Clinical activity of MCLA-128 (zenocutuzumab) in combination with endocrine therapy (ET) in ER+/HER2-low, non-amplified metastatic breast cancer patients with ET-resistant disease who had progressed on a cyclin-dependent kinase (CDK) 4/6 inhibitor

#1037

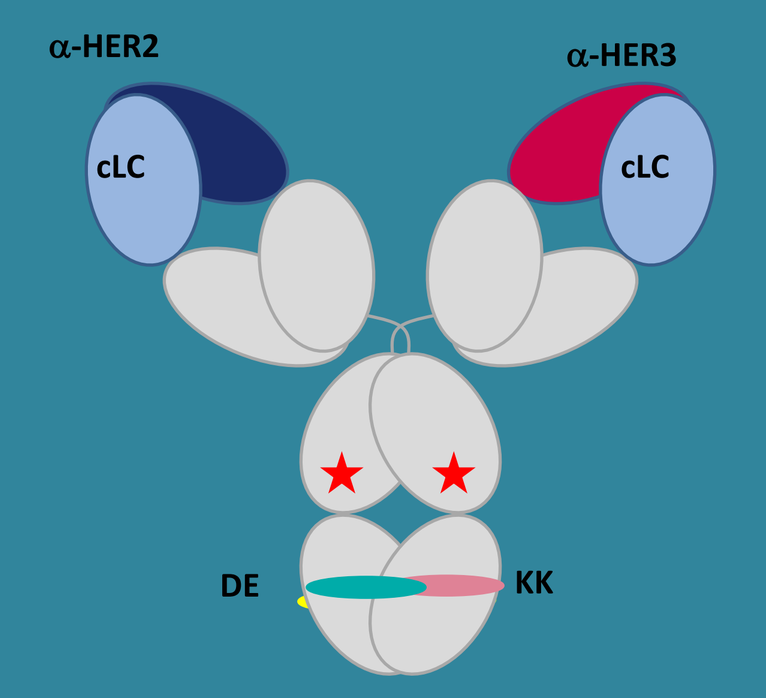
α-HER2
α-HER3
cLC
cLC
DE
KK

B Pistilli[1], H Wildiers[2], EP Hamilton[3], AA Ferreira[4], F Dalenc[5], M Vidal[6], J Gavila[7], A Goncalves[8], C Murias[9], MA Mouret-Reynier[10], JL Canon[11], F Bazan[12], S Ladoire[13], LA Sirulnik[14*], M Bekradda[15], K Bol[14], V Stalbovskaya[14], A Murat[14], J Ford[14], FC Bidard[16]

[1]Gustave Roussy, Villejuif, France; [2]University Hospitals Leuven, Leuven, Belgium; [3]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [4]Instituto Português de Oncologia, Porto, Portugal; [5]Institut Claudius Regaud, IUCT-Oncopole, CRCT, Inserm, Toulouse, France; [6]Hospital Clinic de Barcelona, Barcelona, Spain; [7]Fundacién Instituto Valenciano de Oncologla, Valencia, Spain; [8]Institut Paoli-Calmettes, Marseille, France; [9]Sarah Cannon Research Institute, London, UK; [10]GINECO-Centre Jean Perrin, Clermont-Ferrand, France; [11]Grand Hôpital de Charleroi, Charleroi, Belgium; [12]CHU Minjoz, Besançon, France; [13]Centre Georges-François Leclerc, Dijon, France; [14]Merus NV, Utrecht, Netherlands; *former employee; [15]Oncology Therapeutic Development, Clichy, France; [16]Institut Curie, Saint Cloud, France.

BACKGROUND

MCLA-128 (zenocutuzumab) is a bispecific humanized full-length IgG1 antibody that binds the transmembrane receptor tyrosine kinase human epidermal growth factor receptors 2 and 3 (HER2 and HER3).
MCLA-128 acts via two independent mechanisms of action: 1) inhibition of HER2:HER3 signaling and 2) elimination of tumor cells via enhanced antibody-dependent cell-mediated cytotoxicity (ADCC).

Figure 1. DOCK & BLOCK® mechanism action

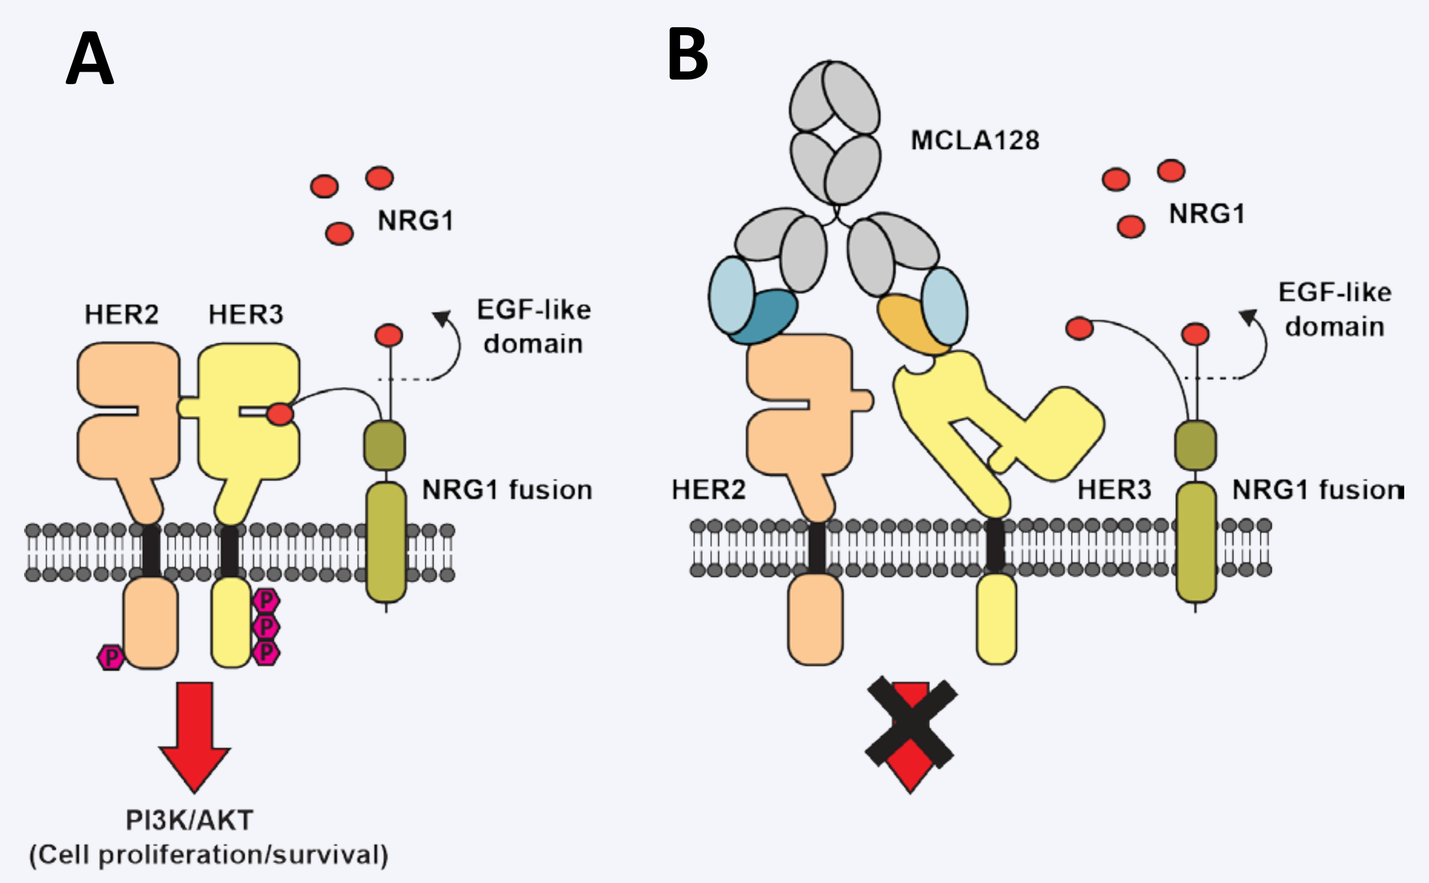
A
B
NRG1
HER2
HER3
EGF-like domain
NRG1 fusion
PI3K/AKT
(Cell proliferation/survival)
MCLA128
NRG1
EGF-like domain
HER2
HER3
NRG1 fusion

A) Ligands (such as NRG1) bind to HER3 with high affinity and promote HER2/HER3 dimerization and downstream signaling.
B) MCLA-128 inhibits the HER3-ligand interaction; one arm of the antibody binds the HER2 receptor. This optimally positions the anti-HER3 arm to block the ligand/receptor interaction, preventing HER2/HER3 dimerization and PI3K/AKT/ mTOR pathway activation.[1]

Bidirectional crosstalk between the estrogen receptor and HER2/3 may contribute to ET resistance, increasing expression of HER2 and HER3, promoting dimerization, and activating the PI3K-AKT pathway. HER2:HER3 heterodimers can also activate estrogen receptor phosphorylation.

In HER2-low breast cancer xenografts, MCLA-128 combined with endocrine therapy outperformed single-agent endocrine treatments. Consistent antitumor activity was observed with single-agent MCLA-128 in heavily pretreated HER2-amplified breast cancer patients in the first-in-human phase 1/2 study.[2]

The current open-label phase 2 study was designed to explore the efficacy of MCLA-128 to rescue patients with ET-resistant metastatic breast cancer who have progressed on a CDK4/6 inhibitor.

STUDY DESIGN

Population

- Metastatic or locally advanced breast cancer
- Hormone receptor positive & low-HER2 expression (IHC 1+ or IHC 2+ with no FISH amplification)
- Up to 3 lines of endocrine therapy (metastatic setting)
- Progression on a CDK 4/6 inhibitor
- ≤2 prior chemotherapy regimens for advanced/metastatic disease
- Measurable disease (RECIST v1.1); for bone only disease, lytic only or mixed lesions were accepted

Prior treatment

Endocrine therapy (≥12 weeks)
CDK 4/6 inhibitor
→ PD →

Study treatment

MCLA-128 + same endocrine therapy

Pre-medication | MCLA-128 750 mg 2 h IV, Day 1, q3w | Endocrine Therapy: Fulvestrant: 500 mg IM, D1, 15, 29, q4w; Exemestane: 25 mg per os, QD; Letrozole: 2.5 mg per os, QD; Anastrazole: 1 mg per os, QD

Assessments

- CT/MRI tumor assessment every 6 weeks
- AEs (CTCAE v4.03); LVEF every 3 months
- PK analyses for serum MCLA-128
- Biomarkers: customized Archer LiquidPlex 30-gene panel in baseline ctDNA (liquid biopsy)

Endpoints

- Primary: Clinical benefit rate (CBR) at 24 weeks, RECIST 1.1, per investigator
- Secondary: Overall response rate, duration of response, safety/tolerability, pharmacokinetics

PATIENT POPULATION

A total of 50 patients have been treated with the MCLA-128 + ET combination, and 6 of them were ongoing at the time of the efficacy cut-off of 31 March 2020. The 50 treated patients had received a median of 3.5 cycles [range 1-23].

Table 1. Demographics and disease characteristics

	N=50
Age (years), median [range]	56 [27-82]
ECOG PS (0/1), N (%)	35 (70%) / 15 (35%)
ER-positive, N (%)	50 (100%)
HER2 IHC 1+ / IHC 2+, N (%)	26 (52%) / 24 (48%)
Prior therapies	
N hormone therapy lines, median [range]	2 [1-5]
N chemotherapy lines (all settings), median [range]	1 [1-3]
Prior CDK4/6 inhibitor, N (%)	50 (100%)
N metastatic sites*, median [range]	**3 [1-6]**
Bone	37 (74%)
Liver	33 (66%)
Lymph nodes	27 (54%)
Lung	14 (28%)

* Sites present in >20% of the cohort.

SAFETY

At the safety data cut-off date of 14 November 2019, the 48 treated patients had received a median of 3 cycles [range 1-17].

Table 2. Treatment related AEs in >5% of patients and all grade ≥3 events (N=48)

	All grades	Grade 3
N patients with ≥1 related AE	**37 (77%)**	**5 (10%)**
Diarrhea	12 (25%)	0
Nausea	10 (21%)	0
Fatigue	8 (17%)	0
Asthenia	5 (10%)	1 (2%)
Infusion related reaction (IRR)	4 (8%)	2 (4%)
Abdominal pain	3 (6%)	0
Myalgia	3 (6%)	0
Stomatitis	3 (6%)	0
Vomiting	3 (6%)	0
Hypersensitivity	1 (2%)	1 (2%)
Hypokalemia	1 (2%)	1 (2%)
Hypotension	1 (2%)	1 (2%)

- No grade 4 related AEs occurred.
- No LVEF decreases considered clinically significant were reported in evaluable patients.
- IRRs (grouped term for AEs associated with the infusion) were reported in 25% of patients; 2 IRRs (both grade 3) led to treatment discontinuation.
- MCLA-128 dose interruptions occurred in 19% of patients.

ANTITUMOR ACTIVITY

- Antitumor activity (RECIST v1.1) was evaluated in 48 evaluable patients with locally confirmed hormone receptor positive, HER2-low disease, at the efficacy cut-off of 31 March 2020.
- The CBR (CR + PR + [SD at 24 weeks]) was 16.7% [90%CI 8.6-28.1].
- One patient had PR (lasting 12.3+ weeks, after SD of 55 weeks), and 20 (42%) patients had SD (from 9 to 37 weeks; Fig. 2).

Table 3. CBR, ORR, and BOR, Investigator assessed (RECIST v1.1)

	N=48
Clinical benefit rate at 24 weeks, N (%) [90%CI]	8 (16.7%) [8.6-28.1]
Overall response rate, N (%) [90%CI]	1 (2.1%)[0.1-9.5]
Best overall response (confirmed)	
Complete response	0
Partial response	1 (2.1%)
Stable disease	20 (41.7%)
Disease progression	26 (54.2%)
Unknown*	1 (2.1%)

* 1st assessment was SD at week 5; 2nd assessment was PD at week 15

Table 4. Characteristics of patients with clinical benefit

Pt ID	Age	Metastatic sites	N prior mts lines	Prior CDK4/6i + ET (response / tt duration)	MCLA-128 + ET (response /tt duration)	Genetic alterations
#1	52	Bone, liver	2	Palbociclib + letrozole (PD / 17 weeks)	MCLA-128 + letrozole (PR / 72+ weeks)	*KIT, MAP2K2, TP53*
#2	76	Bone, liver pleura	3	Ribociclib + fulvestrant (SD / 122 weeks)*	MCLA-128 + exemestane (SD / 30+ weeks)*	*TP53*
#3	53	Bone, breast	1	Palbociclib + letrozole (PD / 13 weeks)	MCLA-128 + letrozole (SD / 37 weeks)	*DDR2, KIT, TP53, KRAS, ERBB2, PTEN, SMAD4*
#4	56	Bone, liver	4	Palbociclib + fulvestrant (SD / 27 weeks)	MCLA-128 + fulvestrant (SD / 40 weeks)	*ERBB2, NRAS, NTRK3, TP53*
#5	64	Bone, liver	2	Palbociclib + fulvestrant (PR / 41 weeks)	MCLA-128 + fulvestrant (SD / 25 weeks)	*ESR1, PTEN*
#6	61	Bone, rectum, LN	2	Palbociclib + letrozole (SD / 156 weeks)	MCLA-128 + letrozole (SD / 26 weeks)	Not assayed
#7	60	Bone	3	Palbociclib + fulvestrant (SD / 43 weeks)	MCLA-128 + fulvestrant (SD / 30+ weeks)	*ALK, ERBB2, KRAS, PTEN, TP53*
#8	52	Bone, liver	5	Palbociclib + fulvestrant (SD / 88 weeks)	MCLA-128 + fulvestrant (SD / 26 weeks)	*ERBB2, ESR1, PIK3CA, PTEN*

mts= metastatic; tt = treatment. *Following the combination, the patient received single agent exemestane for 17 months before MCLA-128 was added.

- Of the 8 patients with clinical benefit, 1 (12.5%) had an actionable *PI3K* mutation; the 3 *ERBB2* mutations identified were not actionable. Further evaluation is warranted.

Figure 2: Plot of duration of exposure (weeks), onset of response, and patients with clinical benefit (N=48)

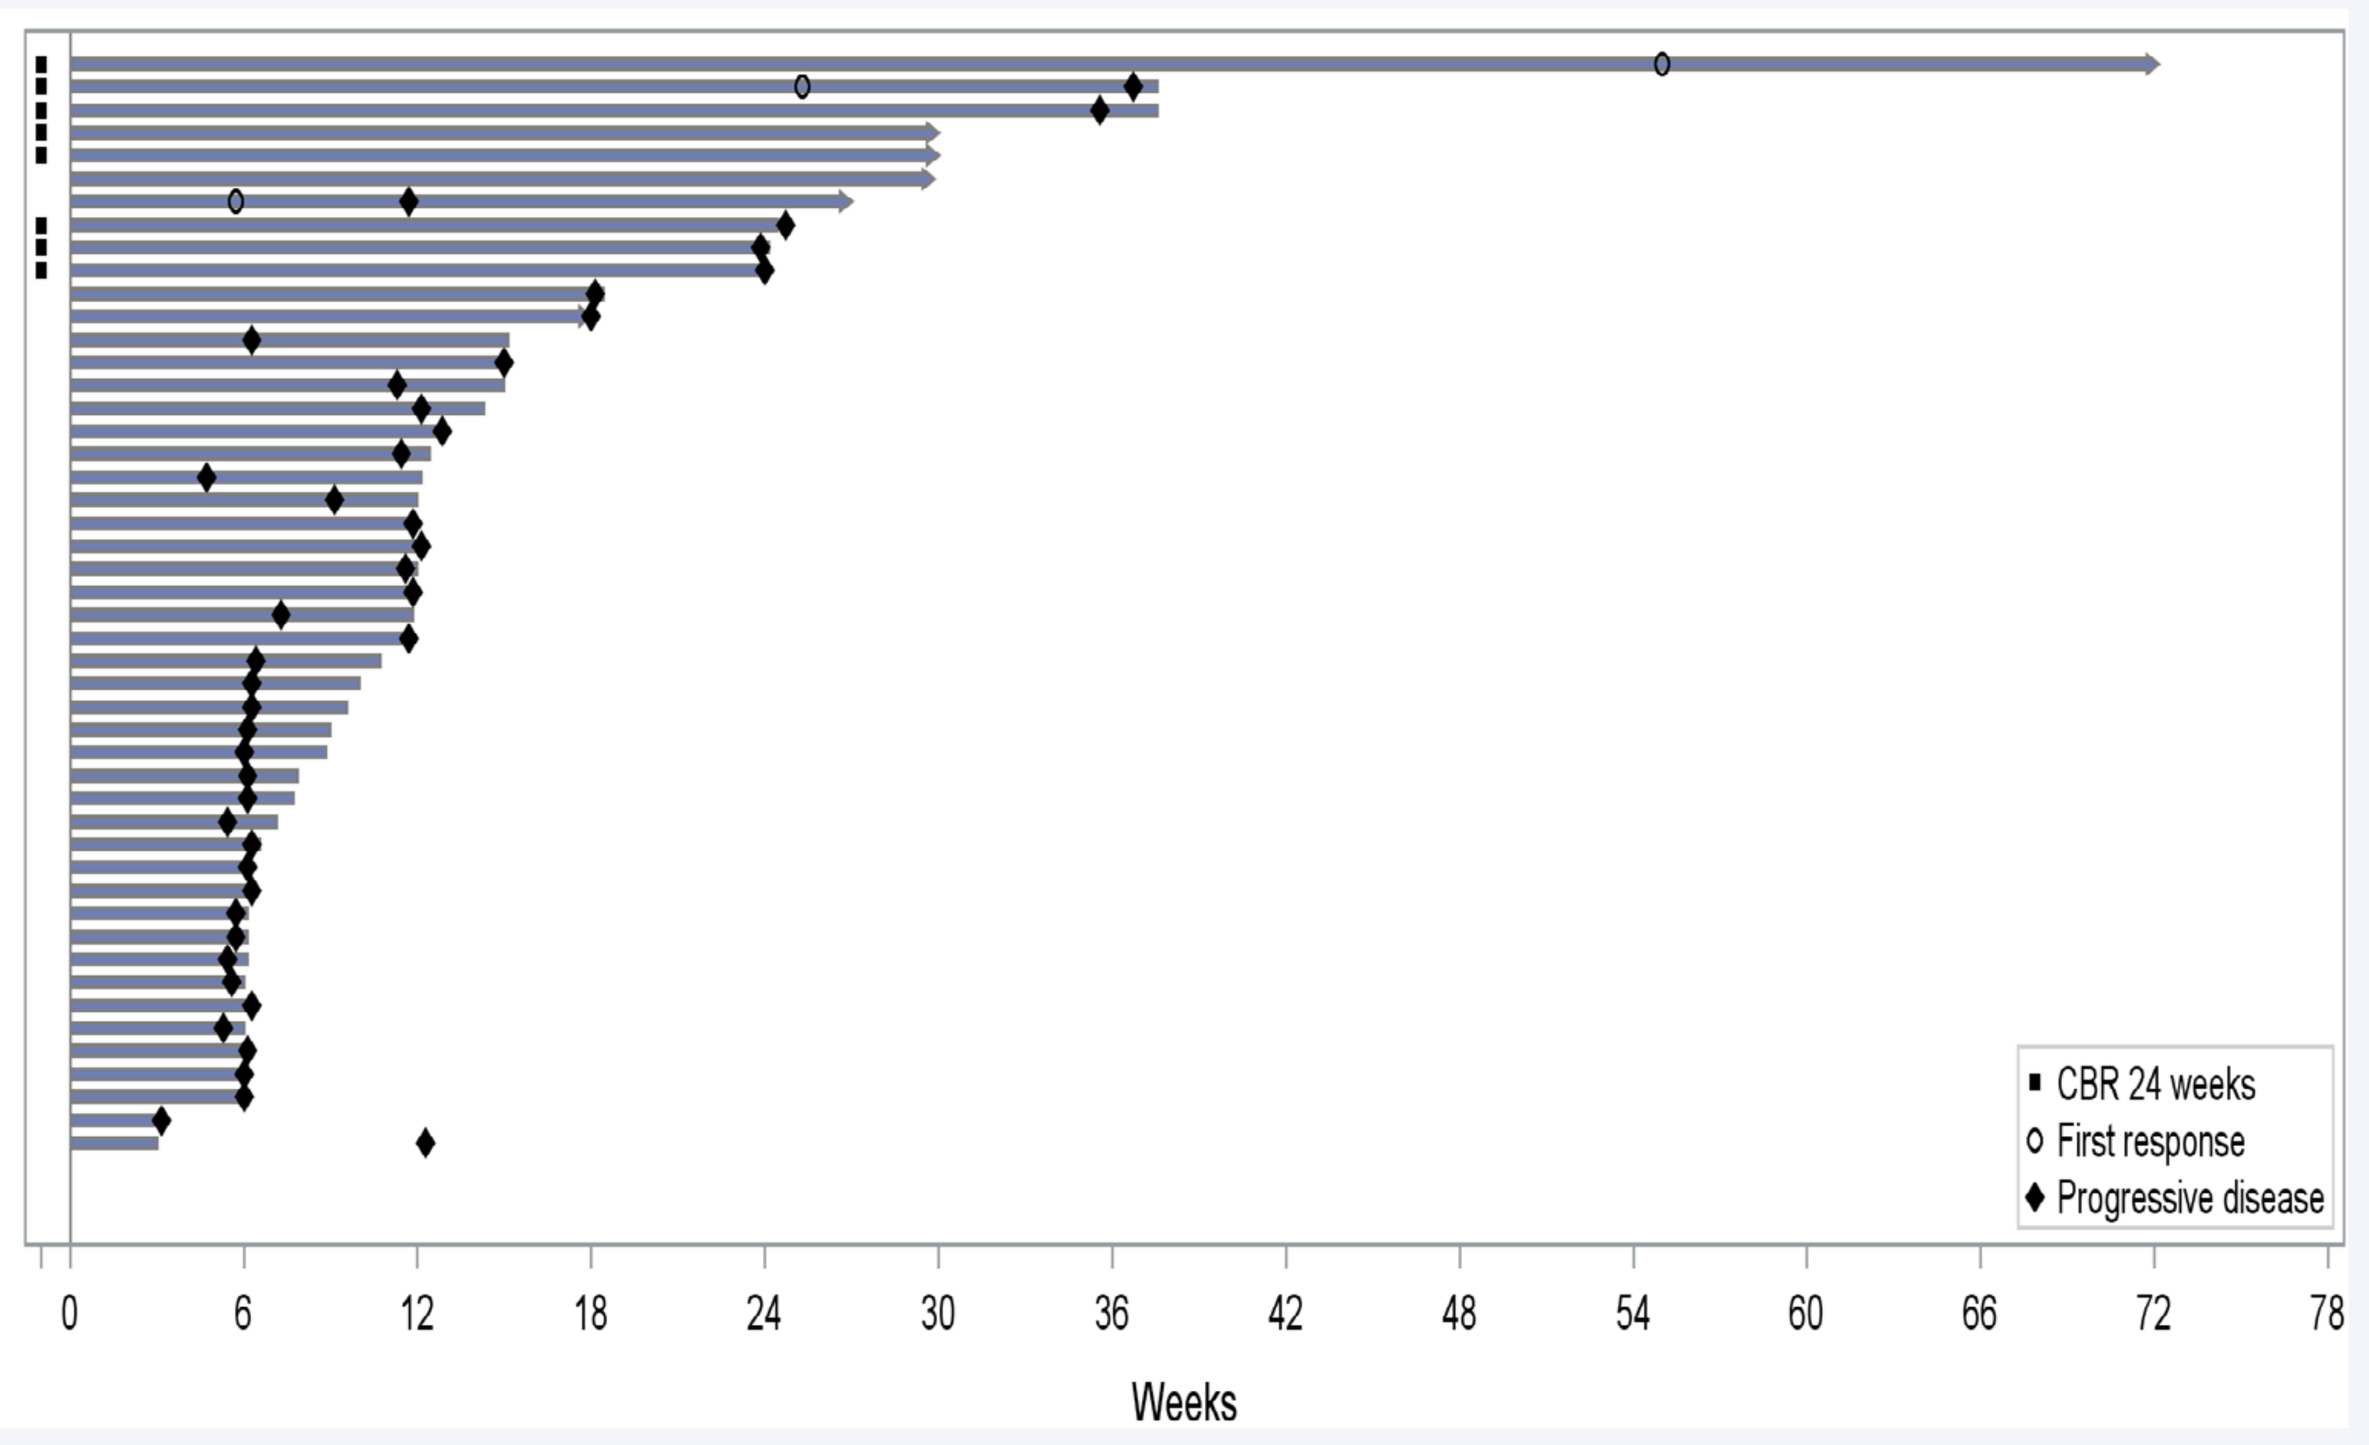
CBR 24 weeks
First response
Progressive disease
0
6
12
18
24
30
36
42
48
54
60
66
72
78
Weeks

Arrows indicate patients with ongoing study treatment

PHARMACOKINETICS

Figure 3: Serum concentration vs time curve for MCLA-128 in cycle 1

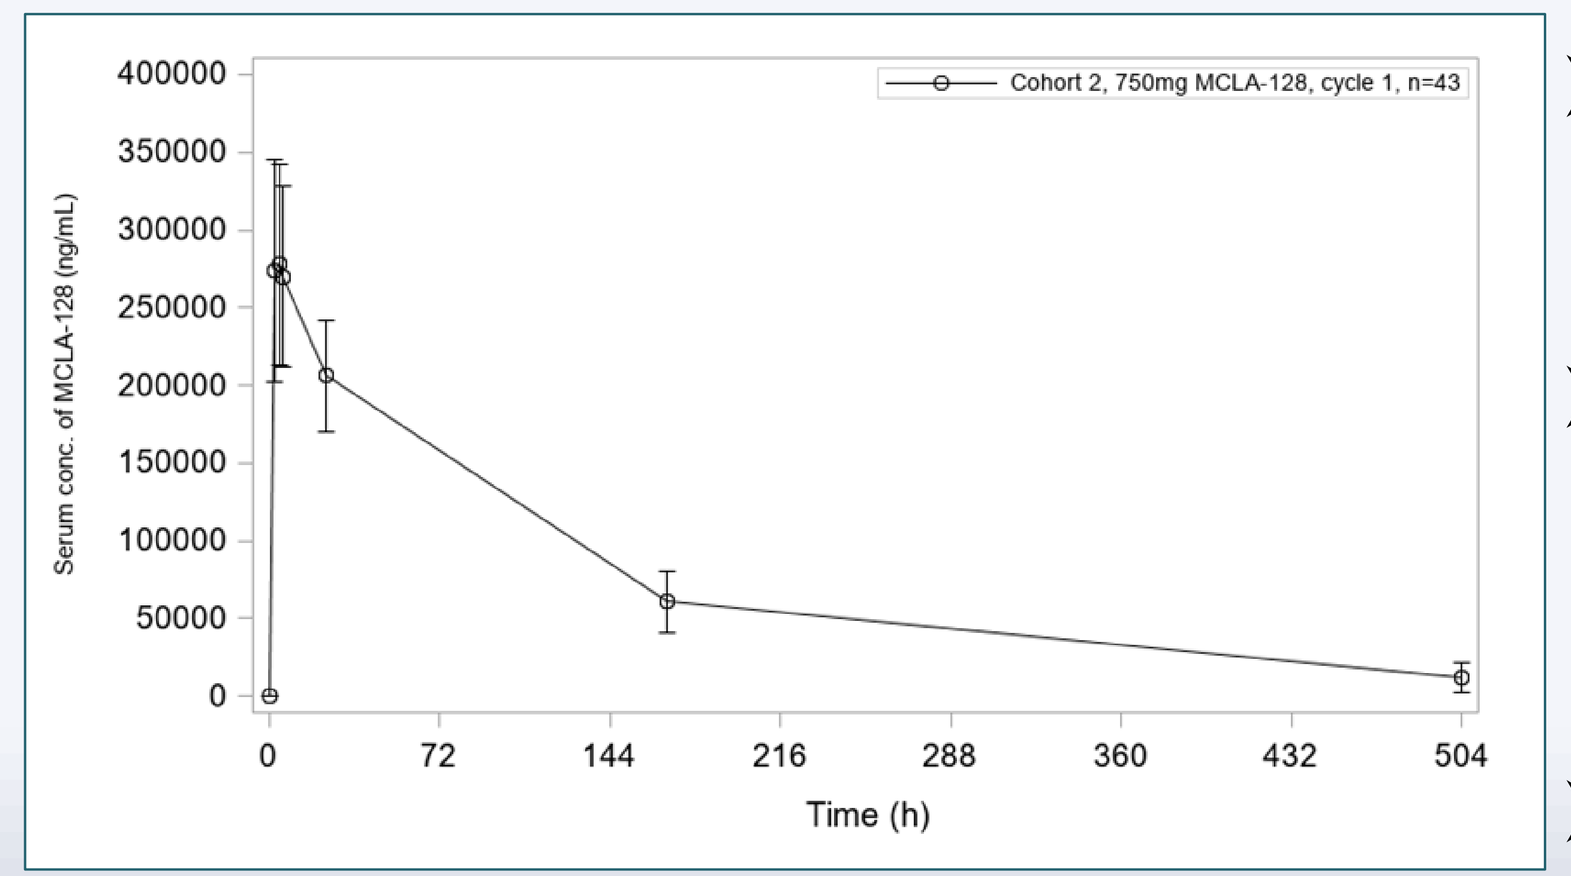
Cohort 2, 750mg MCLA-128, cycle 1, n=43
Serum conc. of MCLA-128 (ng/mL)
400000
350000
300000
250000
200000
150000
100000
50000
0
0
72
144
216
288
360
432
504
Time (h)

- PK data from 43 patients were assessed by non-compartmental analysis.
- Mean C_{max} was 307 µg/mL; AUC_{∞} was 39832 µg·h/mL; V_{ss} was 3.0 L; CL was 20.3 mL/h; t½ was 110 h.
- At mean trough level (12 µg/mL), predicted receptor occupancies for HER2 and HER3 are >90%, suggesting relevant pharmacological activity for the entire 3-week dosing interval.
- When given as part of an ET combination, MCLA-128 PK was similar to single agent PK analyses.[3]

CONCLUSIONS

- **In metastatic breast cancer patients who are endocrine-resistant and have progressed on a CDK4/6 inhibitor, addition of MCLA-128 (zenocutuzumab) to the same most recent hormone therapy resulted in clinical rescue in 17% of patients, with clinically meaningful benefit for at least 6 months.**
- **The combination of MCLA-128 with endocrine therapy is safe and well tolerated.**

References

1. Geuijen et al. *Cancer Cell.* 2018;33(5):922-36
2. Alsina et al. *J Clin Onc.* ASCO 2017; 35 (15 Suppl): 2522
3. De Vries Schultink et al. *Clinical Pharmacokinetics.* 2020; doi: 10.1007/s40262-020-00858-2

Contact
Ernesto Wasserman e.wasserman@merus.nl
Study funded by Merus N.V., enquiries@merus.nl

ASCO Annual Meeting, May 29-June 2, 2020 (Virtual Meeting)
Due to the COVID-19 pandemic, not all data were source verified.