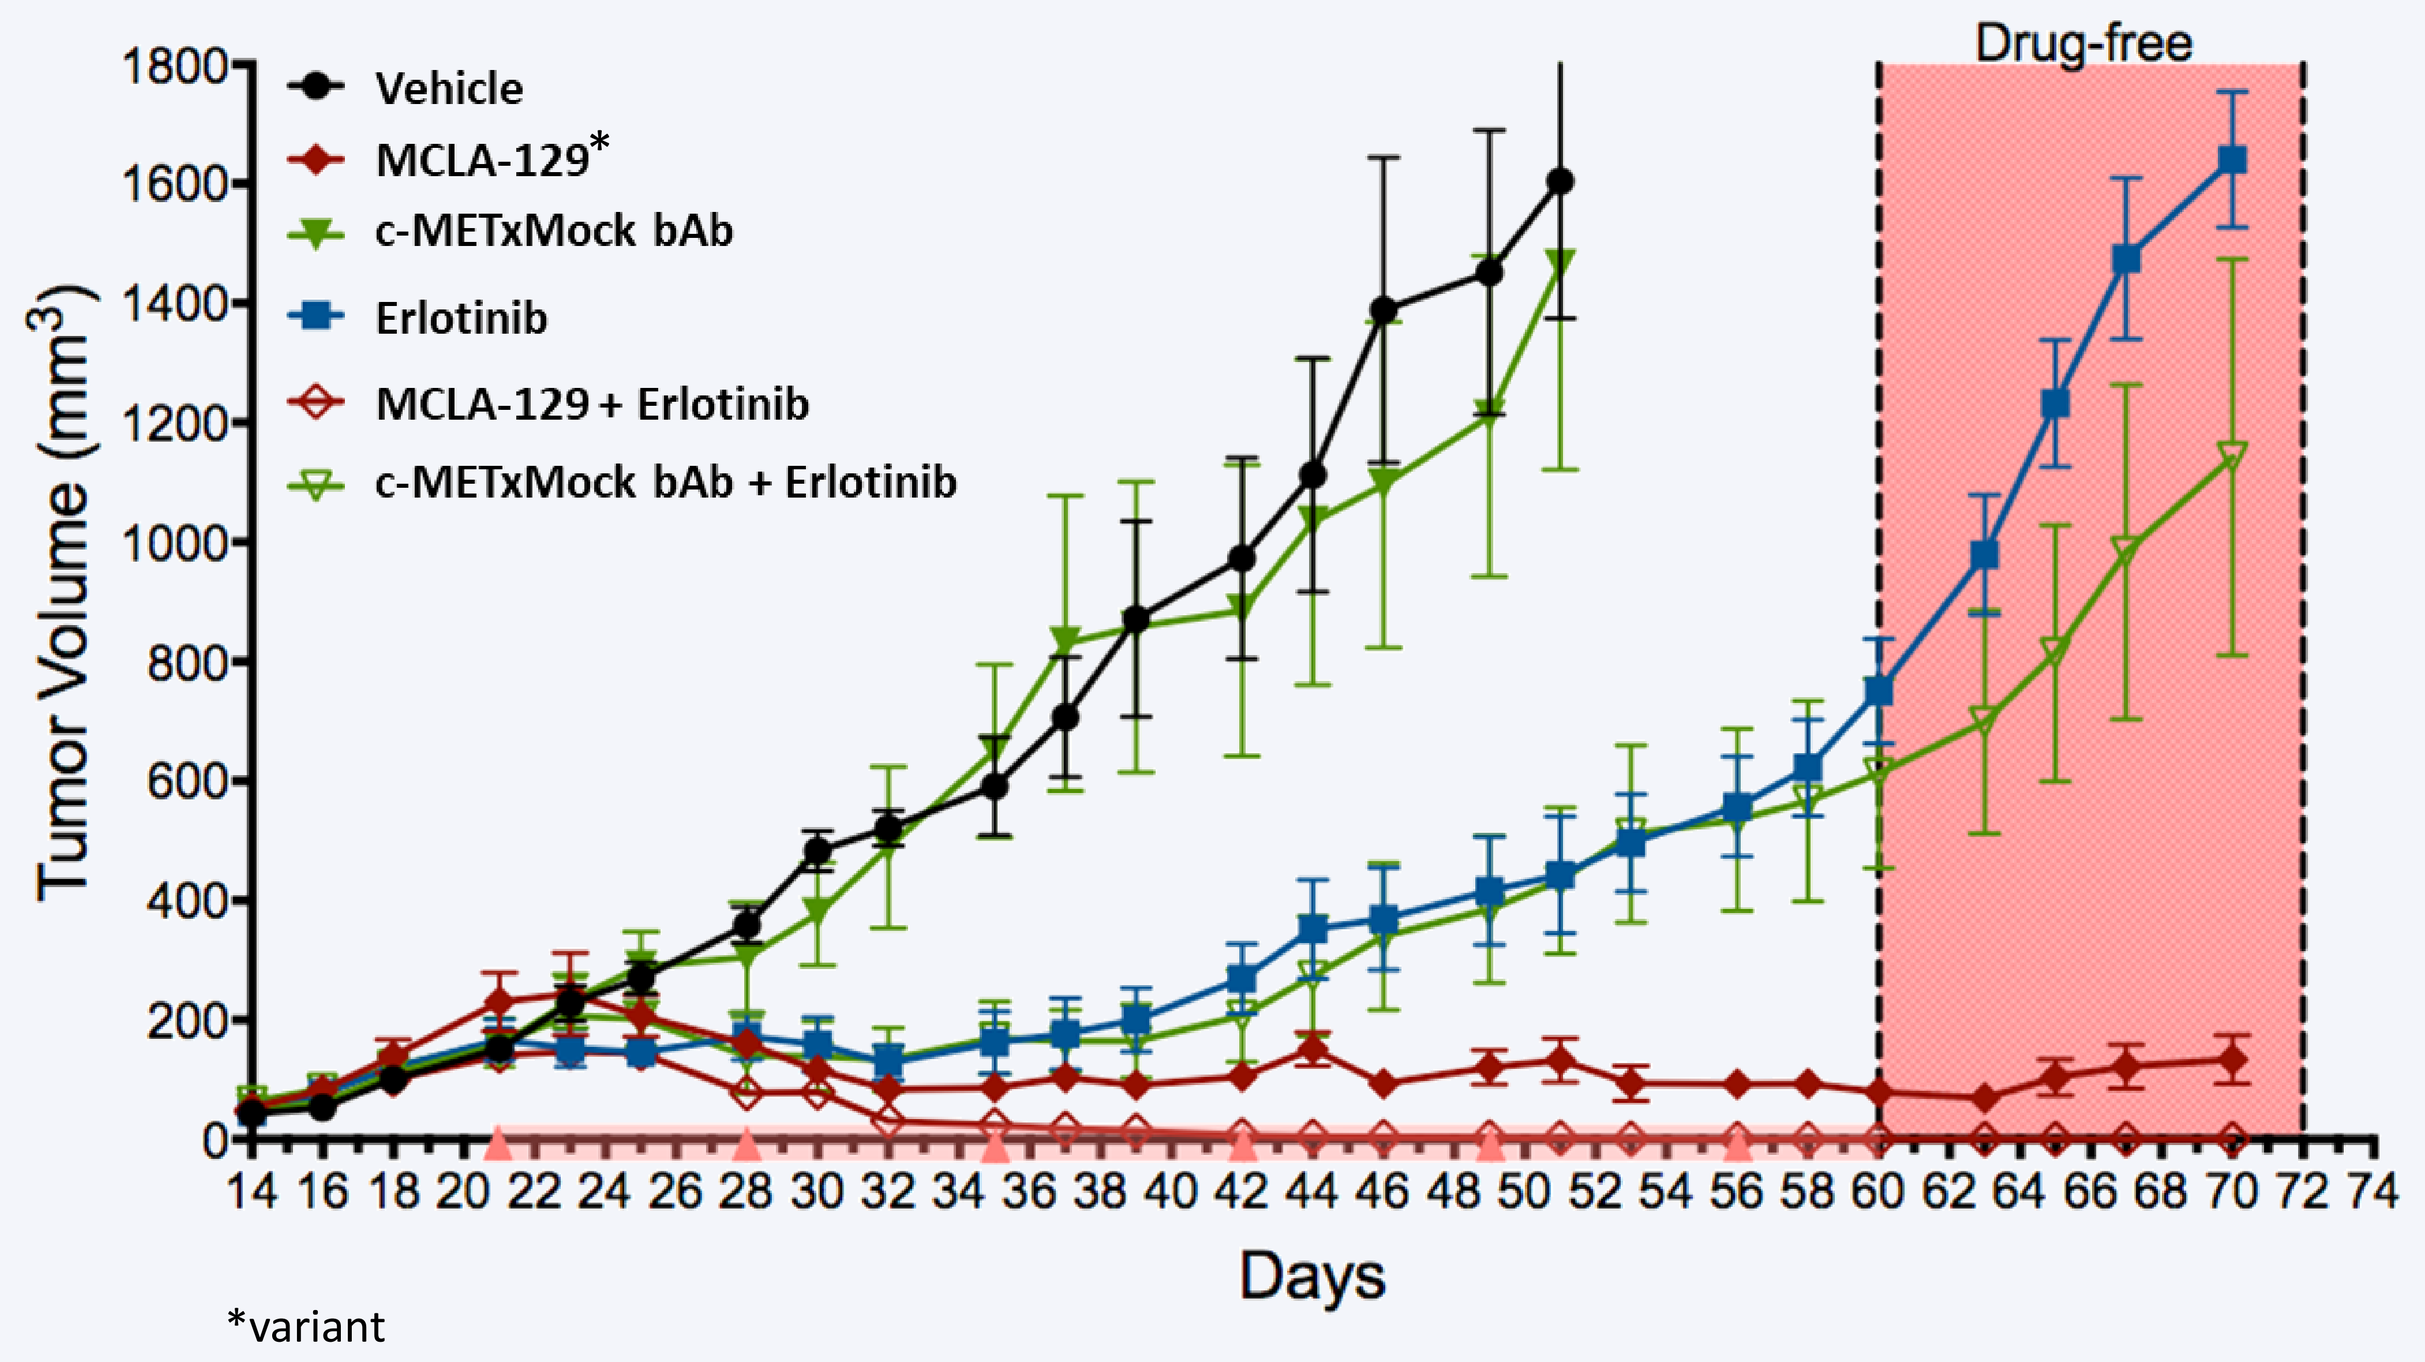
Drug-free
Vehicle
MCLA-129*
c-METxMock bAb
Erlotinib
MCLA-129 + Erlotinib
c-METxMock bAb + Erlotinib
Tumor Volume (mm³)
Days

*variant

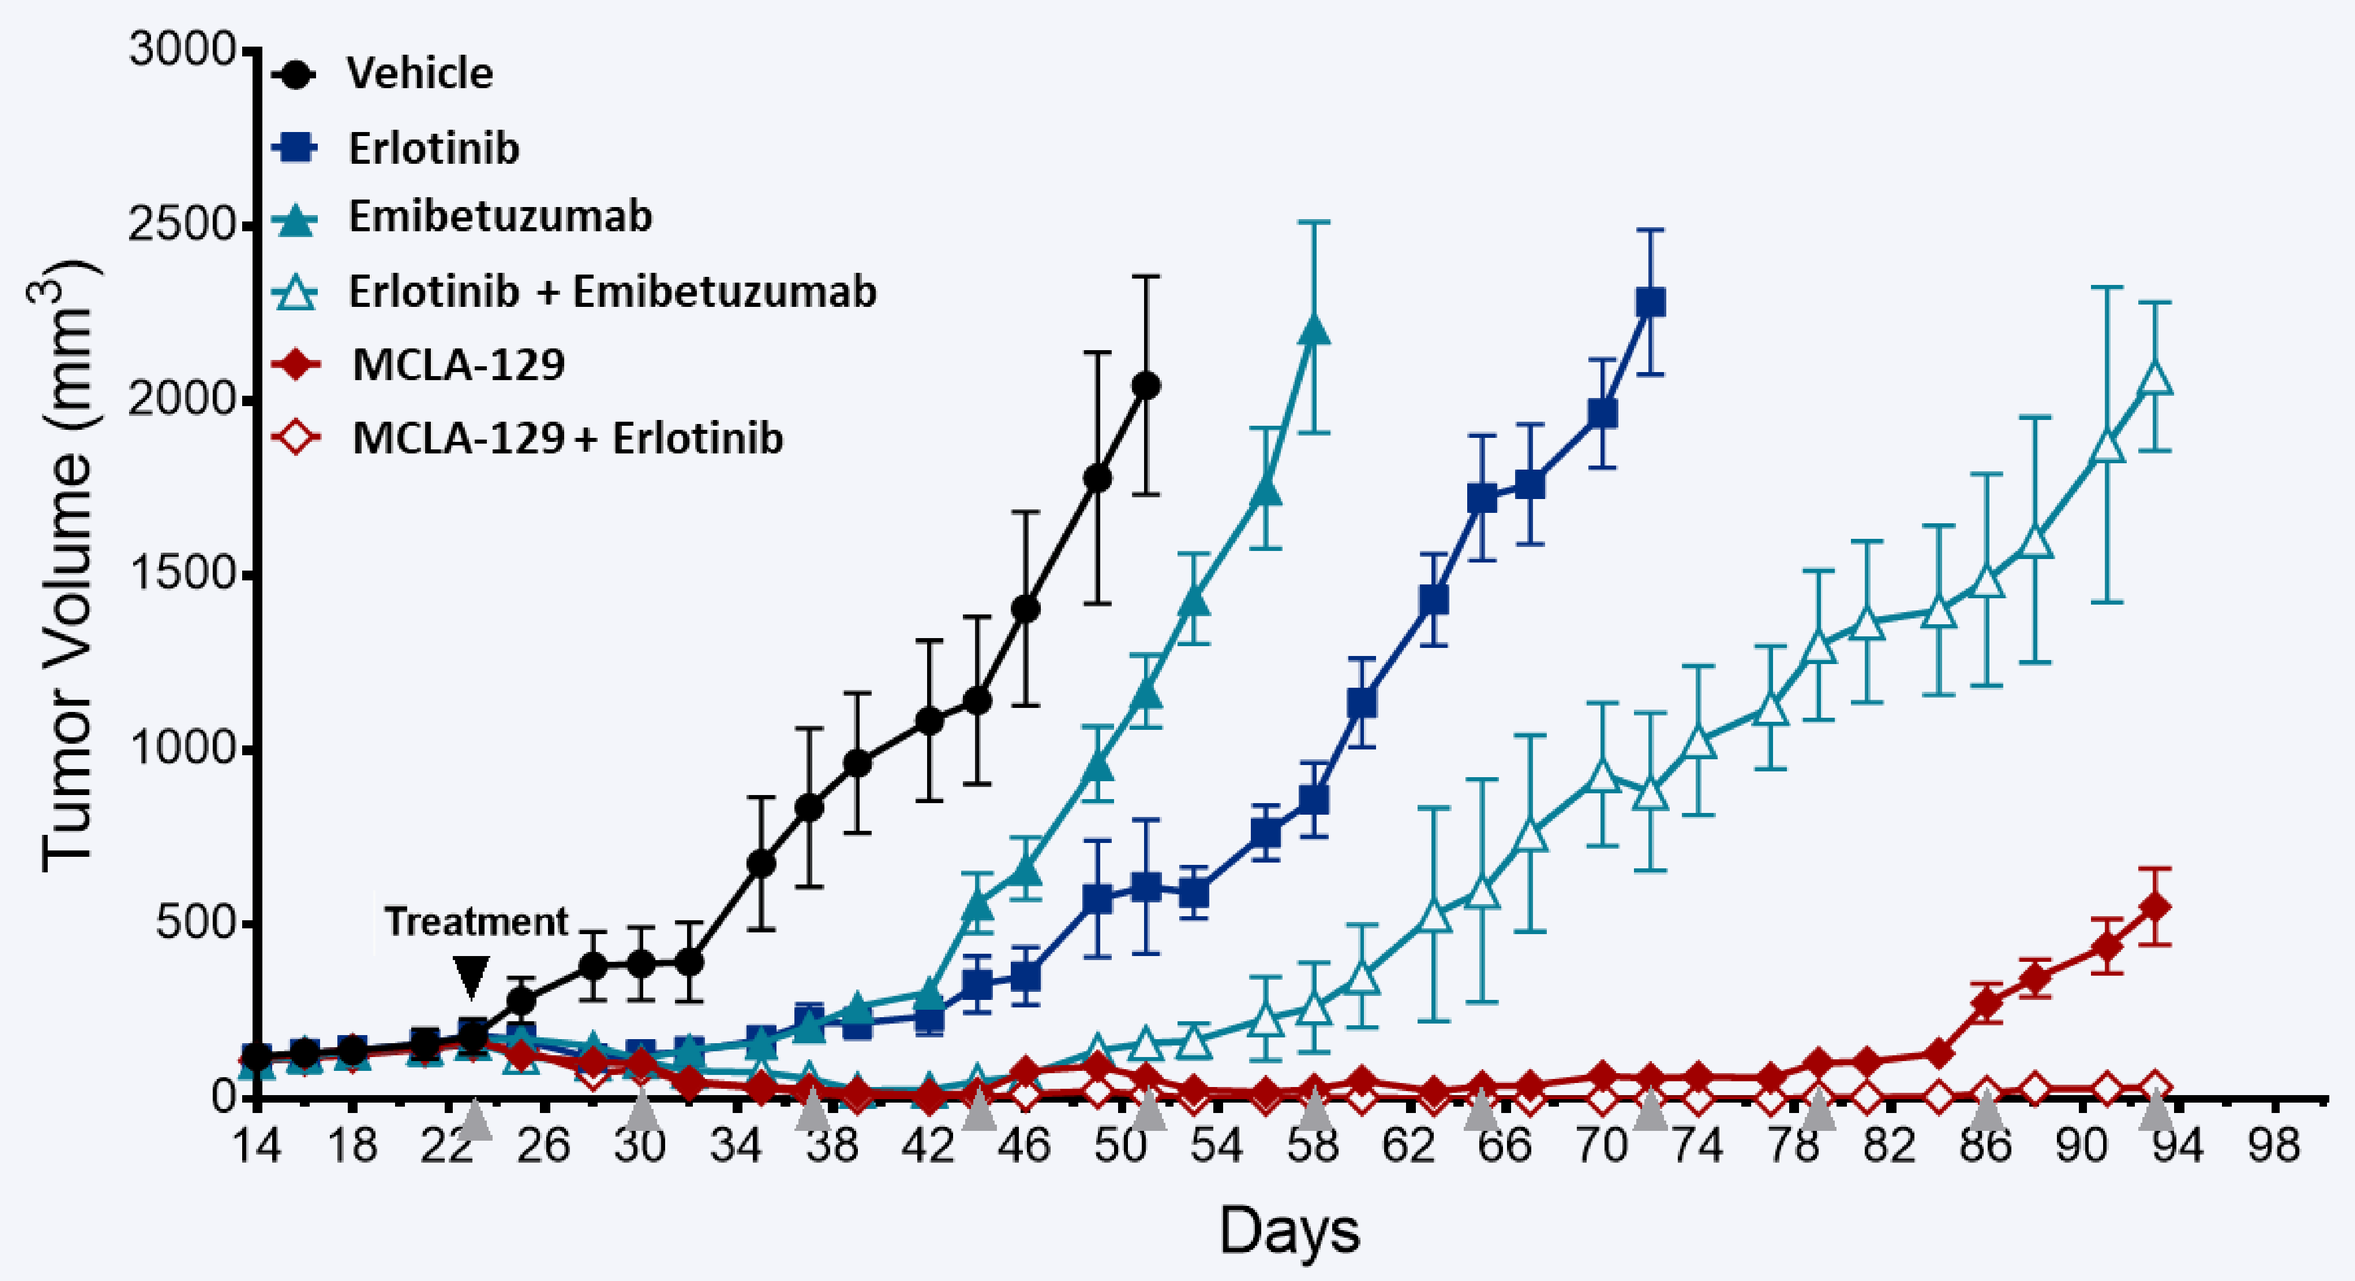
Vehicle
Erlotinib
Emibetuzumab
Erlotinib + Emibetuzumab
MCLA-129
MCLA-129 + Erlotinib
Treatment
Tumor Volume (mm³)
Days

- Treatment with MCLA-129 led to sustained HCC827 growth inhibition both alone or when combined with Erlotinib and this activity was maintained after treatment was stopped.
- Inhibition of tumor growth induced by MCLA-129 was greater than that induced by the anti-c-MET benchmark antibody Emibetuzumab.
- The combination treatment of MCLA-129 with Erlotinib fully inhibited HCC827 tumor growth up to 94 days of treatment in contrast to all other treatments.

MCLA-129 REVERSES ACQUIRED TKI RESISTANCE

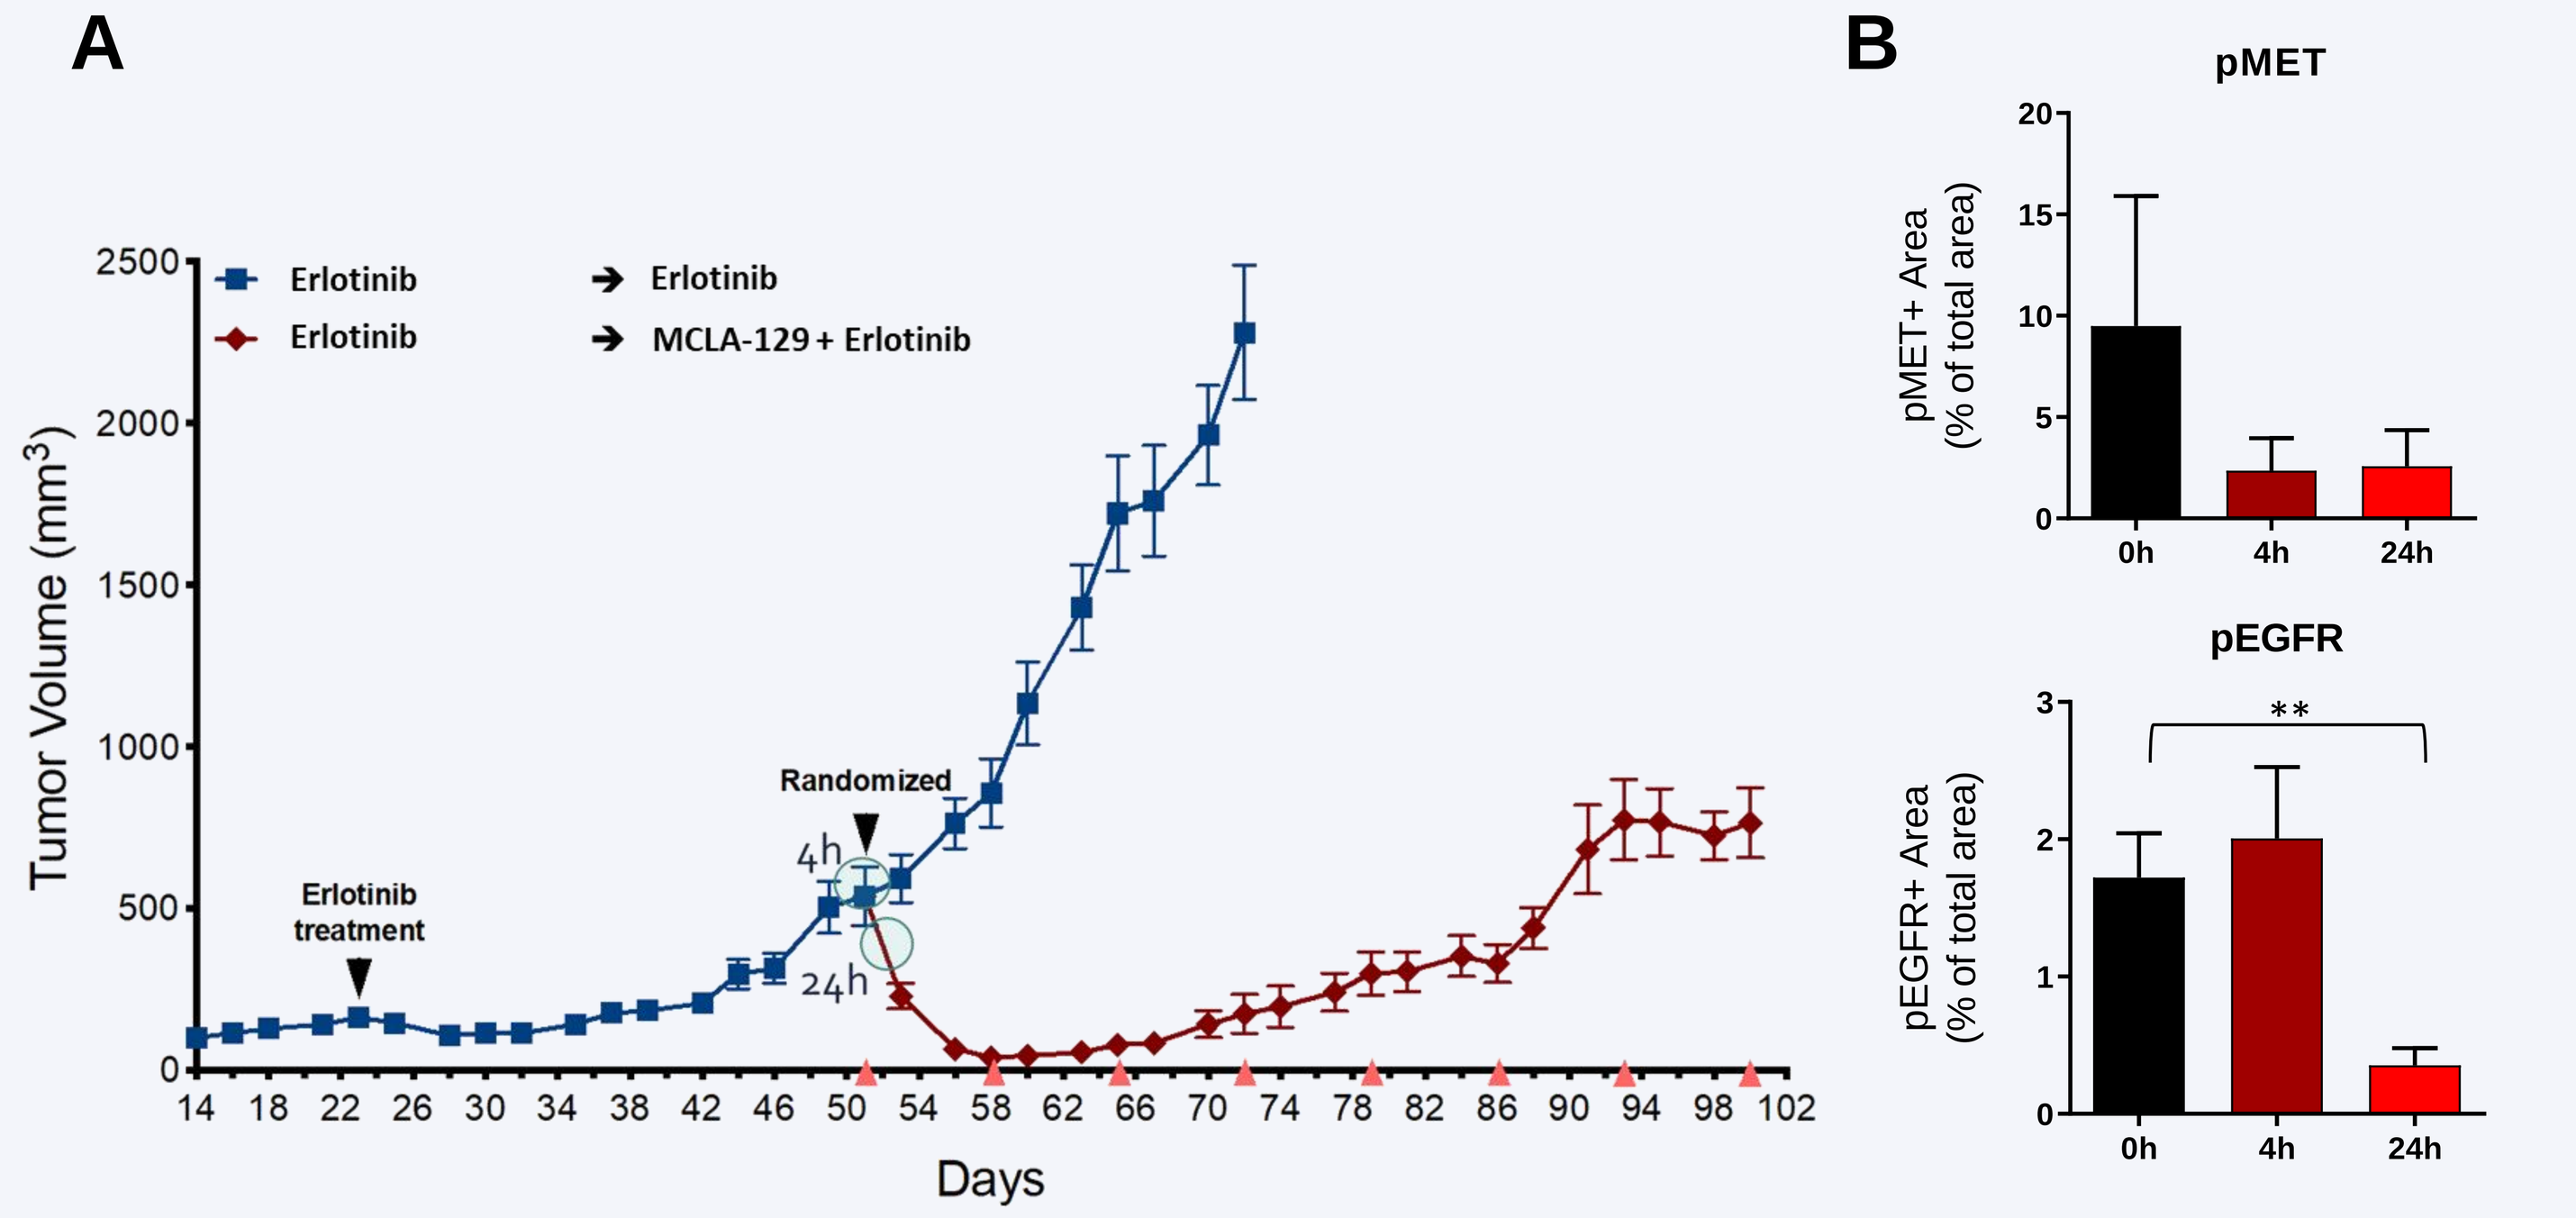
A
Erlotinib
Erlotinib
→ Erlotinib
→ MCLA-129 + Erlotinib
Erlotinib treatment
Randomized
4h
24h
Tumor Volume (mm³)
Days
B
pMET
pMET+ Area (% of total area)
pEGFR
pEGFR+ Area (% of total area)
0h
4h
24h

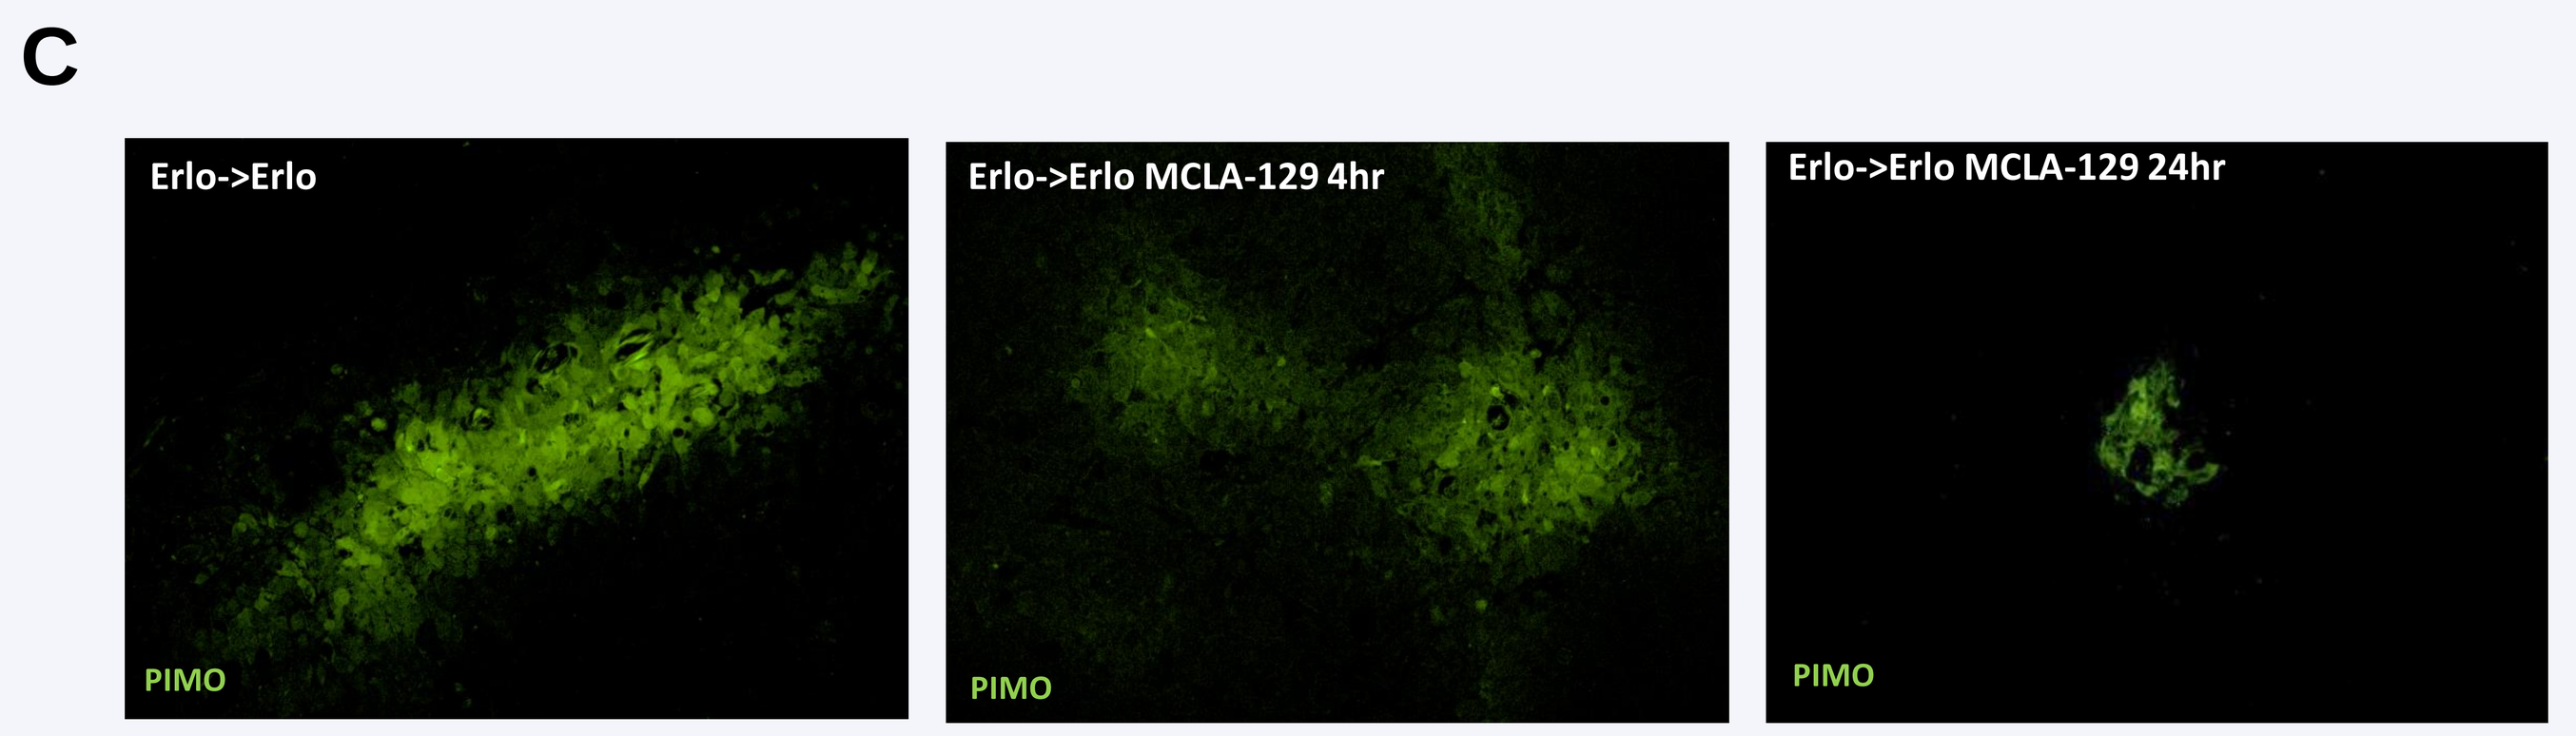
C
Erlo->Erlo
Erlo->Erlo MCLA-129 4hr
Erlo->Erlo MCLA-129 24hr
PIMO

PIMO: pimonidazole staining (representing hypoxic regions within the tumor)

*(A) Tumor volume (circles), (B) % of phosphorylated receptors in tumor sections, and (C) hypoxic tumor area in mice sacrificed: 2hr post Erlotinib 6mg/kg QD treatment (0h) or 4h and 24h after MCLA-129 administration and 2hr after Erlotinib 6mg/kg QD treatment. *p<0.05.*

- hHGFKi mice engrafted with HCC827 tumors and treated daily with Erlotinib exhibited rapid tumor growth after a period of control; at day 51 (tumors >500 mm³) mice were randomized to continue Erlotinib treatment alone or in combination with MCLA-129.
- In mice receiving the combination of Erlotinib and MCLA-129 tumor volume decreased dramatically (A) and was associated with inhibition of EGFR and c-MET phosphorylation (B) and a decrease in the hypoxic tumor area (C).

KEY FINDINGS AND CONCLUSIONS

- MCLA-129 is a high-affinity, ADCC-enhanced cLC Biclonics® targeting human EGFR and c-MET.
- MCLA-129 was selected from a large panel of bispecific antibodies using an unbiased screen of ligand dependent proliferation and migration assays.
- MCLA-129 can overcome HGF mediated Erlotinib resistance in NSCLC cell lines in vitro.
- MCLA-129 shows tumor shrinkage in the Erlotinib resistant HCC827 in immunocompromised NSG-hHGFki mice by inhibiting phosphorylation of EGFR and c-MET.
- These preclinical data suggest MCLA-129 could benefit NSCLC patients that become resistant to EGFR targeted therapies and warrants clinical evaluation.

Scan the QR code for an electronic copy of the poster. These materials are for personal use only and may not be reproduced without permission.

References

1. Tsuji et al. *Oncotarget*. 2017;8(42):71805-71816
2. Engelman et al. *Science.* 2007;316(5827):1039-1043.
3. Goyama et al. *Blood.* 2015;125(17):2630-2640.

Disclosures

Cecile Geuijen, Tristan Gallenne, Roy Nijhuis, Therese Visser, Willem Bartelink, Carina Bartelink-Clements, Vanessa Zondag-van der Zande, Eric Rovers, Karin de Cortie, Linda Kaldenberg-Hendriks, Berina Eppink, Rinse Klooster, John de Kruif, Mark Throsby:
Employment and stock ownership – *Merus NV*
This study was sponsored by Merus NV (Utrecht, the Netherlands)